Dr. Lord, MD, at BRI, Seattle

MOZART THERAPEUTICS

Orchestrating The Immune System

Bispecific CD8 Treg Modulators Regulate A Novel Regulatory CD8 T cell Network And Eliminate Pathogenic CD4 T cells In Live Cell Co-Culture System

Gardell, JL; Crane, C; Bowser, J; Fasnacht, R; Julien, S; Maurer, ME; Templeton, M; Therriault J; Yang SJ; Swiderek, KM

Introduction

Others have described a subset of CD8 T cells (CD8 Treg) with immunosuppressive characteristics in inflammatory disease settings. CD8 Treg activation through canonical T cell receptors results in their oligoclonal expansion and perforin dependent elimination of pathogenic CD4 T cells. We have described the CD8 Treg network in Celiac patients and its potential to eliminate pathogenic CD4 T cells*. Here we describe bispecific CD8 Treg modulators that activate CD8 Tregs, resulting in pathogenic CD4 T cell death. Here, we have demonstrated that a panel of bispecific CD8 Treg modulators with monovalent binding affinities in the low nanomolar range selectively bind the CD8 Treg subset. Within 48 hours of CD8 Treg modulator addition to T cell co-cultures, CD8 Tregs had a rapid increase of cytolytic markers and degranulation in response to CD4 T cell targets. We observed a concomitant increase of activated CD4 T cell death and decrease of pro-inflammatory cytokines. This regulatory network is present and can be modulated in organoids generated from ex vivo intestinal samples and in a humanized GVHD mouse model.

*Please see Mozart Poster #889: *"Demonstration of regulatory CD8 T cell prevalence, phenotype, and functions in autoimmune patients treated with a tolerizing peptide vaccine"*, presented on Sunday, May 8, 2022, 2:30-3:45 pm.

In autoimmunity, regulatory CD8 T cells fail to control expansion of pathogenic CD4 T cells resulting in downstream inflammation

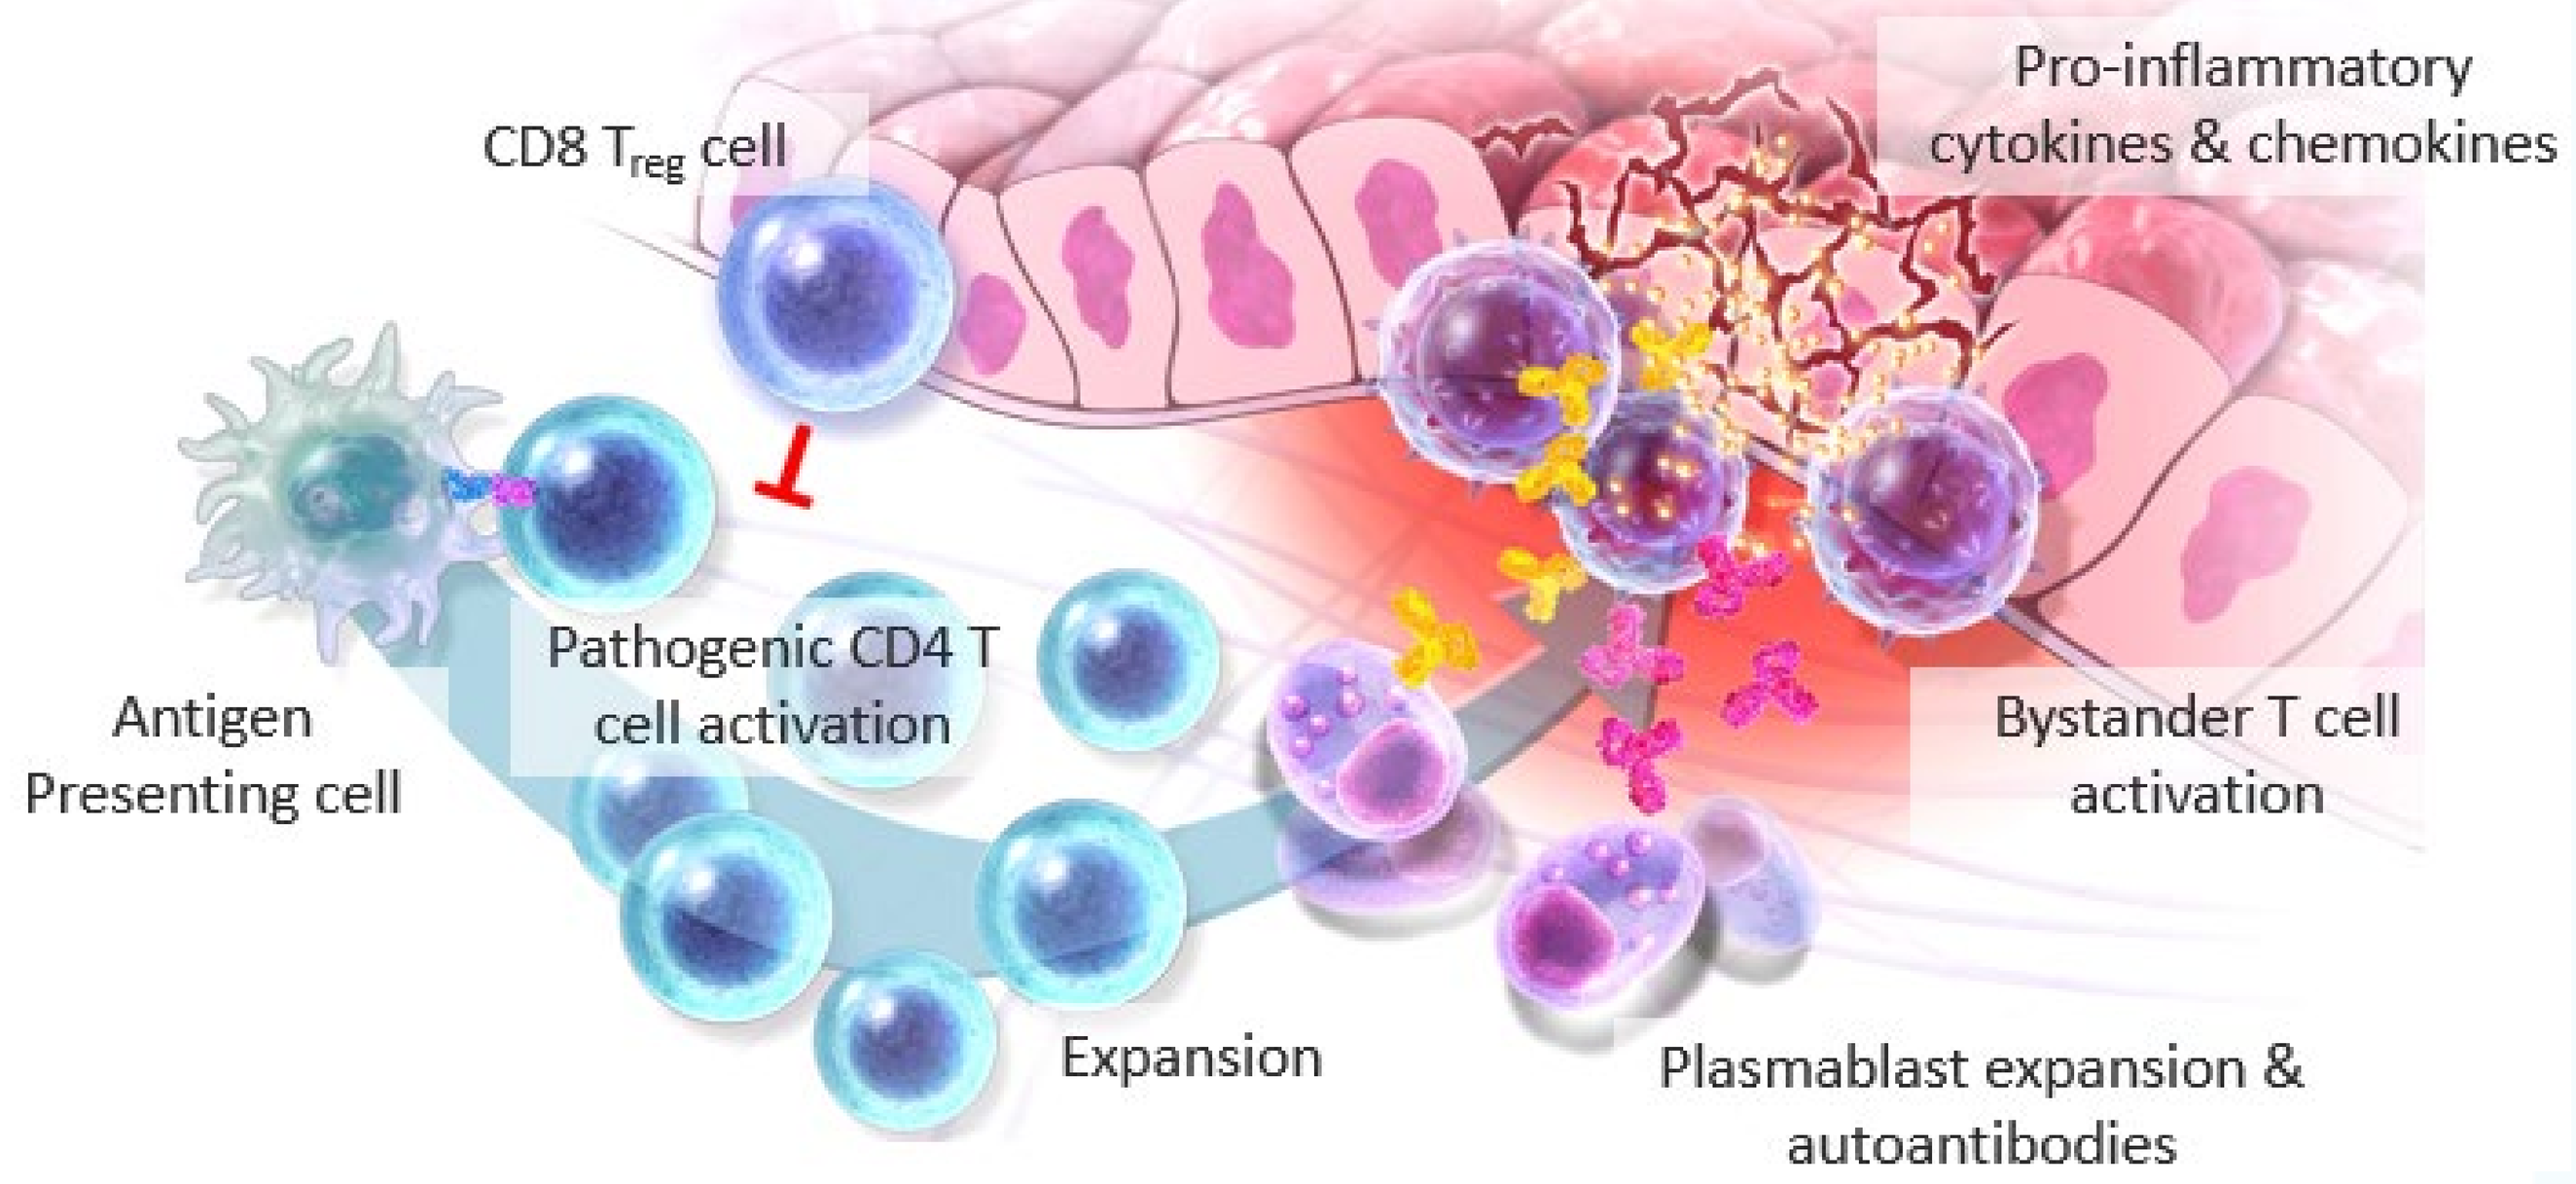
CD8 T$_{reg}$ cell
Pro-inflammatory cytokines & chemokines
Antigen Presenting cell
Pathogenic CD4 T cell activation
Bystander T cell activation
Expansion
Plasmablast expansion & autoantibodies

Methods

- Novel bispecific CD8 Treg modulators were tested for target specificity by Octet and cell binding
- The functional impact of CD8 Treg modulators was evaluated using flow and Luminex assays, and in a live cell co-culture system using Celiac patient derived CD8 Tregs and gliadin activated CD4 T cell targets
- CD8 Treg modulators with specific molecular formats were then ranked for functional efficacy

Surface receptors identified on CD8 Tregs allow for selective targeting and engagement

Recombinant Protein Binding

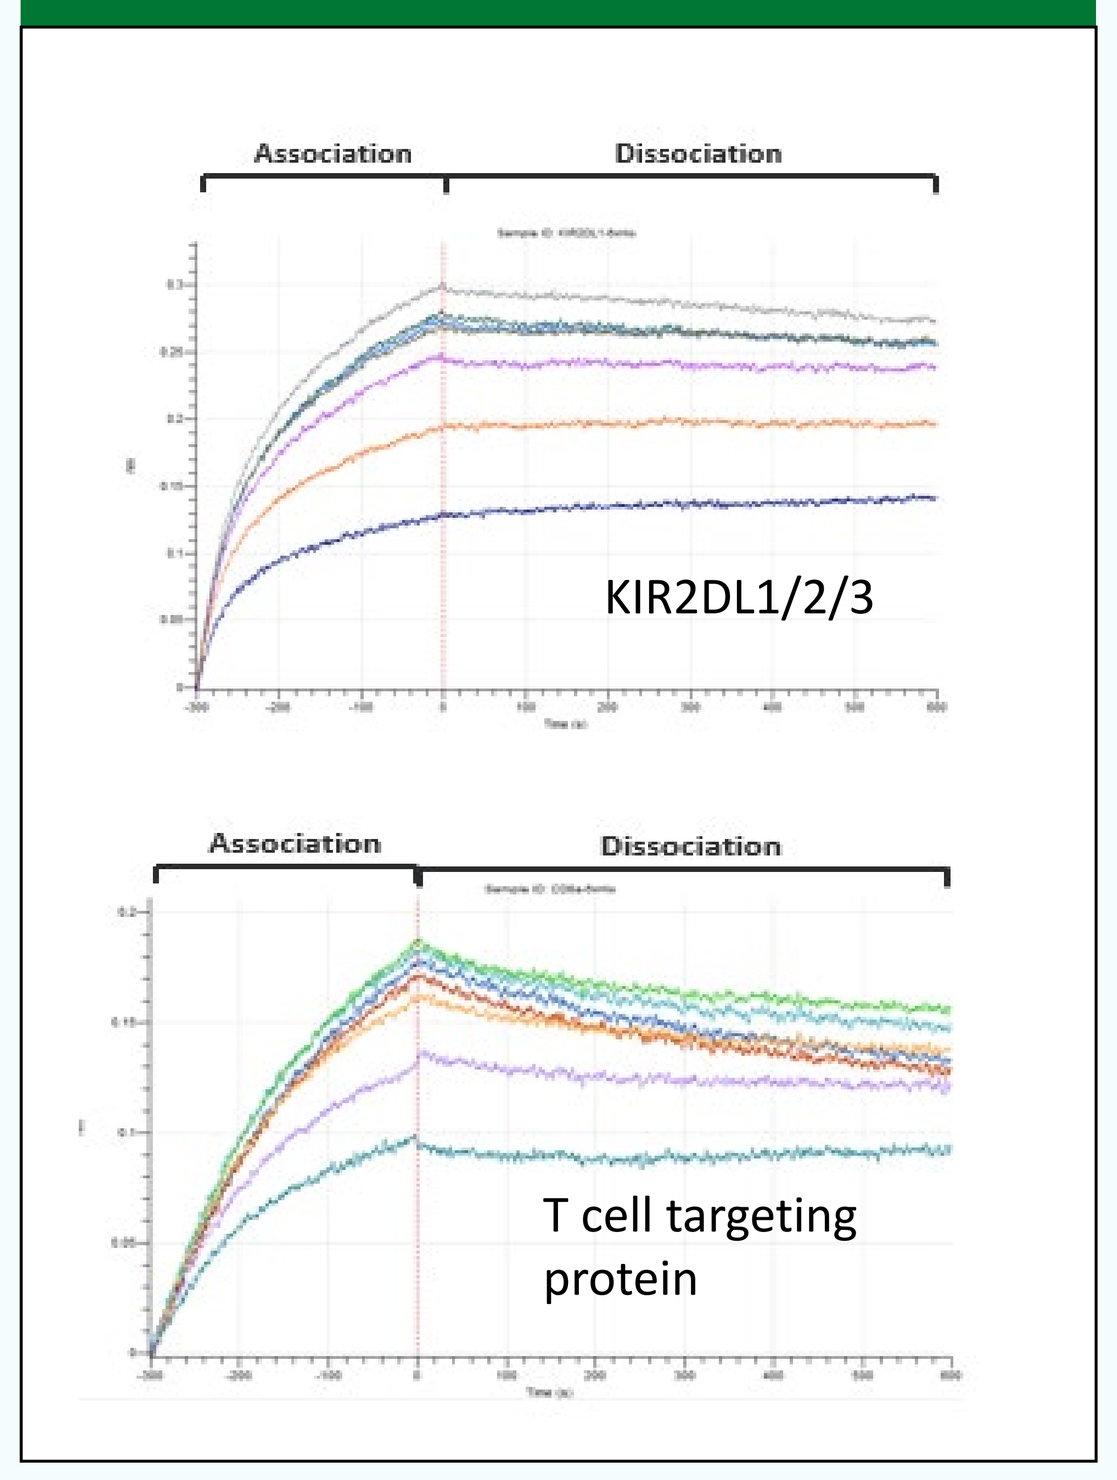
Association
Dissociation
KIR2DL1/2/3
Association
Dissociation
T cell targeting protein

CD8 Treg Target Receptor Binding

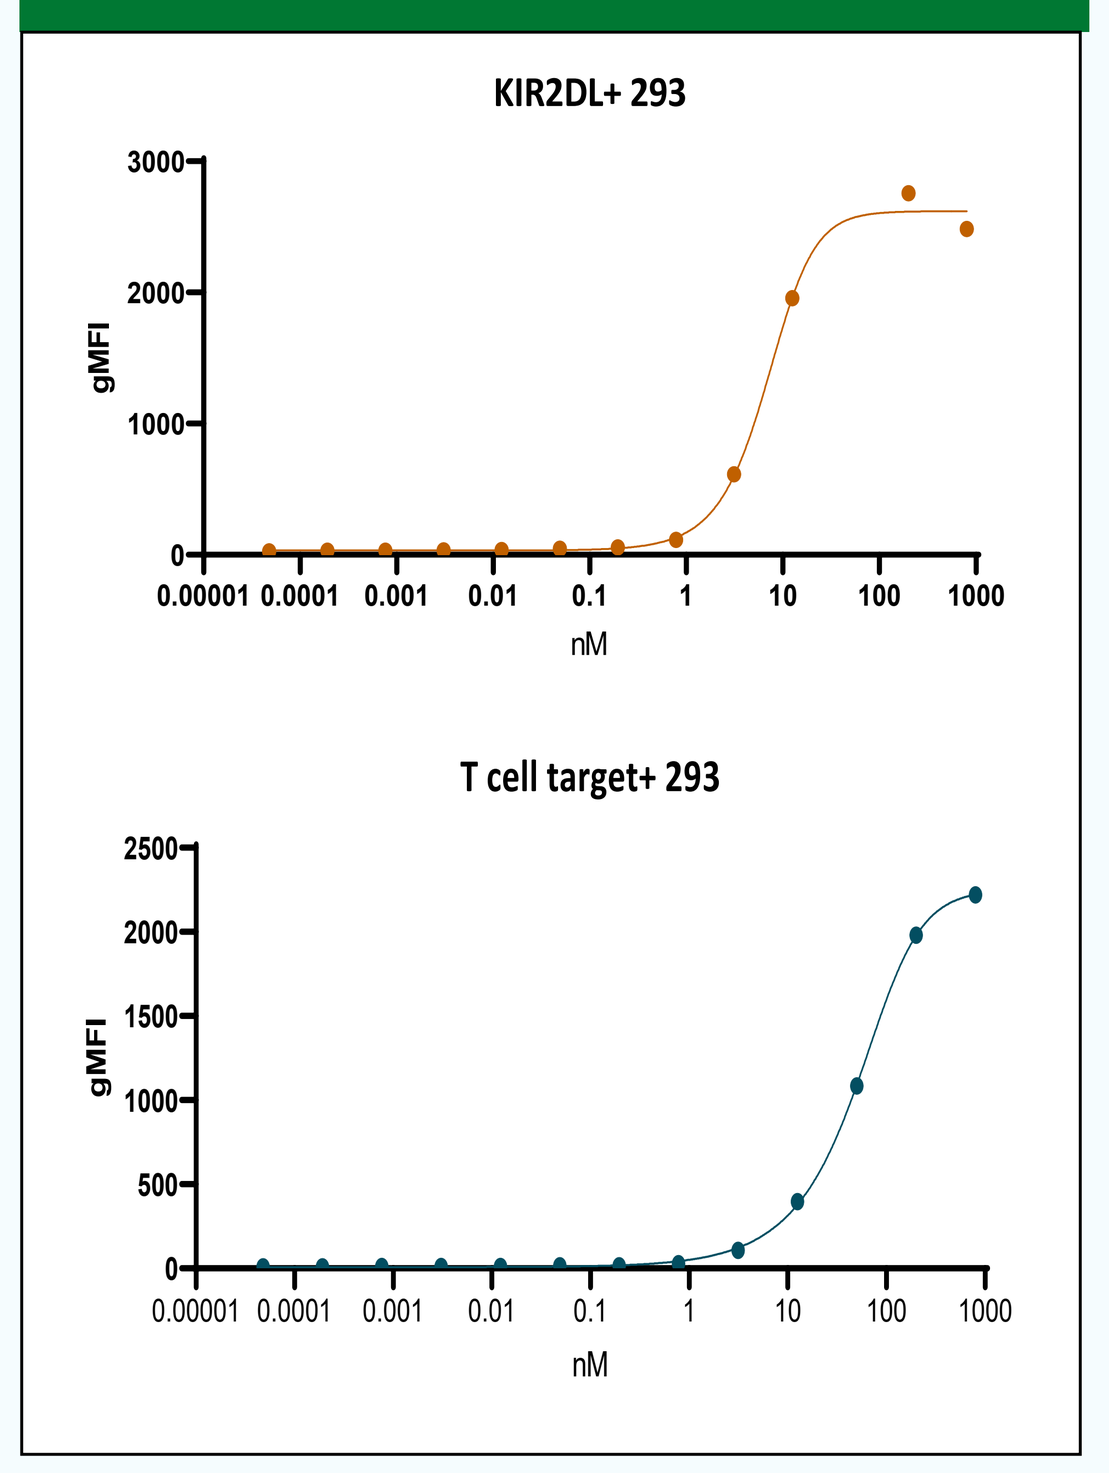
KIR2DL+ 293
gMFI
nM
T cell target+ 293
gMFI
nM

Dual Target Binding

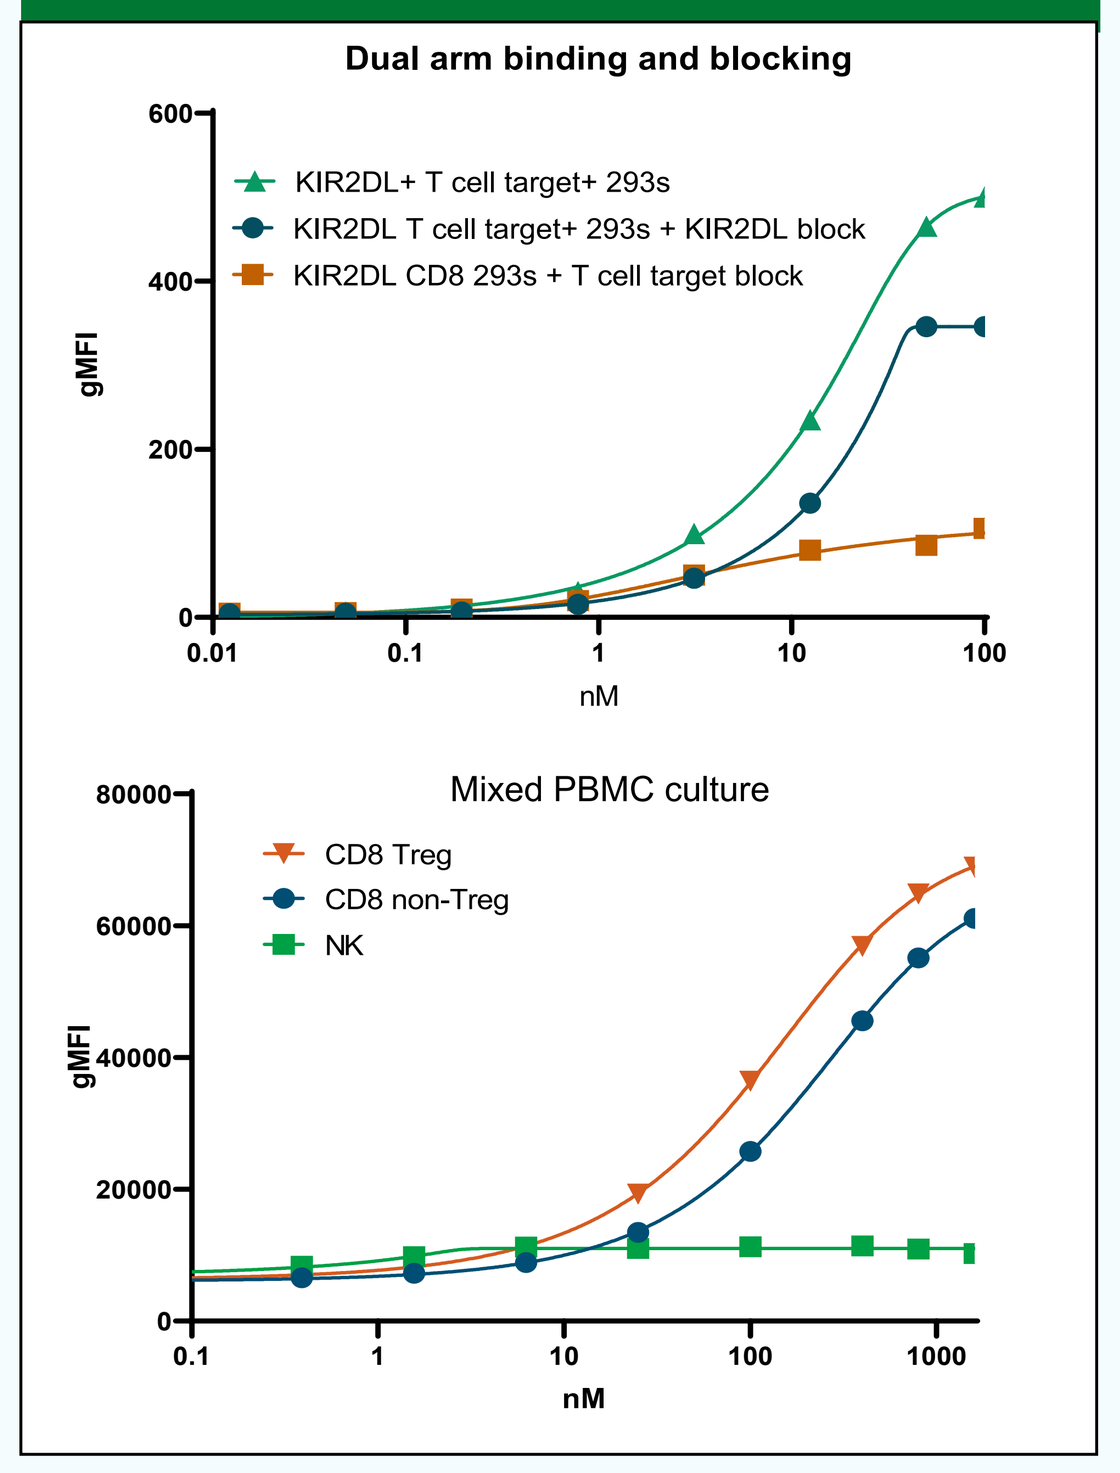
Dual arm binding and blocking
KIR2DL+ T cell target+ 293s
KIR2DL T cell target+ 293s + KIR2DL block
KIR2DL CD8 293s + T cell target block
gMFI
nM
Mixed PBMC culture
CD8 Treg
CD8 non-Treg
NK
gMFI
nM

Figure 1 Binding of bispecific CD8 Treg modulator to target recombinant proteins as detected by Octet or with anti-Fc secondary antibody following incubation with cells transfected with CD8 Treg target proteins. Bispecific CD8 Treg modulator dual binding was evaluated in mixed PBMC cultures in presence and absence of KIR and T cell targeting protein blockade, respectively.

Bispecific CD8 Treg modulators induce cytolytic function and elimination of activated pathogenic, gliadin reactive CD4s while decreasing production of proinflammatory cytokines

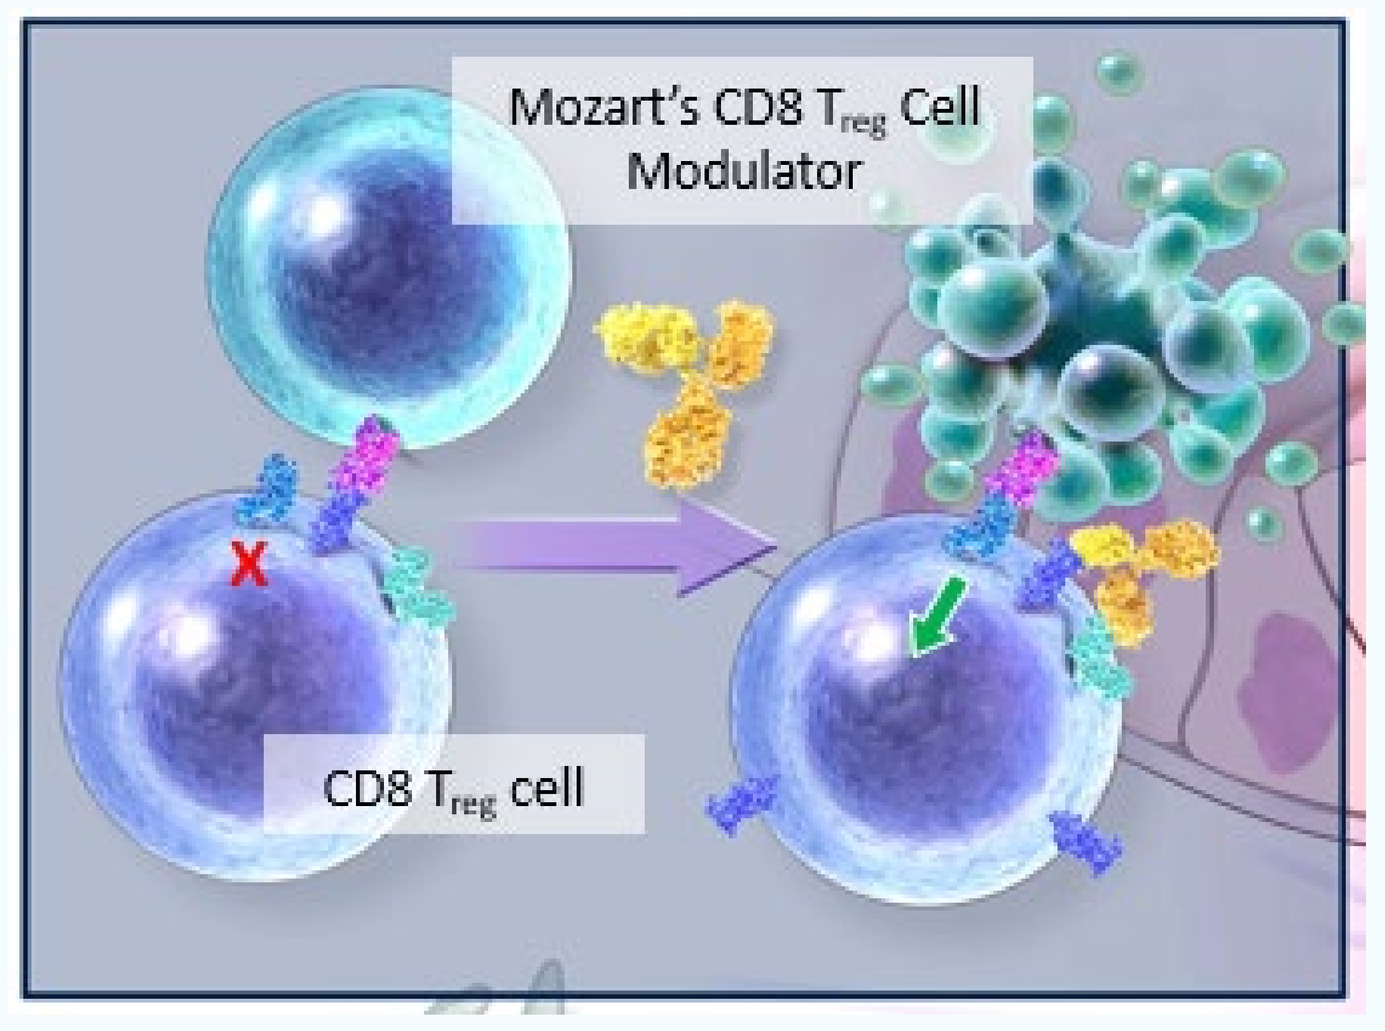
Mozart's CD8 T$_{reg}$ Cell Modulator
CD8 T$_{reg}$ cell

Cytolytic Function

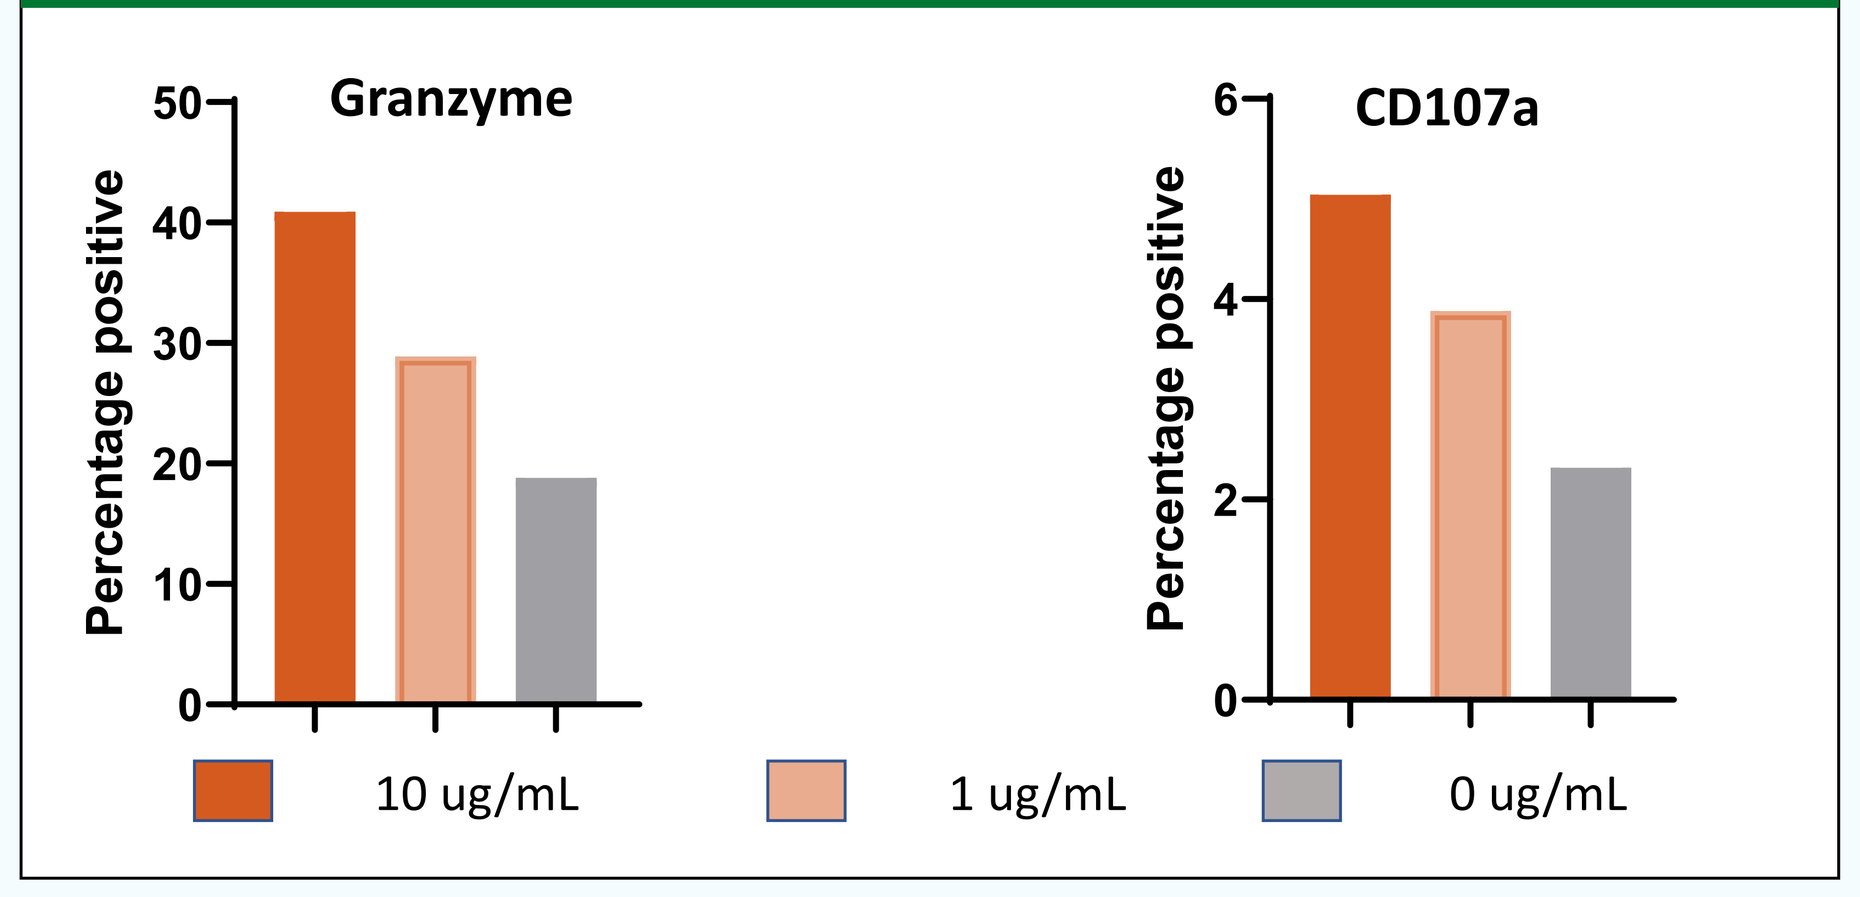
Granzyme
Percentage positive
CD107a
Percentage positive
10 ug/mL
1 ug/mL
0 ug/mL

Decreased Proinflammatory Cytokines

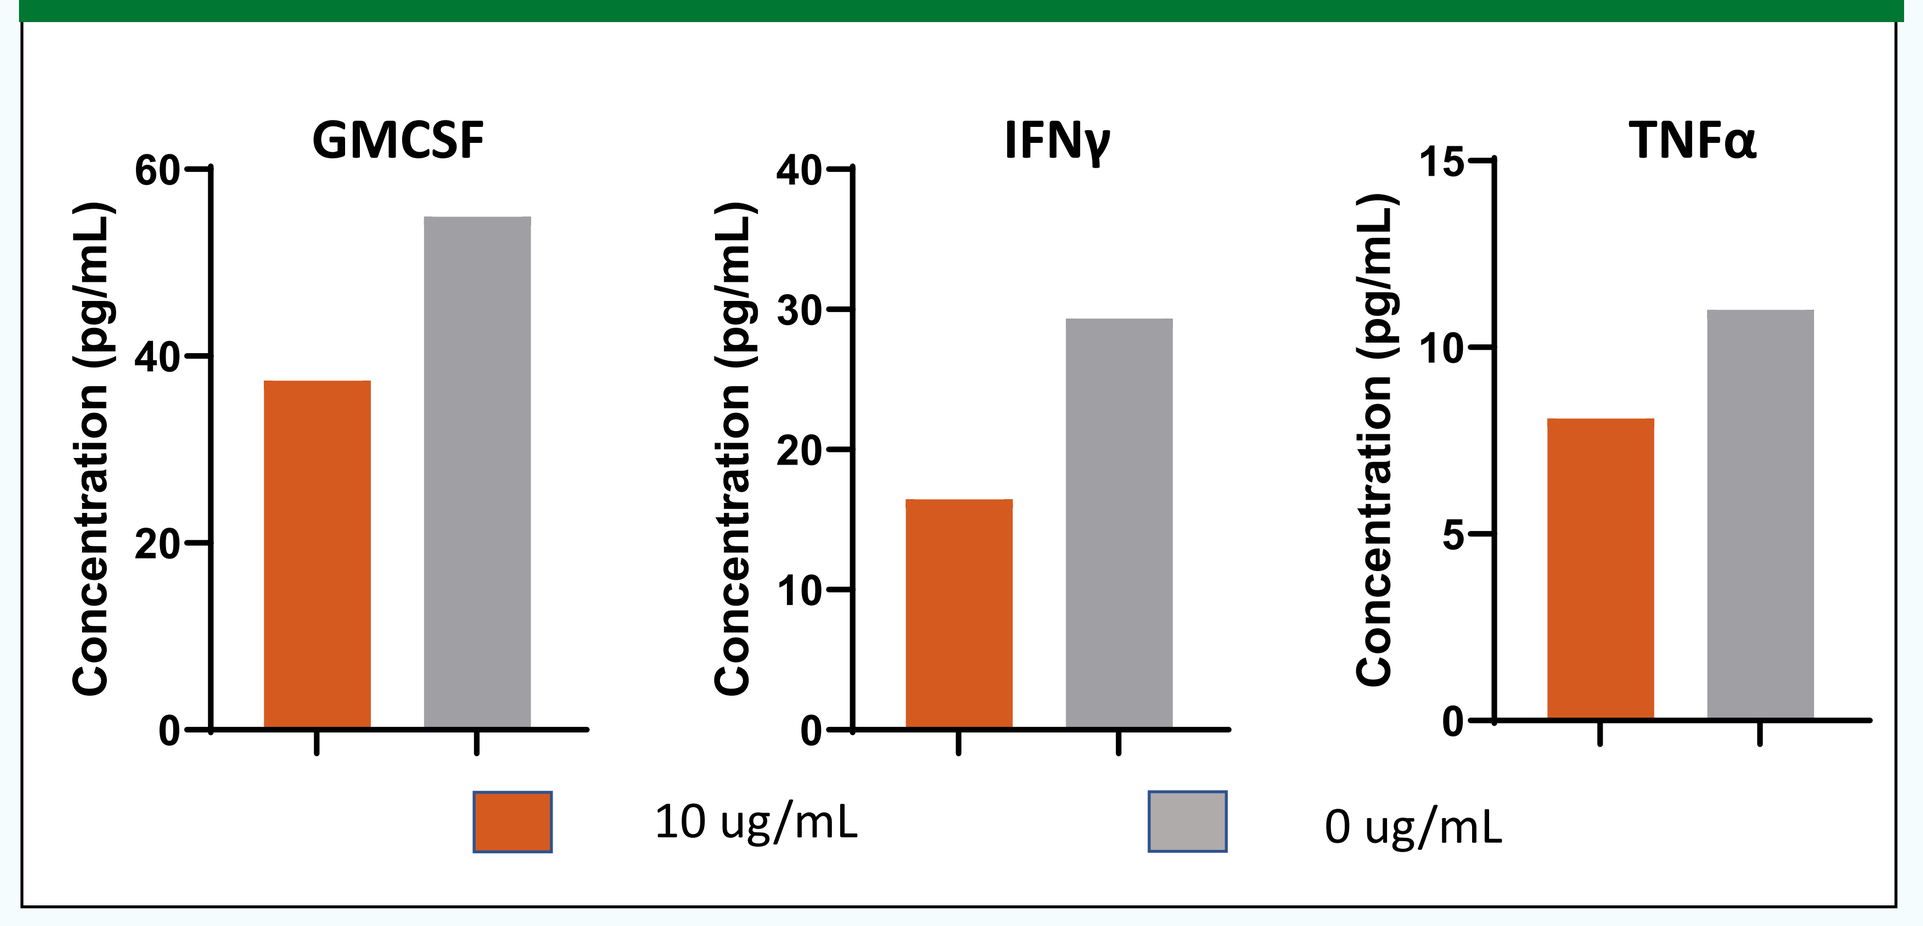
GMCSF
IFNγ
TNFα
Concentration (pg/mL)
10 ug/mL
0 ug/mL

Gliadin-specific CD4+ SKW Cell Death

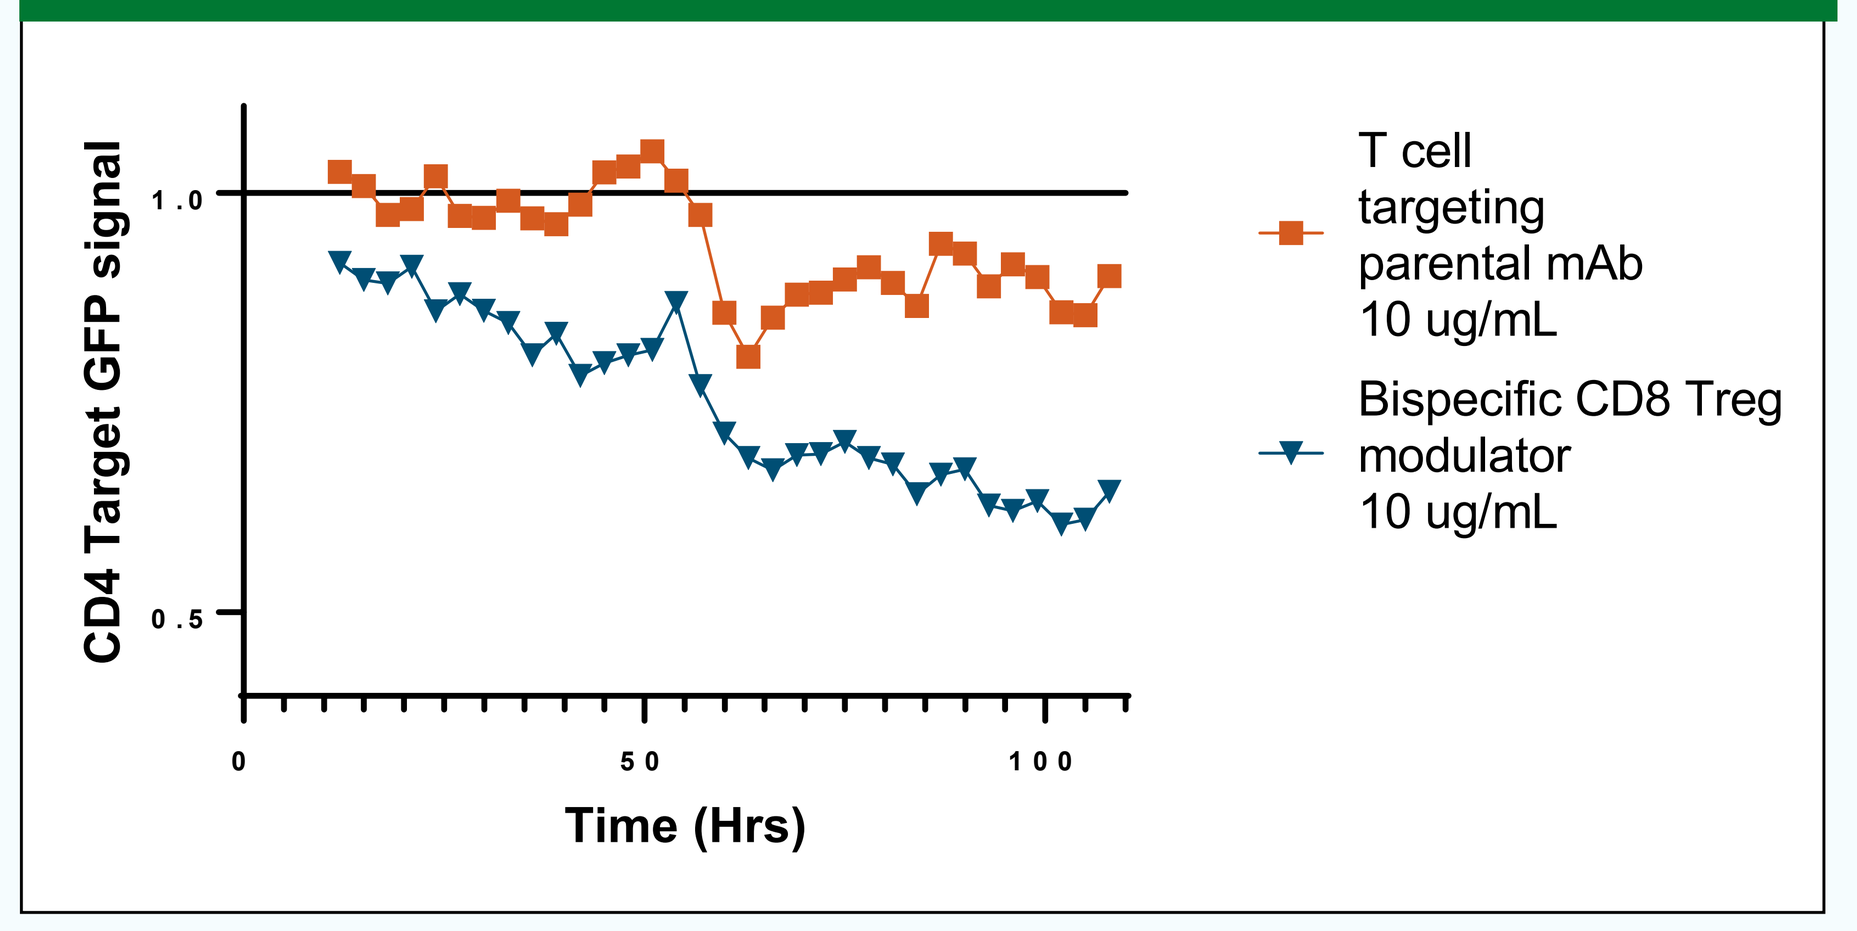
CD4 Target GFP signal
Time (Hrs)
T cell targeting parental mAb 10 ug/mL
Bispecific CD8 Treg modulator 10 ug/mL

Figure 2 Gliadin pulsed celiac PBMCs were plated with bispecific CD8 Treg modulators in the presence of 0.1 μg/mL anti-CD3 . Following incubation, CD8 Tregs were stained for Granzyme and CD107a. CD8 cells isolated from gliadin peptide pulsed PBMC cultures were incubated with bispecific CD8 Treg modulators in the presence of anti-CD3. Following incubation, supernatants were collected, and cytokine production was evaluated by Luminex assay. CD8 T regulatory cells were isolated from PBMC co-cultures and placed together with CD4 targets cells at 1 Treg:5 target cell ratio in the presence of parental or bispecific CD8 Treg modulators and gliadin-pulsed APCs. Decrease in target cell signal was measured over time by incucyte assay.

Bispecific CD8 Treg modulators decreased activated CD4s in duodenal biopsy organoid cultures

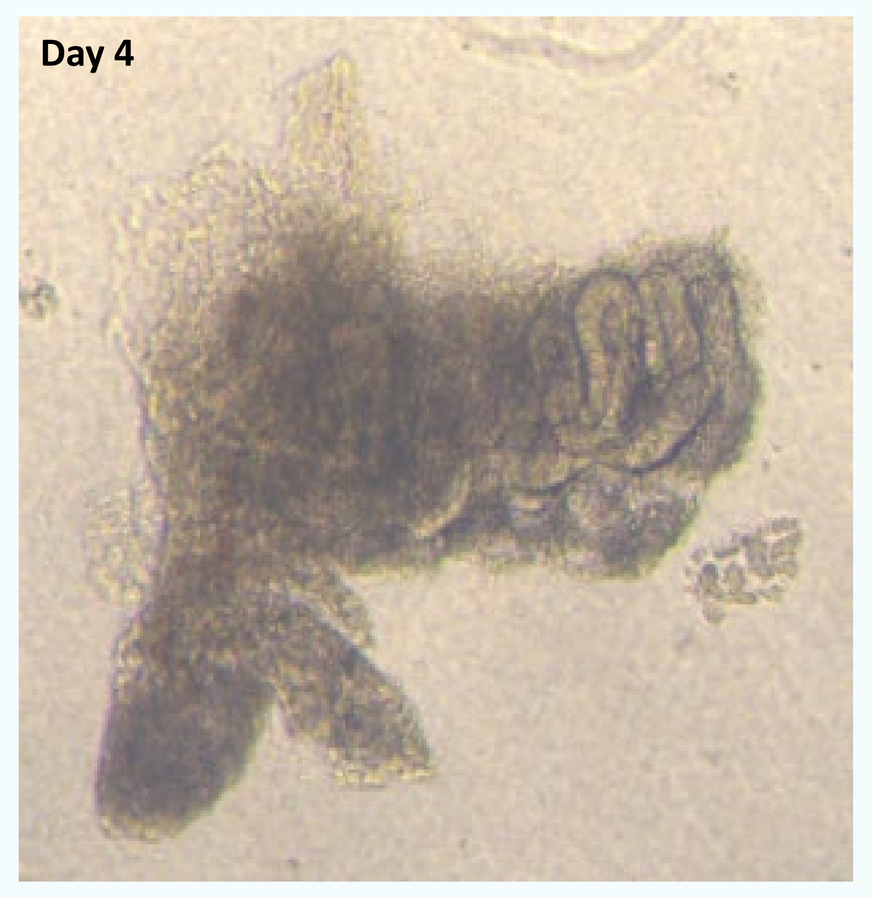
Day 4

Organoid Immune Cell Populations

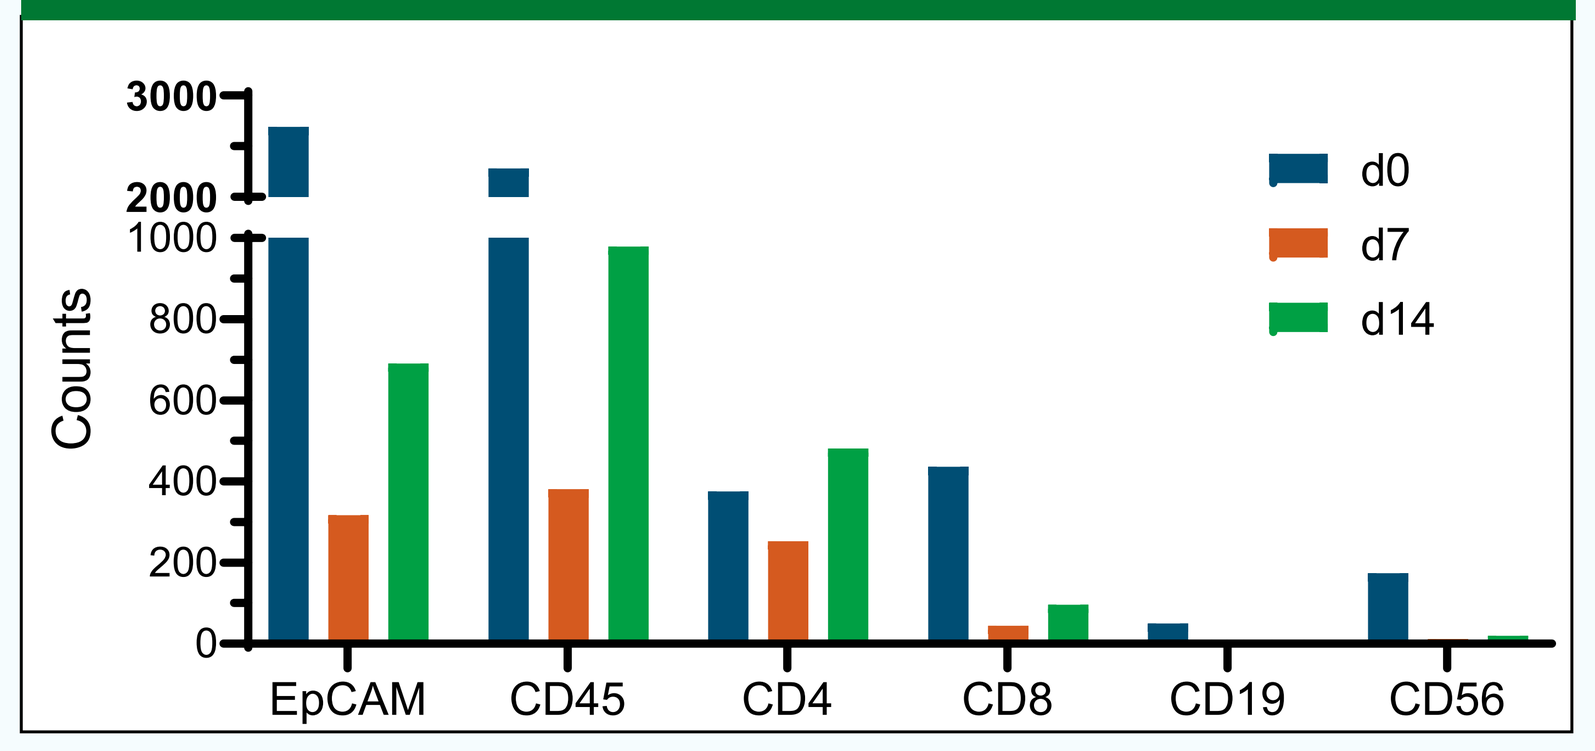
Counts
d0
d7
d14
EpCAM
CD45
CD4
CD8
CD19
CD56

Decrease in Activated CD4s

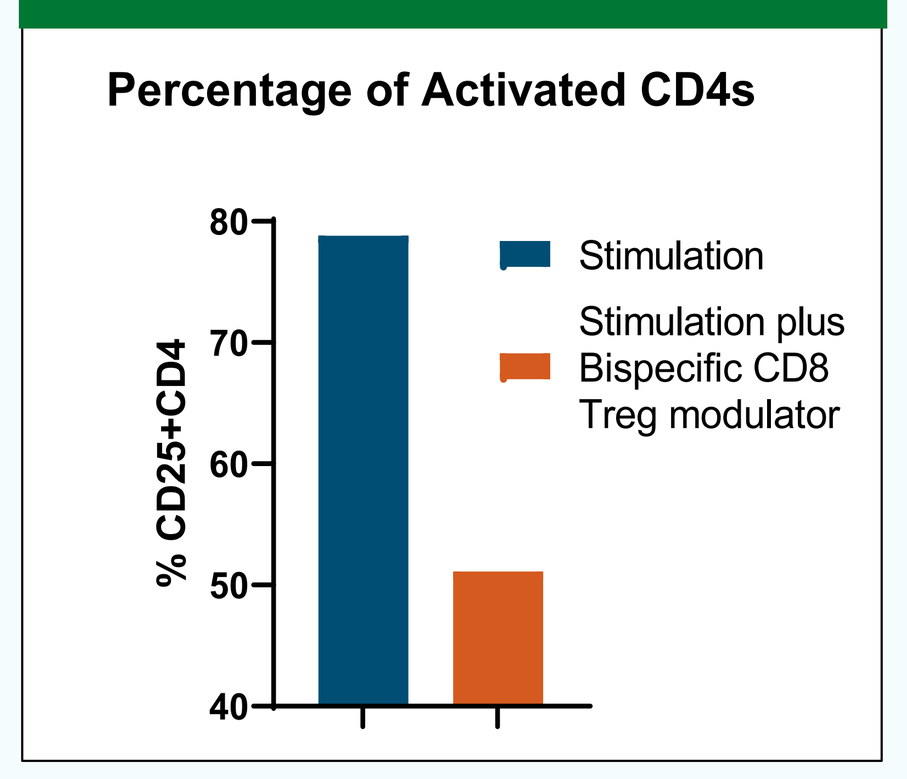
Percentage of Activated CD4s
% CD25+CD4
Stimulation
Stimulation plus Bispecific CD8 Treg modulator

Figure 4. Image of an organoid generated from an intestinal biopsy that cultured for four days in a collagen matrix in the presence of essential growth factors. Organoid were harvested following culture and processed for flow cytometry. Graphs show numbers of different immune cell populations detected. Organoids were treated with anti-CD3 and CD28 to expand T cell numbers. The addition of the bispecific CD8 modulator resulted in a reduced percentage of activated CD4s.

Bispecific CD8 Treg modulators ameliorate CD4 mediated pathology in Human PBMC-NSG™ GvHD Model

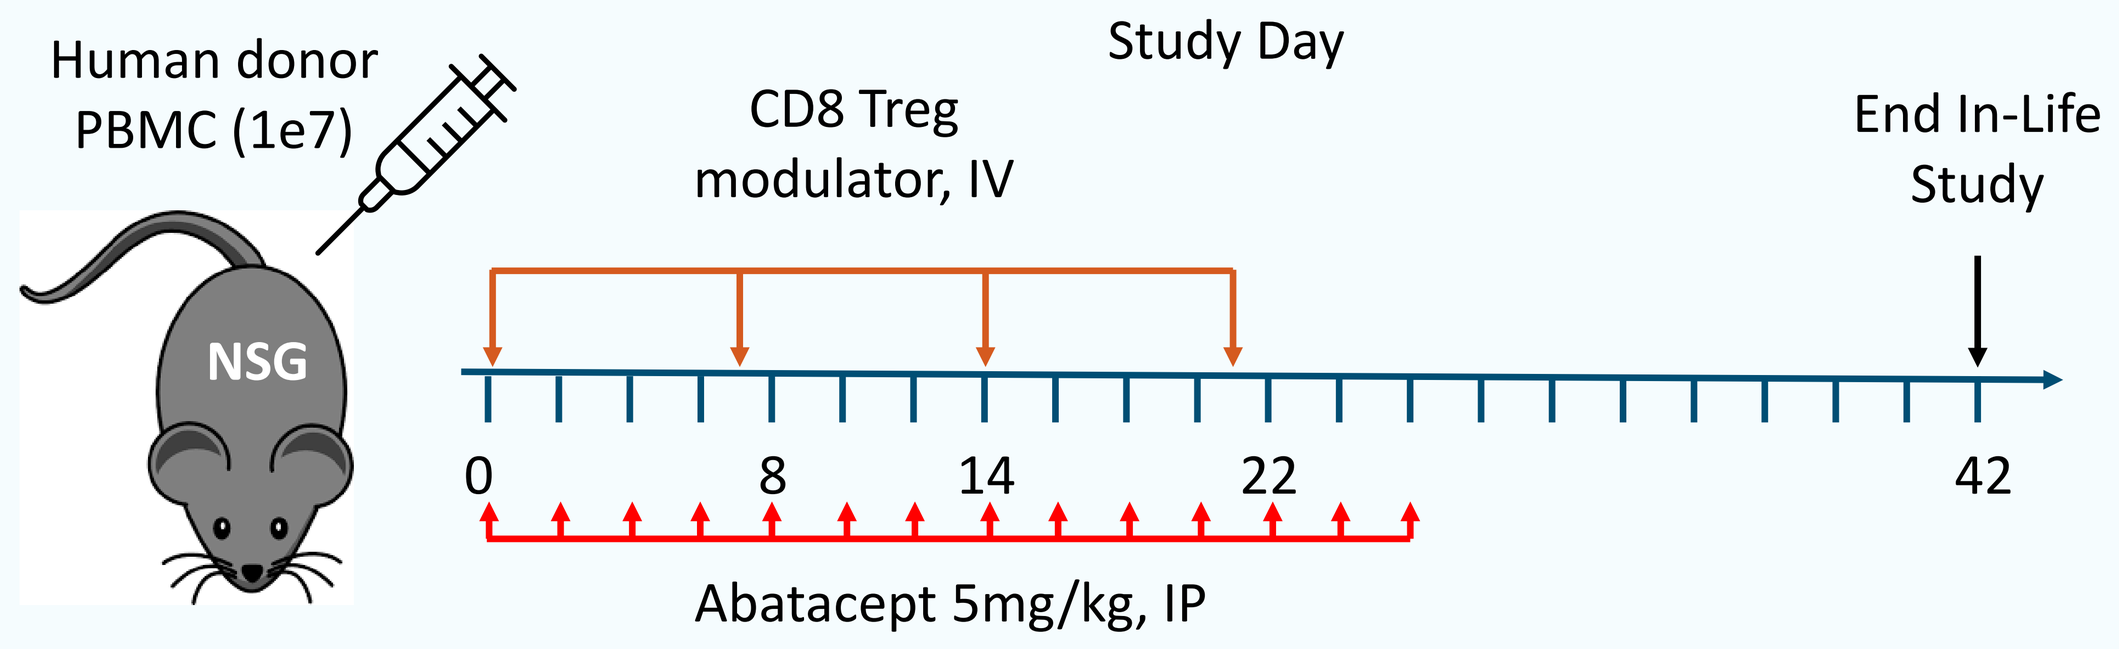
Human donor PBMC (1e7)
Study Day
CD8 Treg modulator, IV
End In-Life Study
NSG
0
8
14
22
42
Abatacept 5mg/kg, IP

Day 14 Cytokine Results

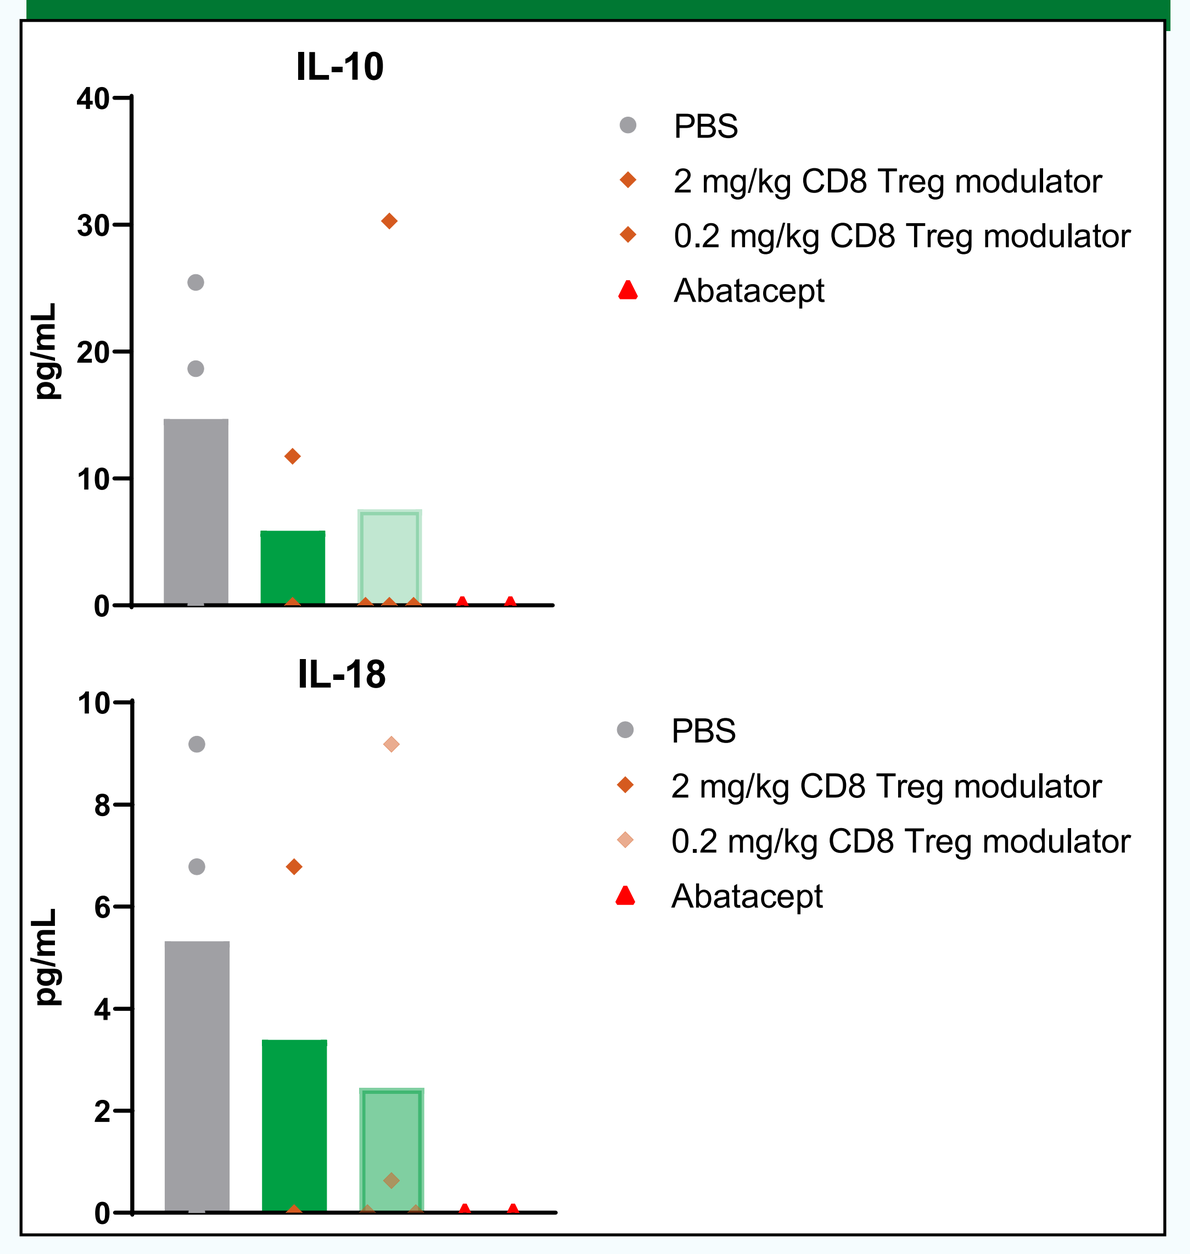
IL-10
pg/mL
PBS
2 mg/kg CD8 Treg modulator
0.2 mg/kg CD8 Treg modulator
Abatacept
IL-18
pg/mL
PBS
2 mg/kg CD8 Treg modulator
0.2 mg/kg CD8 Treg modulator
Abatacept

Histology

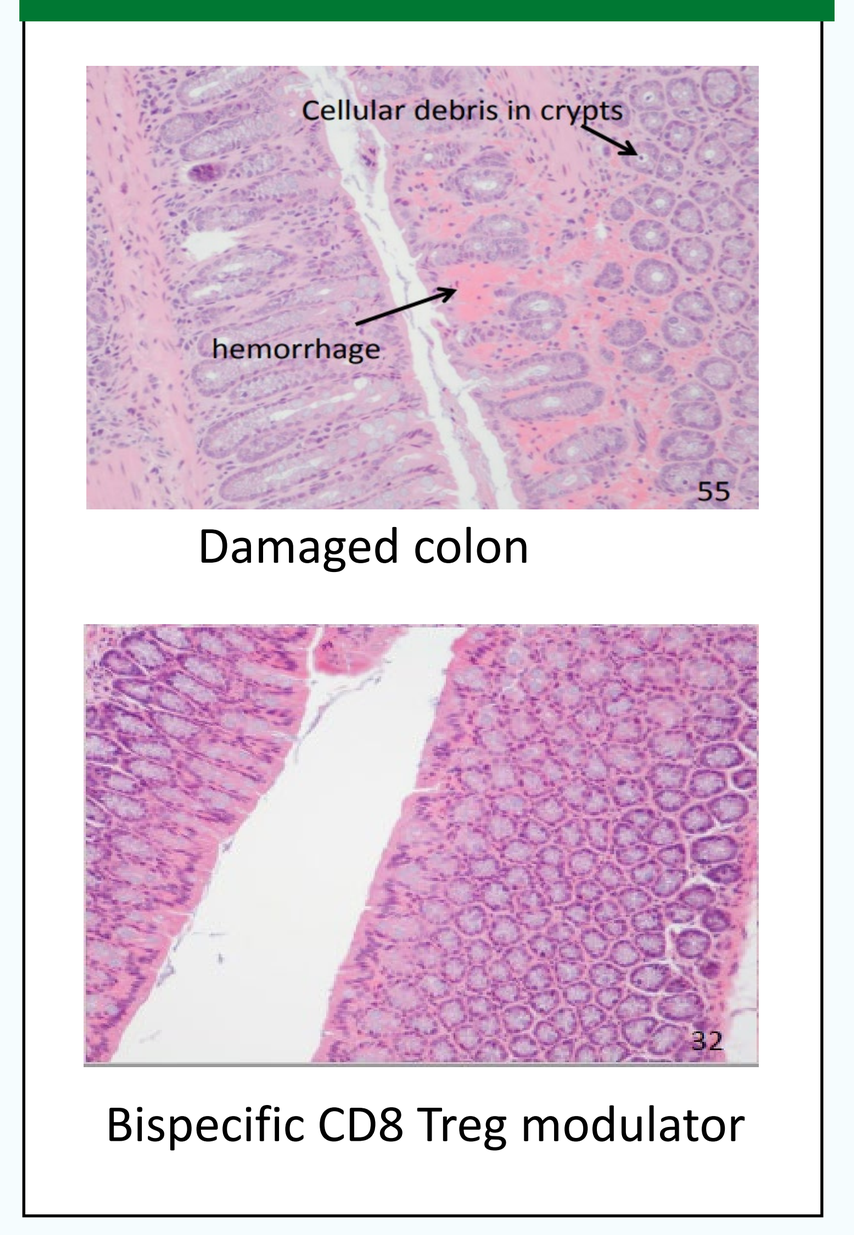
Cellular debris in crypts
hemorrhage
Damaged colon
Bispecific CD8 Treg modulator

Clinical Disease Score

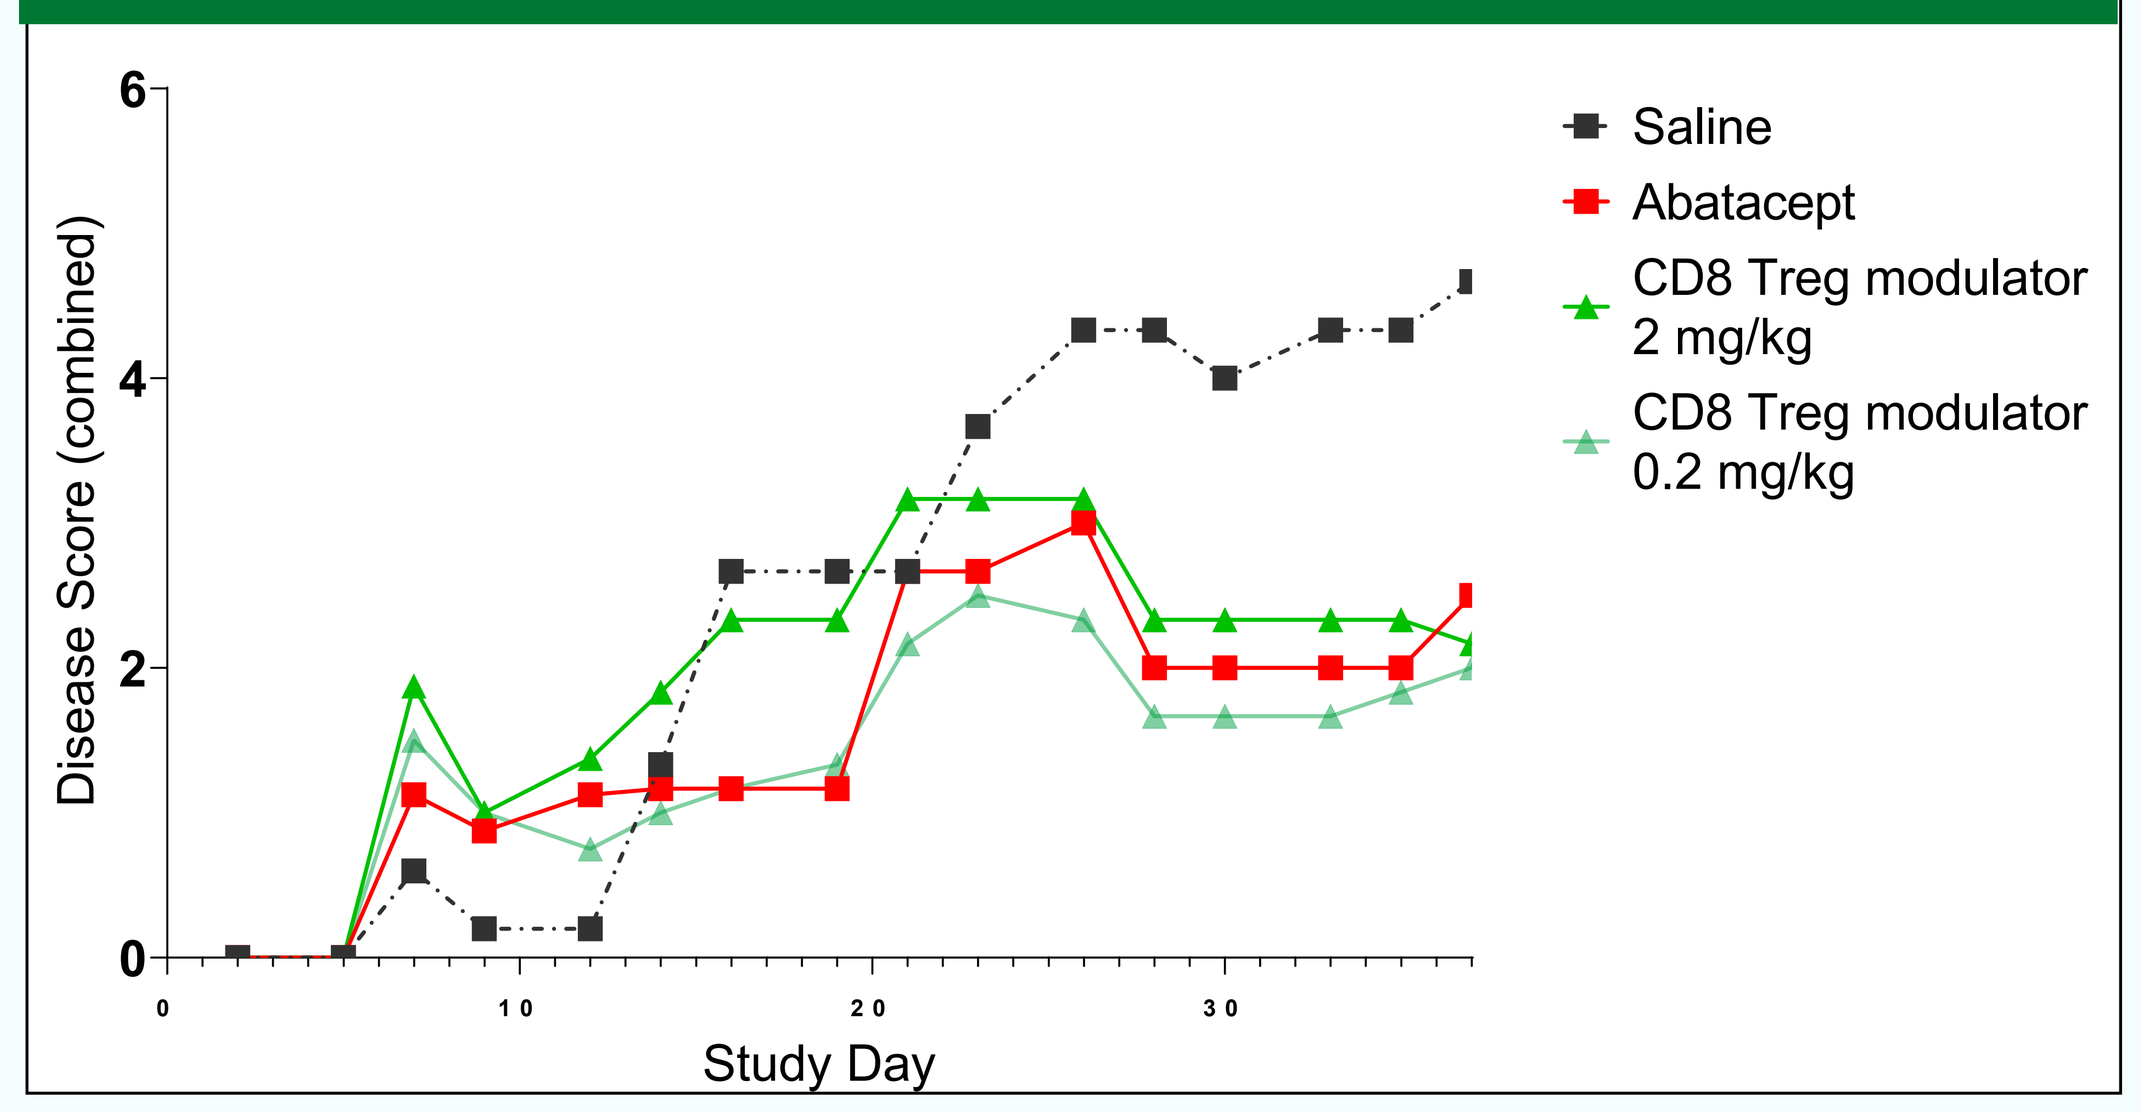
Disease Score (combined)
Study Day
Saline
Abatacept
CD8 Treg modulator 2 mg/kg
CD8 Treg modulator 0.2 mg/kg

Figure 5 NSG Mice were injected with human PBMCs followed by four weekly doses of bispecific CD8 Treg modulators or abatacept control every 48 hours for a total of 14 doses. H & E staining of colon from mice at the terminal time point revealed that mice that received the bispecific CD8 Treg modulator has normal colon structure and no sign of loss of structural integrity. Clinical disease score determined by mouse activity level, fur texture, paleness, posture, skin integrity and weight loss based on a grade 0 to 2 showed bispecific CD8 Treg modulated injected mice had scores similar to the Abatacept control group.

Conclusions

- A targeted approach to engage and activate CD8 Treg reduces pathogenic CD4 T cells in vitro.
- In an ex vivo duodenal organoid culture, we demonstrate that CD8 Treg modulator have the capacity to eliminate activated CD4 T cells.
- The CD8 Treg modulator demonstrates the capacity to ameliorate disease in a human PBMC NSG mouse GvHD model.
- We postulate that bispecific CD8 T cell modulators may represent a novel, selective and broadly applicable therapeutic approach for the treatment of human autoimmune diseases.

Acknowledgements:
The authors would like to thank Dr. Lord and patients for duodenal biopsies, members of the Kuo laboratory for assistance with the organoid culturing methods, and Jackson laboratories for performing GVHD studies.

Contact:
Follow up questions can be directed to Kristine Swiderek, Chief Scientific Officer, Mozart Therapeutics kswiderek@mozart-tx.com or by visiting the website at https://www.mozart-tx.com/

References:
Li et al. KIR $^+$ CD8 $^+$ T cells suppress pathogenic T cells and are active in autoimmune diseases and COVID-19. *Science.* 2022
DOI: 10.1126/science.abi9591