Nonclinical Toxicology and Safety Studies of MTX-101, an Inhibitory KIR2DL x CD8 Targeting Bispecific CD8 Treg Modulator, Enabling Clinical Development as Therapeutic for the Treatment of Autoimmune Disease

Catherine J. McMahan, Minh Pham, Daniel Boster, Gleda Hermansky, Lisa Bogatzki, Phoenicia Quach, Jennifer Gardell, Susan Julien, Courtney Crane and Kristine M. Swiderek

MOZART THERAPEUTICS

Orchestrating The Immune System

Introduction

In healthy individuals, CD8 Treg selectively kill pathogenic CD4 T cells. In autoimmune disease, CD8 Treg appear dysfunctional and insufficient to eliminate self-reactive CD4 T cells, in part due to expression of the autoimmune checkpoint KIR2DL(1/2/3). MTX-101 is designed to target and selectively activate only CD8 Treg to restore immune balance, reduce inflammation, pro-inflammatory cytokines and disease pathology.

MTX-101, at repeat doses up to 100 mg/kg, was evaluated in a GLP toxicology study using a humanized cord blood derived CD34+-engrafted NSG-Tg(Hu-IL15) mouse model. In-life safety, during dosing and recovery, and terminal evaluations demonstrated that MTX-101 was well-tolerated with no drug-related findings observed. Across studies, high levels of target expressing cell binding have been seen, with no adverse changes of immune cell frequency or activation status noted. No cytokine release was detected. High exposure in repeated dose studies resulted in PK consistent with CD34+ NSG-Tg(Hu-IL15), BALB/cJ mice and cynomolgus monkeys.

MTX-101 was evaluated for cytokine release in human whole blood or PBMC (multiple assay formats), with no elevation of cytokine detected. In vitro immunogenicity evaluation suggests low risk of ADA, consistent with a lack of impact to PK of MTX-101 in vivo. MTX-101 was assessed for off-target binding via the Retrogenix protein library microarray platform. No strong binding was observed other than expected interactions to its KIR2DL and CD8 targets.

MTX-101 Mechanism of Action (MOA)

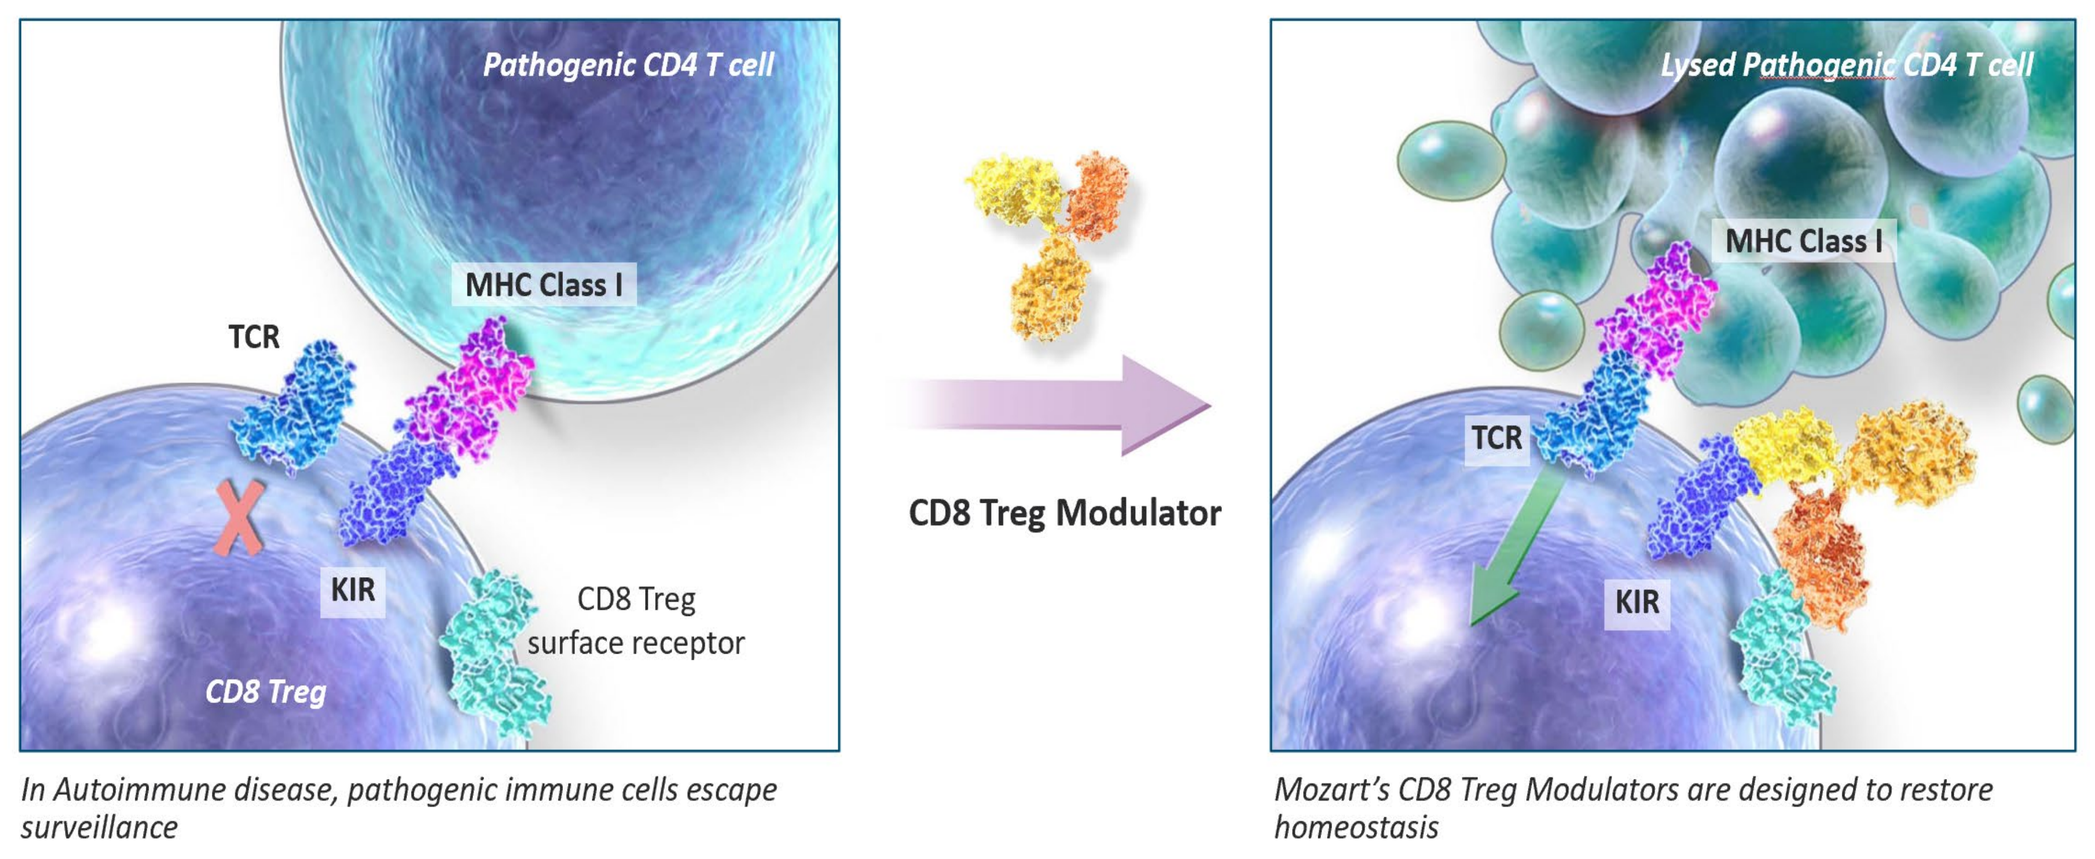
Pathogenic CD4 T cell
MHC Class I
TCR
KIR
CD8 Treg
CD8 Treg surface receptor
CD8 Treg Modulator
Lysed Pathogenic CD4 T cell
In Autoimmune disease, pathogenic immune cells escape surveillance
Mozart's CD8 Treg Modulators are designed to restore homeostasis

Mozart's approach seeks to increase cytolytic activity of dysfunctional CD8 Treg

Methods

- MTX-101, a novel CD8 Treg Modulator, was tested in healthy humanized CD34+ NSG-Tg(Hu-IL15) mice at approximately 15-22 weeks post engraftment with two independent human donor cells at the initiation of dosing.
- The in vivo pharmacological impact of MTX-101 was evaluated using flow cytometry and human U-PLEX Meso Scale Discovery (MSD) assays. In vitro, a cytokine release assessment was performed with 10 individual donors in wet coated and soluble formats.
- Quantitation of MTX-101 in humanized (CD34+ NSG-Tg(Hu-IL15)) mice was performed using an MSD assay using samples collected with a micro-sampling procedure. Serum was collected for analysis of BALB/cJ and cynomolgus macaques pharmacokinetics.
- For tolerability/toxicology assessments, a single dose study (non-GLP) and two independent repeated dose studies (non-GLP and GLP) were performed in humanized mice (CD34+ NSG-Tg(Hu-IL15)). Single dose pharmacokinetics studies were done in BALB/cJ mice and cynomolgus macaques.

Repeated dose toxicology study design

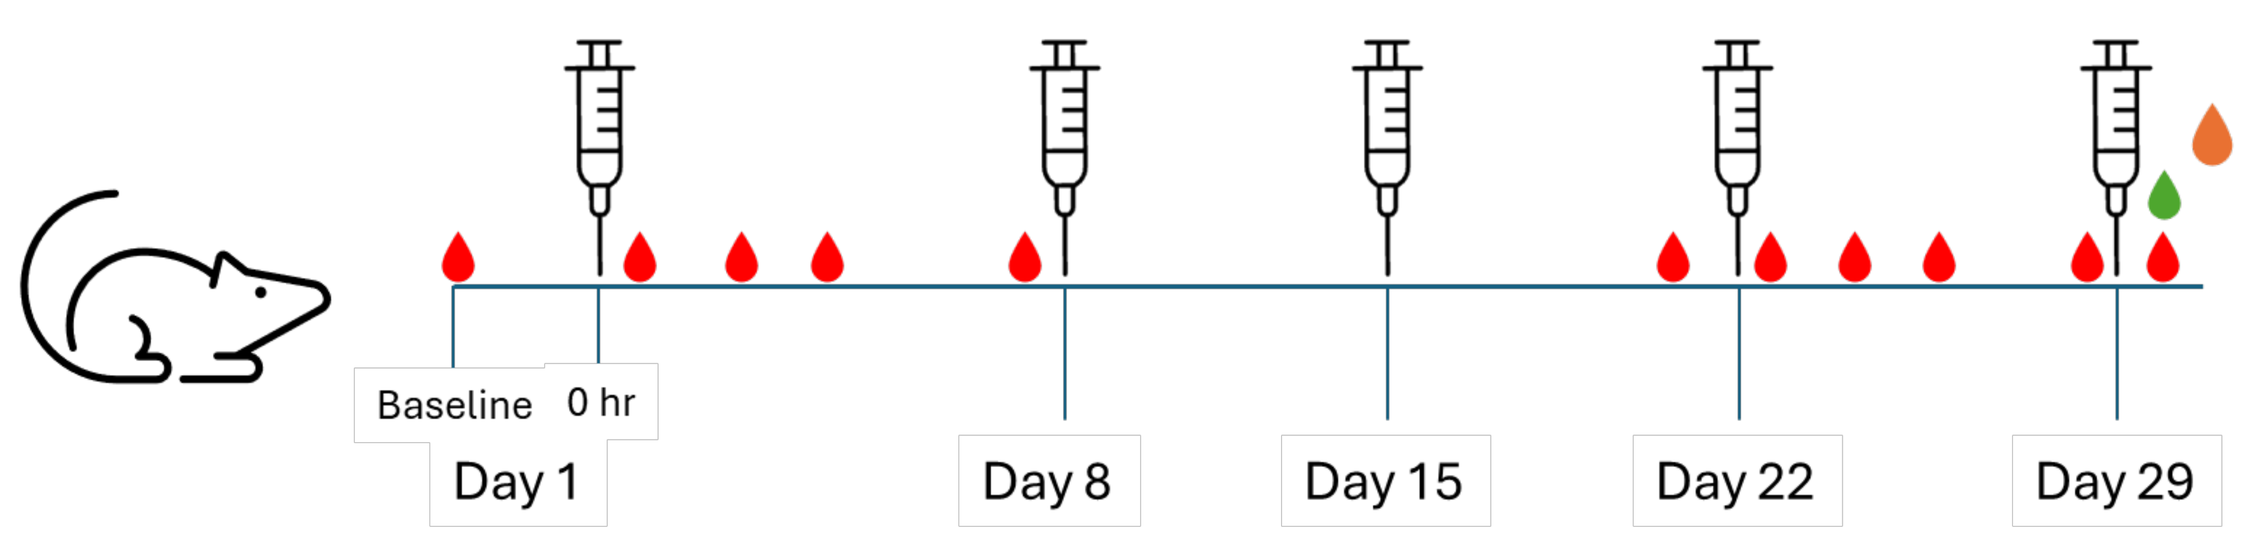
Baseline
0 hr
Day 1
Day 8
Day 15
Day 22
Day 29

Figure 1 Female, human CD34+ cord blood cells from five independent donors were engrafted into NSG-Tg(Hu-IL15) mice and screened for inclusion on study at 12 weeks. Mice with >25% hCD45, >3% hCD3 and >2% hCD56 were accepted for the study. After shipment and acclimation, CD34+ NSG-Tg(Hu-IL15) animals were distributed into toxicology (n=36, 3 donors), immunophenotyping (8, 2 donors) or toxicokinetics (n=8, 2 donors) subsets and treated with five repeated doses (QW) of vehicle or MTX-101 at 15, 50 or 100 mg/kg (~17-18 weeks post engraftment). Following Day 29, the n=4 mice/donor of the toxicology subset were observed for 6 weeks to allow for recovery of any findings.

- Toxicology evaluations (🔸): Day 30 (n=8/donor/dose) and Day 72 (n=4/donor/dose) for clinical and terminal pathology
- PK time points (🔸): Day 1 and Day 22: predose and at 0.5, 48, 96, 168 hr post dose
- Flow cytometry time points (🔸): Day 29: 0.5hr

Similar frequency of human immune cells in peripheral blood at 12 weeks post engraftment in donor cohorts of CD34+ NSG-Tg(Hu-IL15) mice

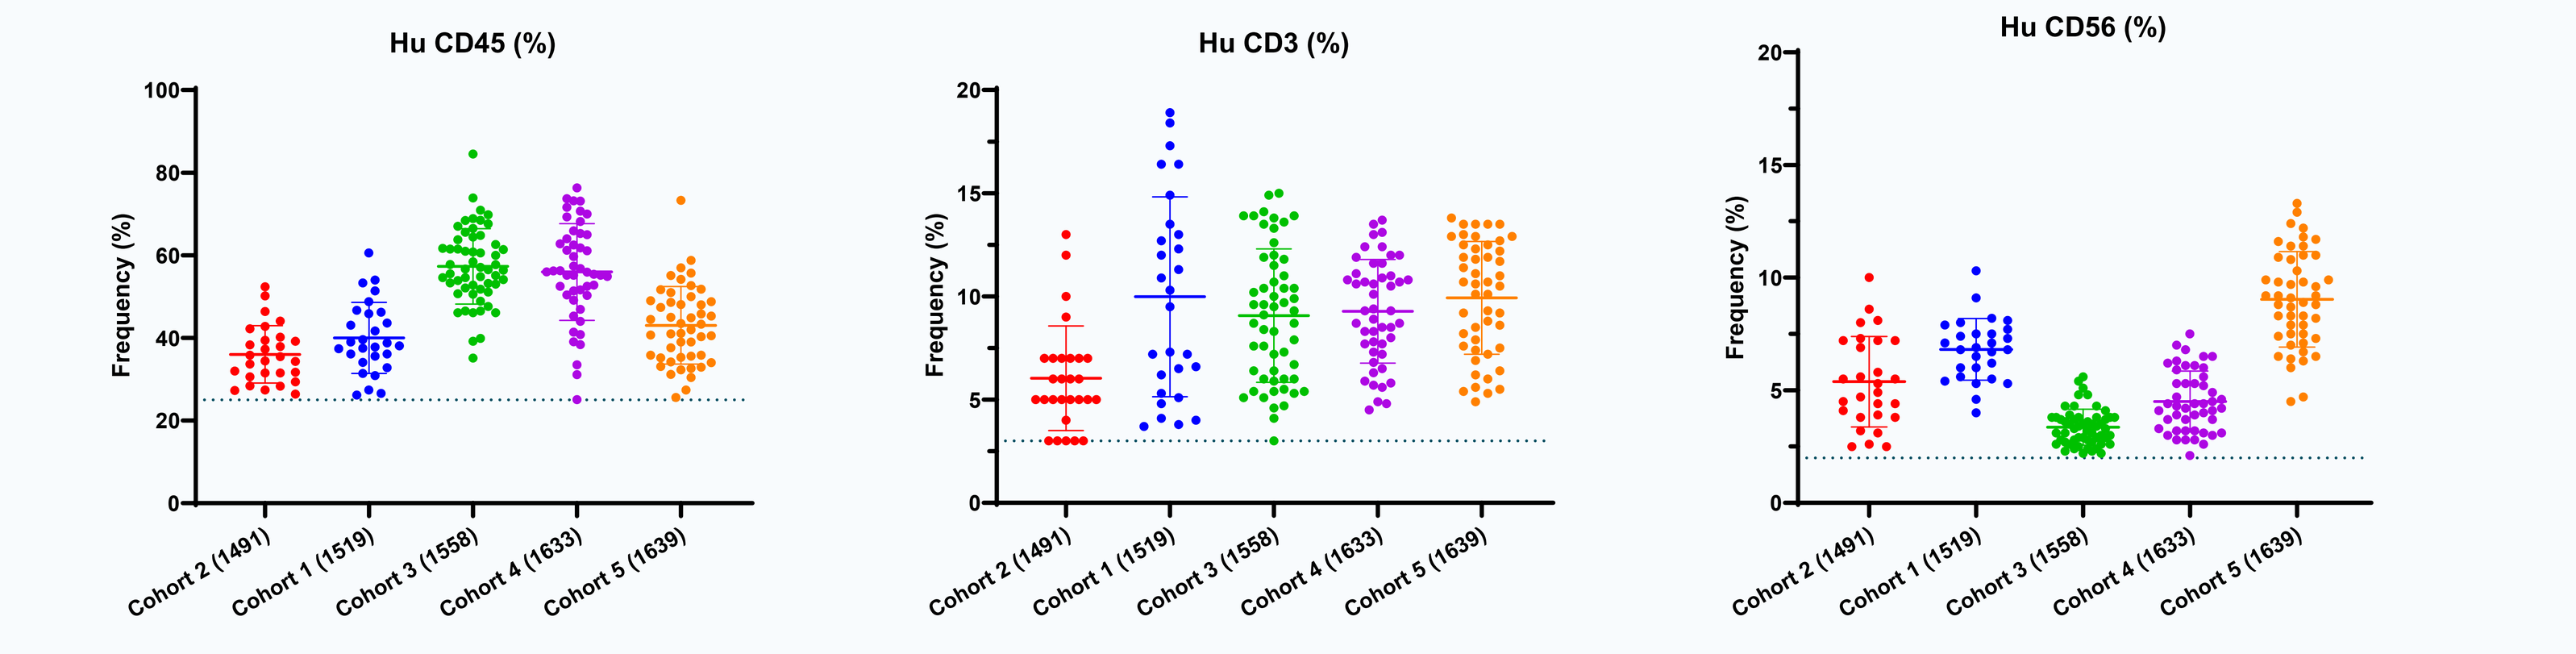
Hu CD45 (%)
Hu CD3 (%)
Hu CD56 (%)
Frequency (%)
Cohort 2 (1491)
Cohort 1 (1519)
Cohort 3 (1558)
Cohort 4 (1633)
Cohort 5 (1639)

Figure 2 The percentages of hCD45, CD3 T, and CD56 NK cells at 12 weeks post engraftment in the mice enrolled for the study. CD34+ cells from five donors were engrafted into NSG-Tg(Hu-IL-15) mice aged 4 weeks, followed by a 12-week post engraftment assessment for human cells expressing hCD45+, CD19+, CD3+, CD33+, and CD56+ at the Jackson Laboratory. Mice exhibiting >25% hCD45, >3% hCD3, and >2% hCD56 were subsequently shipped to the testing facility.

Pharmacologic impact in CD34+ NSG-Tg(HuIL-15) mice following a single dose of MTX-101

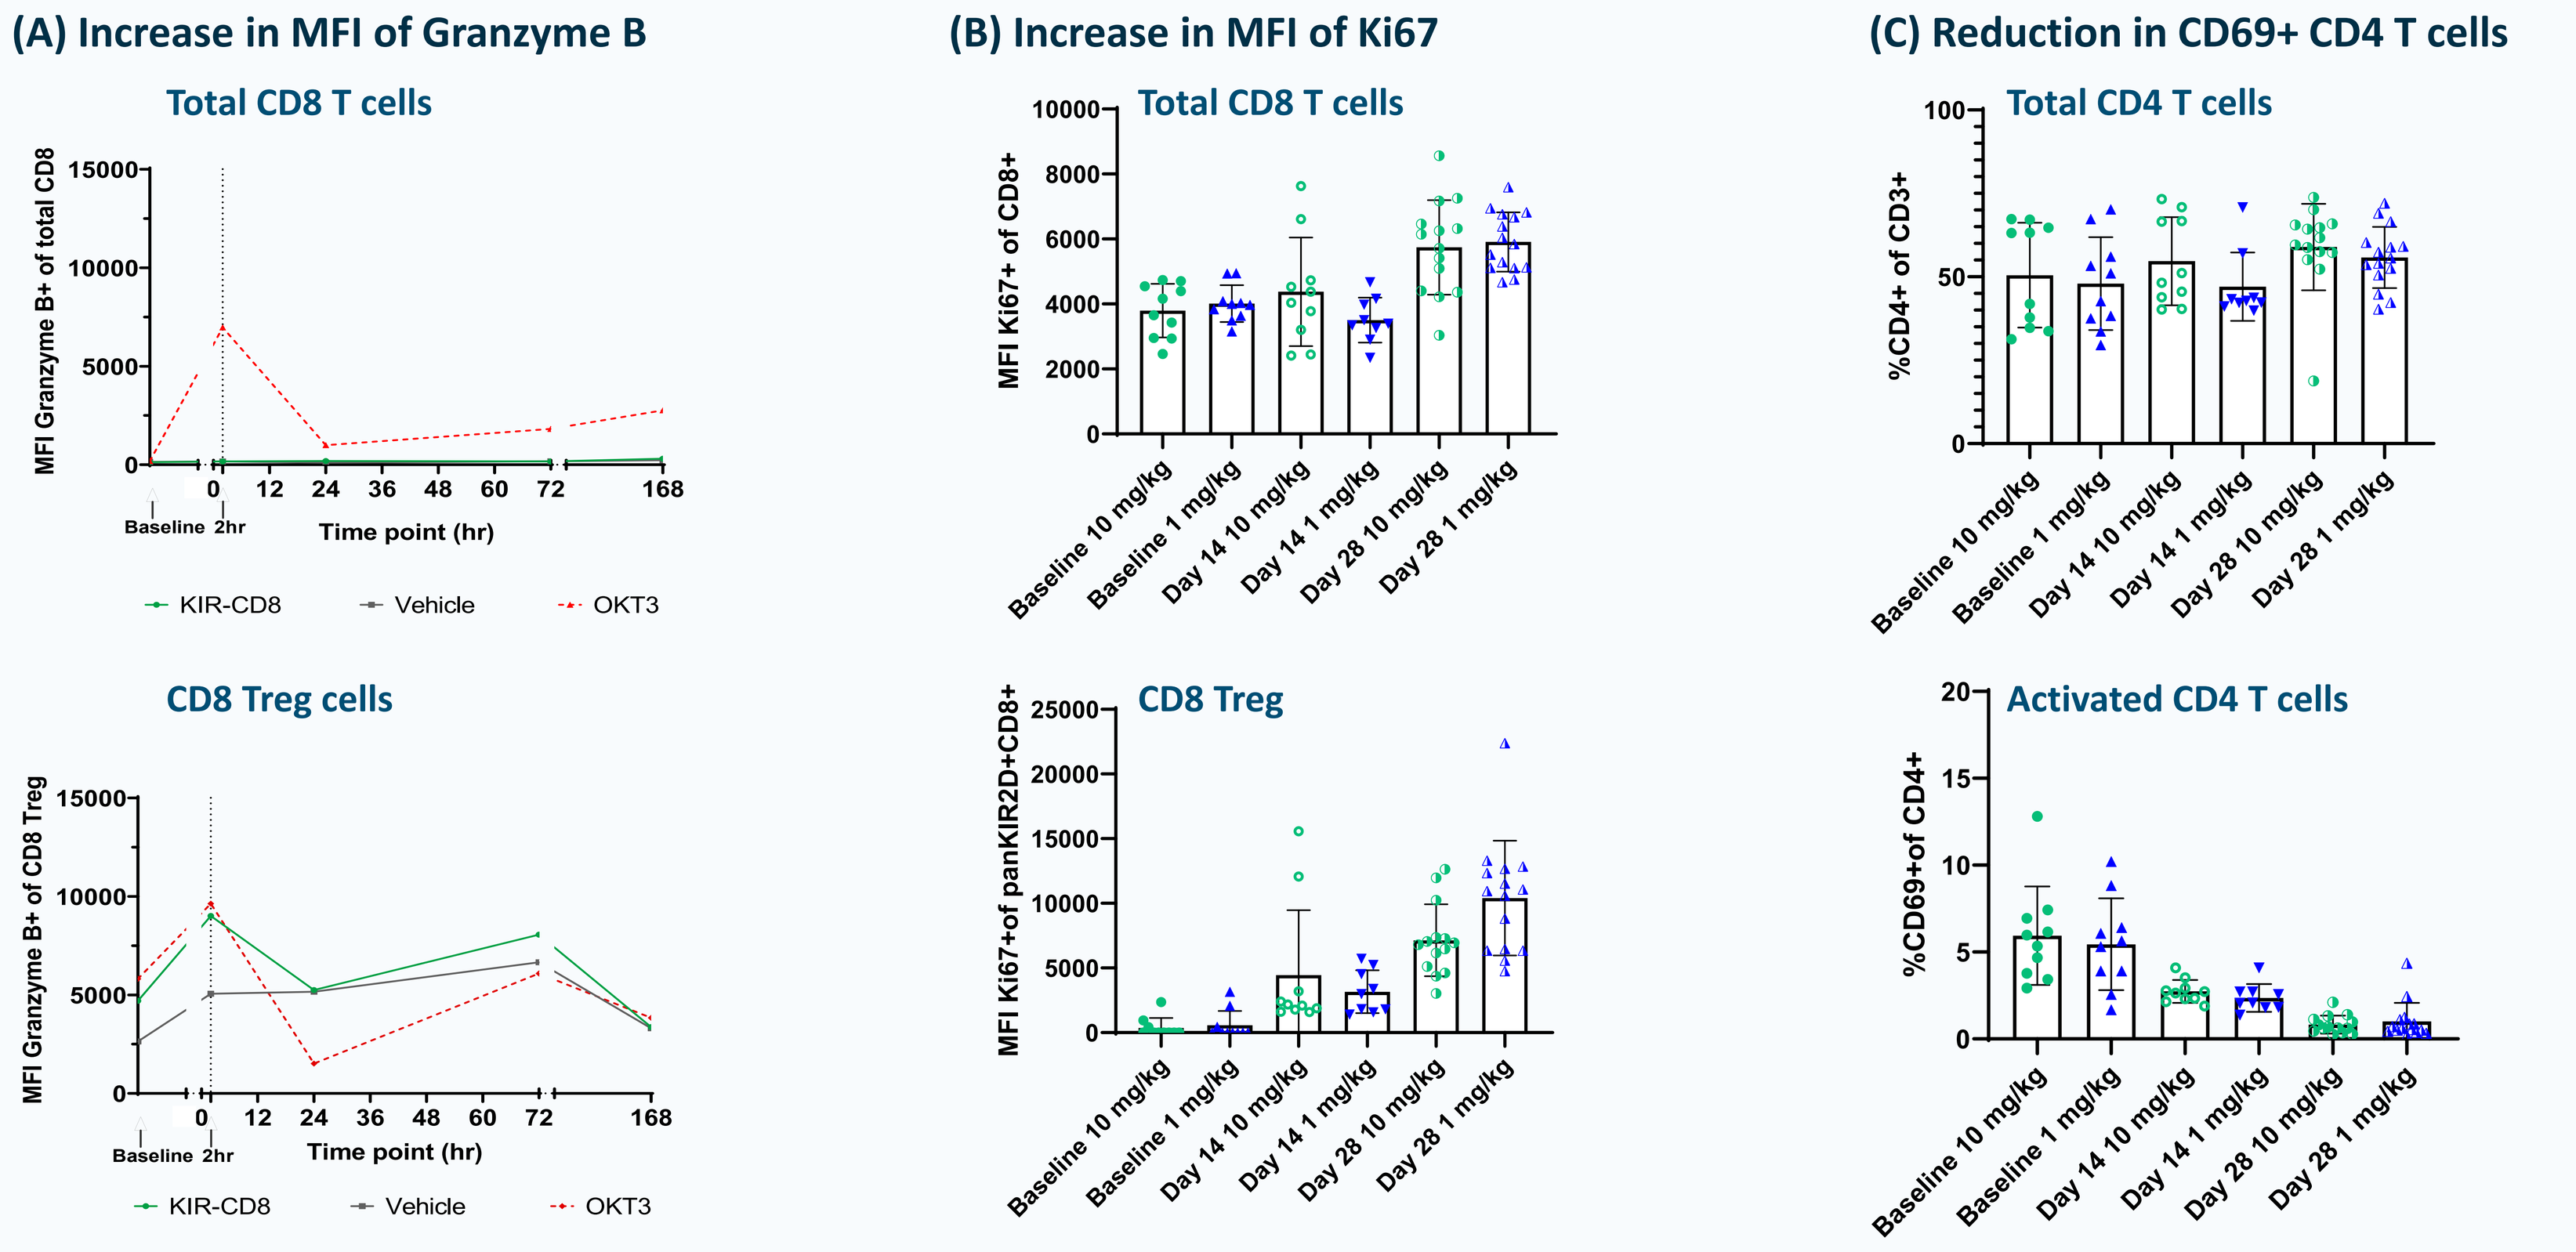
(A) Increase in MFI of Granzyme B
(B) Increase in MFI of Ki67
(C) Reduction in CD69+ CD4 T cells
Total CD8 T cells
Total CD4 T cells
CD8 Treg cells
CD8 Treg
Activated CD4 T cells
MFI Granzyme B+ of total CD8
MFI Granzyme B+ of CD8 Treg
MFI Ki67+ of CD8+
MFI Ki67+of panKIR2D+CD8+
%CD4+ of CD3+
%CD69+of CD4+
Time point (hr)
Baseline 2hr
KIR-CD8
Vehicle
OKT3
Baseline 10 mg/kg
Baseline 1 mg/kg
Day 14 10 mg/kg
Day 14 1 mg/kg
Day 28 10 mg/kg
Day 28 1 mg/kg

Figure 3 (A,B) Top panel, Total CD8 T cells; Bottom panel, KIR+ CD8 Treg cells. (A) Granzyme B was evaluated in an independent study (n=3/dose/donor) with a research stage lot of candidate protein. A transient increase of median fluorescent intensity (MFI) Granzyme B was detected at the 2-hr timepoint post-dose in CD8 Treg cells following CD8 Treg modulator treatment. (B) Following a single dose of MTX-101, an increase in Ki67 MFI Treg cells was observed in CD8 Treg over the course of the study (relative to total CD8 population).
(C) Top panel, total CD4 T cells; bottom panel, CD69+ CD4 T cells. There was an observed decrease in %CD69+ CD4 cells after a single dose of MTX-101, while no change in total CD4+ T cells was detected (Data presented as median, with individual animals)

Frequency of human immune cells in peripheral blood following repeated doses up to 100 mg/kg of MTX-101 in CD34+ NSG-Tg(HuIL15) mice

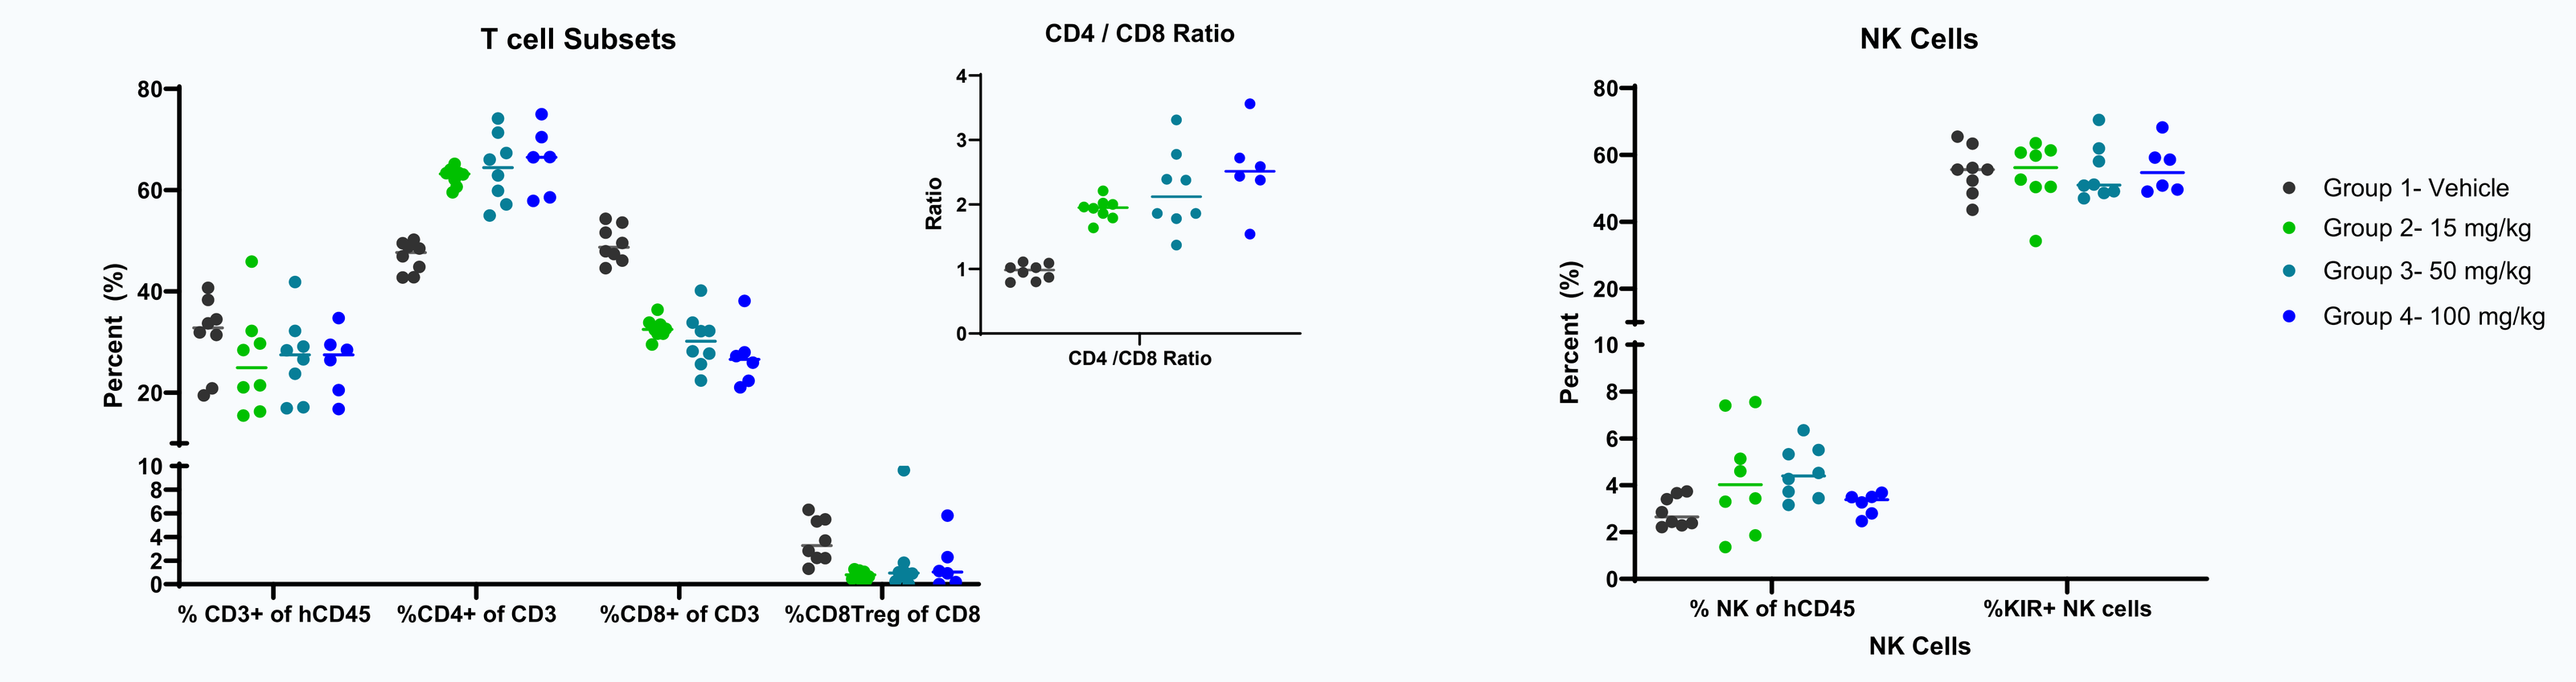
T cell Subsets
CD4 / CD8 Ratio
NK Cells
Percent (%)
Ratio
% CD3+ of hCD45
%CD4+ of CD3
%CD8+ of CD3
%CD8Treg of CD8
CD4 /CD8 Ratio
% NK of hCD45
%KIR+ NK cells
Group 1- Vehicle
Group 2- 15 mg/kg
Group 3- 50 mg/kg
Group 4- 100 mg/kg

Figure 4 Frequency of immune cell subsets, including CD4+ or CD8+ T cell subsets **(A)** and NK cells **(B)**, were assessed by flow cytometry on Day 29 using peripheral blood following 5 weekly IV doses of MTX-101. Skewing of the CD4 to CD8 ratio was not considered adverse and was likely due to a decrease in detection of CD8 T cells due to interference of MTX-101 with anti-CD8 antibodies. A slight reduction of CD8 Treg cells was also observed, potentially due to interference with MTX-101. No MTX-101 related changes were observed in total NK cells or in the KIR+ NK cell subset (B) or CD20+ B cells (not shown).

Profile of MTX-101 concentration in CD34+ NSG-Tg(HuIL-15) mice

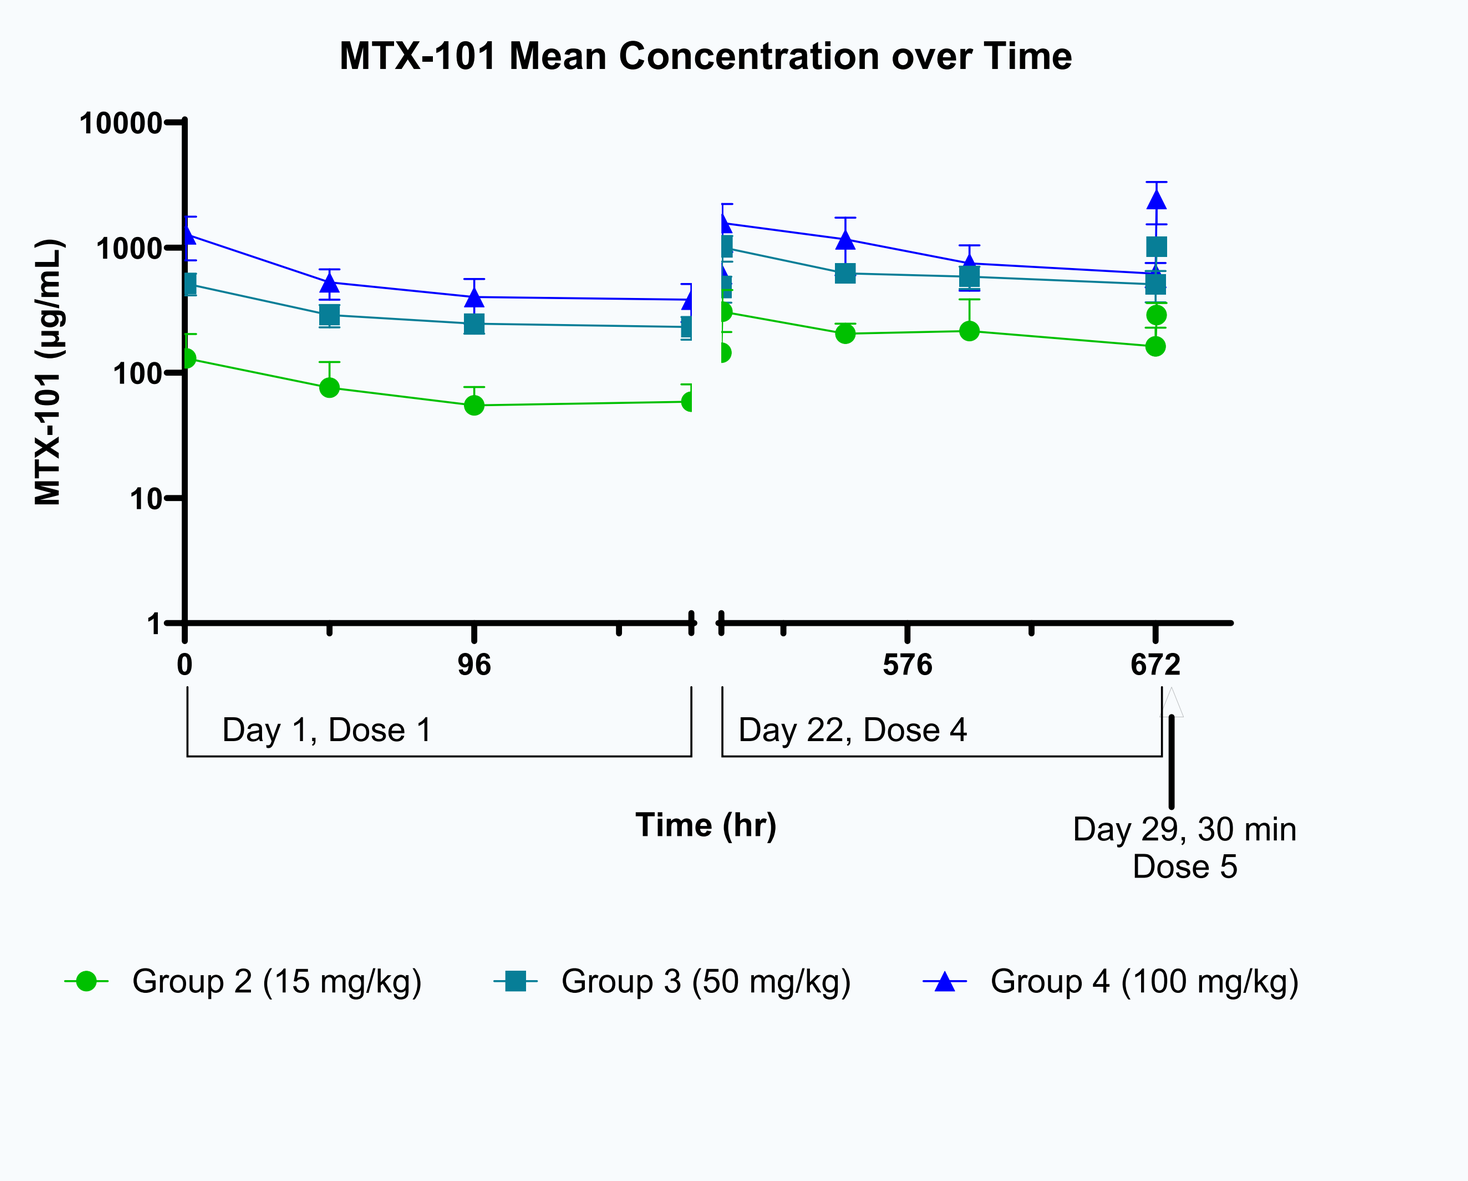
MTX-101 Mean Concentration over Time
MTX-101 (µg/mL)
Time (hr)
Day 1, Dose 1
Day 22, Dose 4
Day 29, 30 min Dose 5
Group 2 (15 mg/kg)
Group 3 (50 mg/kg)
Group 4 (100 mg/kg)

Figure 5 Toxicokinetics (TK) of MTX-101 demonstrated dose proportional exposure with 1.5 to 3-fold accumulation following repeated doses (QW) at 15, 50 or 100 mg/kg (serial microscale samples). Data are presented as mean +/- SD for each dose derived from two donors (n=4 animals / donor/dose level).
No evidence of immunogenicity (ADA) was observed.

Binding to cells expressing both targets of MTX-101 in CD34+ NSG-Tg(HuIL-15) mice

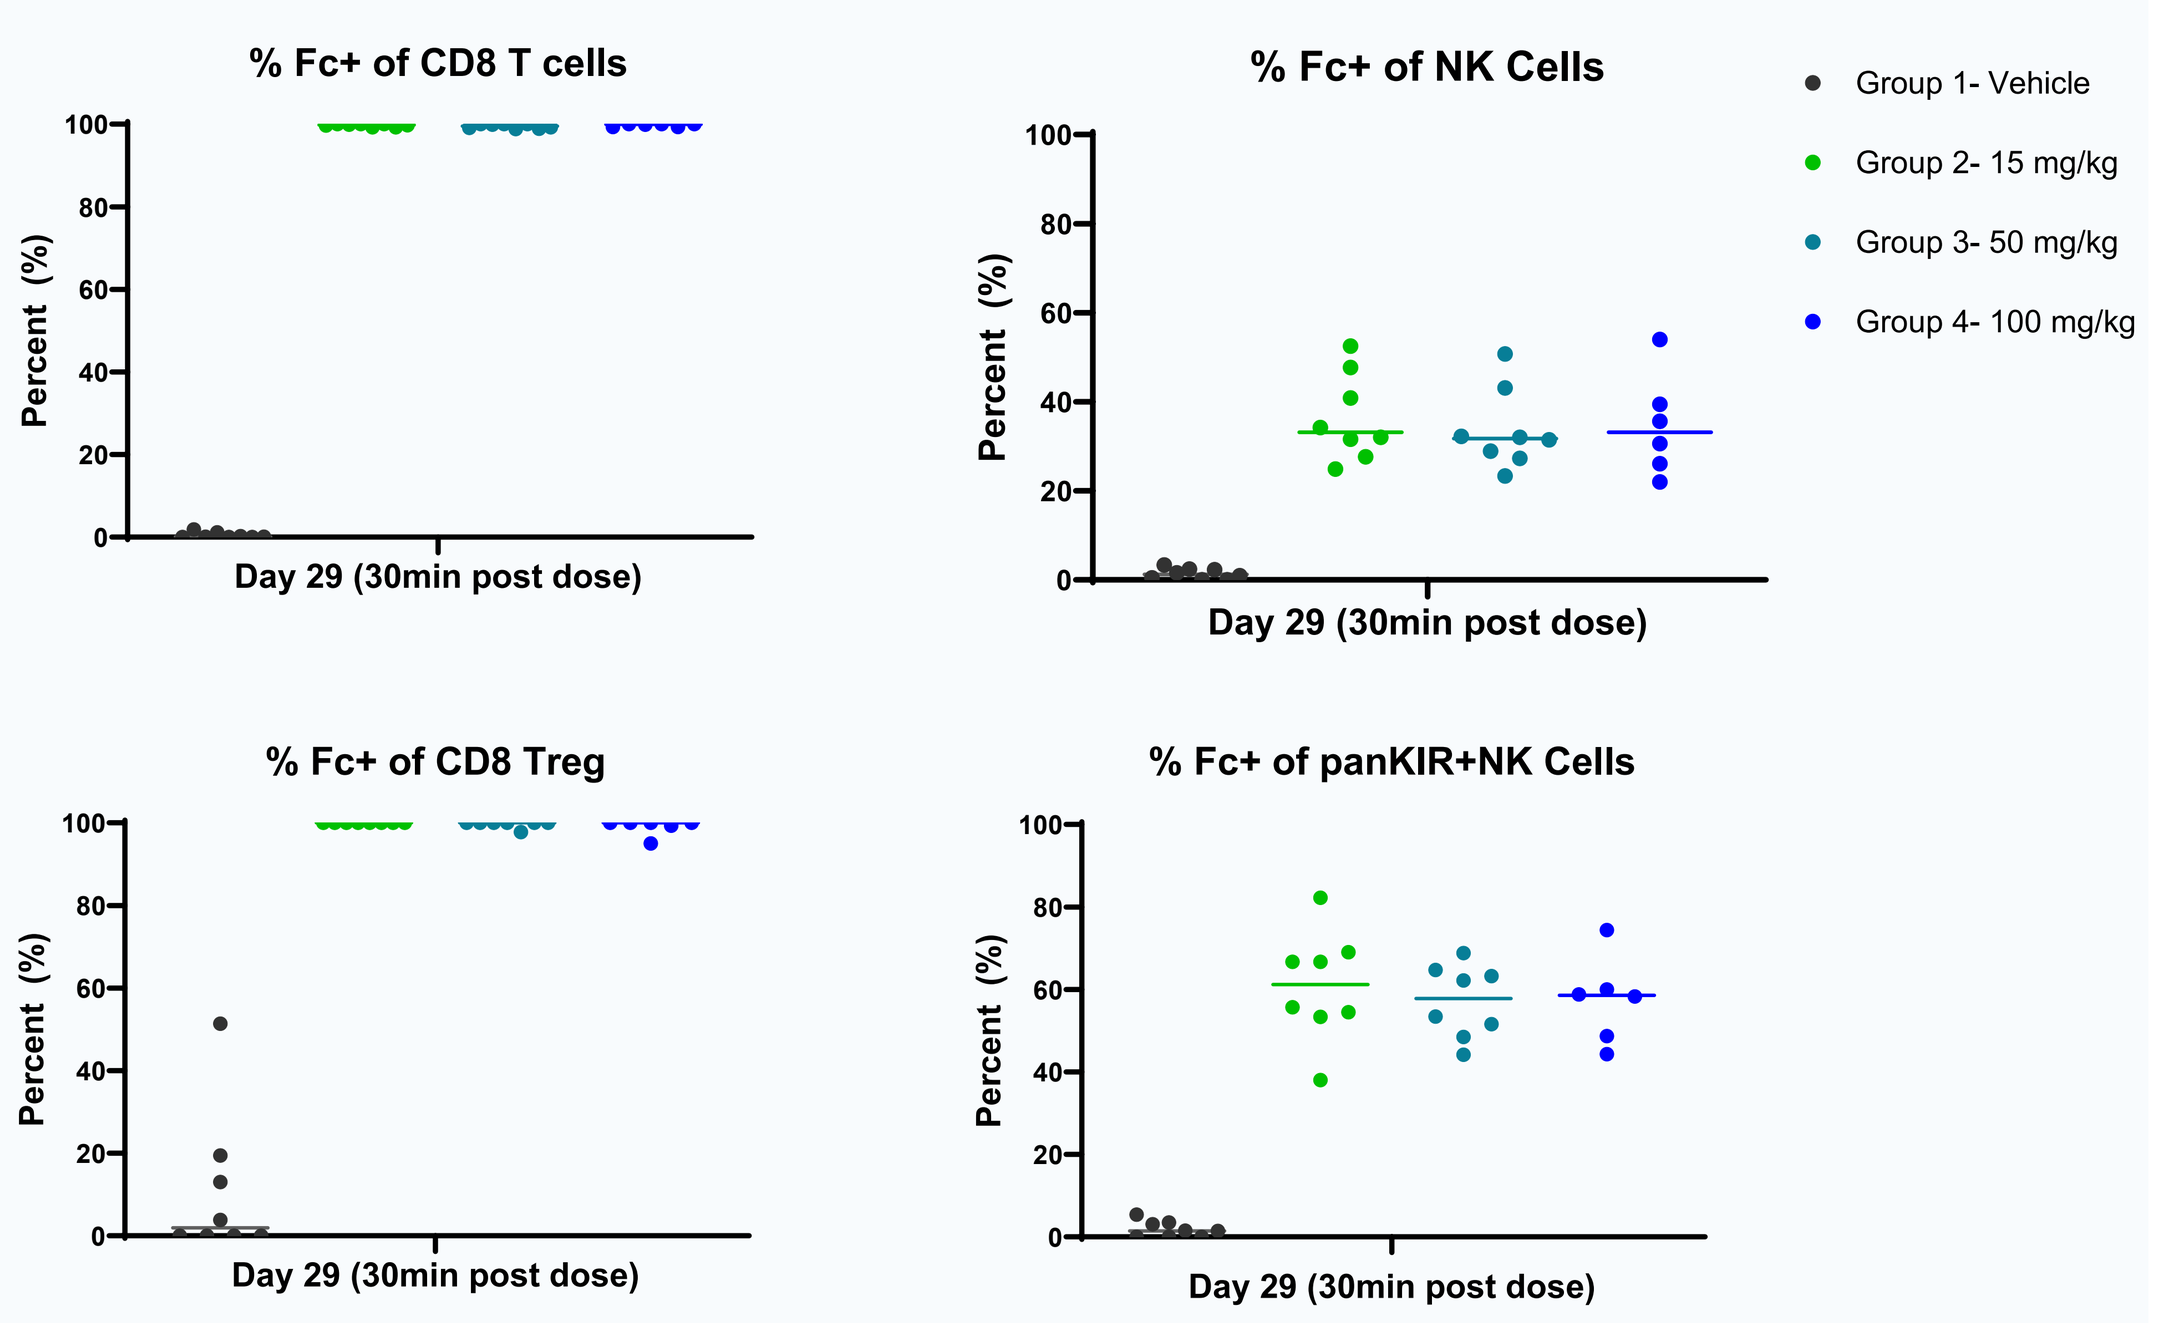
% Fc+ of CD8 T cells
% Fc+ of NK Cells
% Fc+ of CD8 Treg
% Fc+ of panKIR+NK Cells
Percent (%)
Day 29 (30min post dose)
Group 1- Vehicle
Group 2- 15 mg/kg
Group 3- 50 mg/kg
Group 4- 100 mg/kg

Figure 6 Binding of MTX-101 to total CD8, CD8 Treg, NK, and panKIR2D+NK cells via Fc detection were found on cells in blood and in spleen. As expected, no binding was observed to CD4 T cells either in blood or spleen (not shown). Data are presented for individual animals. Uniform high binding was detectable on Day 29 (30 min post dose) to Total T cells and to CD8 Treg cells while more variable binding was detected on NK cells.

No adverse changes in toxicity evaluations following repeated doses of MTX-101 up to 100 mg/kg

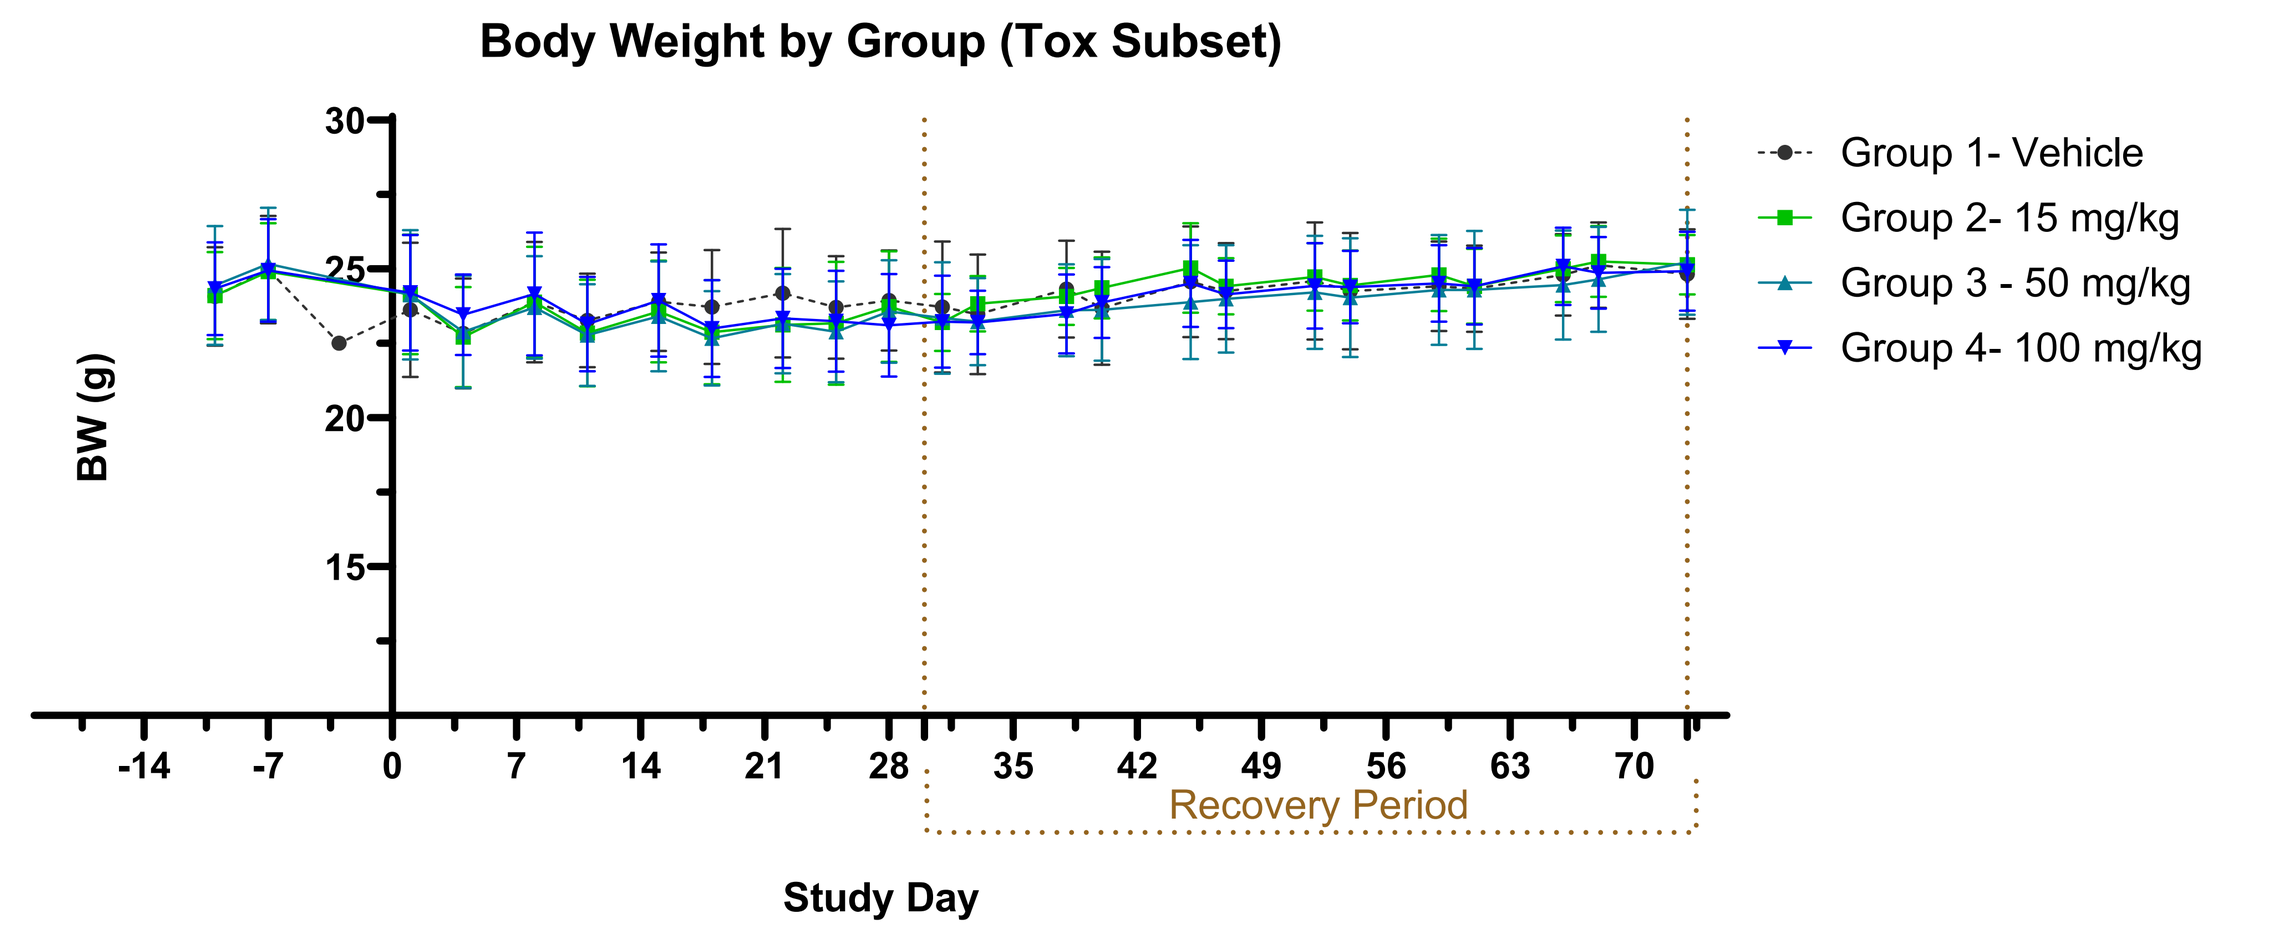
Body Weight by Group (Tox Subset)
BW (g)
Study Day
Recovery Period
Group 1- Vehicle
Group 2- 15 mg/kg
Group 3 - 50 mg/kg
Group 4- 100 mg/kg

Figure 7 Body weight measurements were taken twice per week during main study and recovery periods following repeated doses at 100, 50 or 15 mg/kg in CD34+ NSG-Tg(Hu-IL15) mice. No differences in mean group body weights were observed compared to vehicle treated group. No findings were observed in clinical pathology or anatomic pathology at terminal time points.

Minimal risk of pro-inflammatory cytokines with MTX-101 in Human Cytokine Release Assay

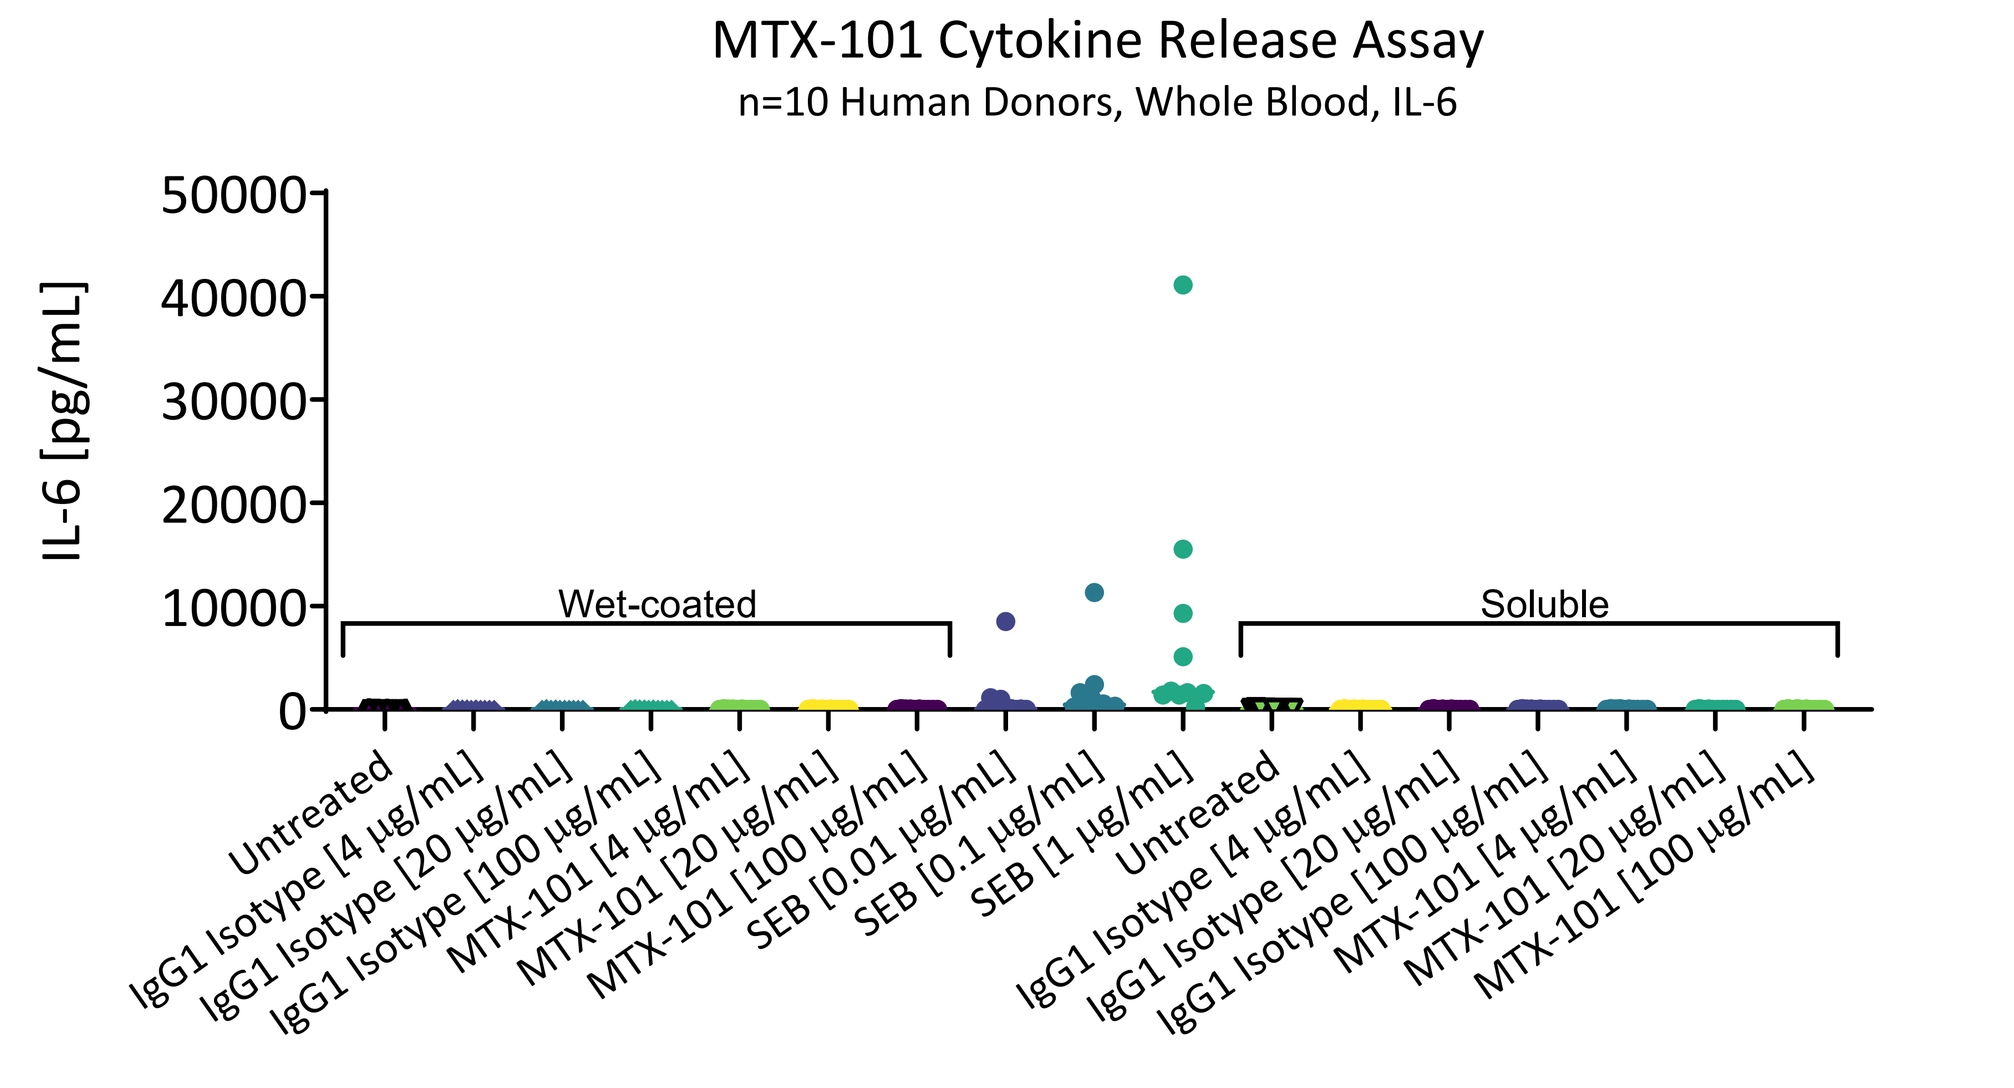
MTX-101 Cytokine Release Assay
n=10 Human Donors, Whole Blood, IL-6
IL-6 [pg/mL]
Wet-coated
Soluble
Untreated
IgG1 Isotype [4 µg/mL]
IgG1 Isotype [20 µg/mL]
IgG1 Isotype [100 µg/mL]
MTX-101 [4 µg/mL]
MTX-101 [20 µg/mL]
MTX-101 [100 µg/mL]
SEB [0.01 µg/mL]
SEB [0.1 µg/mL]
SEB [1 µg/mL]

Figure 8 MTX-101 was evaluated in a cytokine release assessment with dose titration up to 100 µg/mL (~800 nM) using soluble or wet-coated conditions. Ten donors were evaluated in each condition using whole blood. MTX-101 did not cause increased expression of pro-inflammatory serum cytokines (including IL-12, IL-8, IL-10, TNFα or IFNγ). Data (IL-6) are presented for individuals. (Staphylococcus enterotoxin B (SEB) was used as a positive control)

Results

MTX-101 bound to its targets and was well-tolerated up to the highest dose tested (100 mg/kg) and did not adversely impact prevalence or increase activation of NK cells, CD4 T cells or CD8 T cells or detectable pro-inflammatory serum cytokines.

- MTX-101 binding to expected cell types was measurable following single or repeated doses in CD34+ NSG-Tg(Hu-IL15) mice, including CD8 Treg.
- Following repeated doses up to 100 mg/kg of MTX-101, no adverse safety findings were observed, with concentration of MTX-101 indicating high exposure in humanized mice. The highest dose tested is considered NOAEL. The half-life is consistent across species with a $T_{1/2}$ of about 11.5 days
- No elevated cytokine levels have been detected suggesting MTX-101 has low risk of cytokine release, as demonstrated in vivo in CD34+ NSG-Tg(Hu-IL15) mice, as well as in vitro with whole blood or PBMC from multiple human donors.
- Following a single dose of MTX-101, an increase in proliferation and Granzyme B expression were observed, suggesting an impact on CD8 Treg cytolytic capacity.
- Following a single dose of MTX-101, a decrease of activated CD4 T cells was observed at late timepoints in support of the postulated MoA of MTX-101.

Conclusion

MTX-101 is a promising therapeutic approach to address an underlying cause of autoimmune disease via enhancement of CD8 Treg function. Non-clinical safety studies using CD34+ NSG-Tg(HuIL15) mice support safe administration of MTX-101 and clinical entry into a Ph1a/b healthy adult and patient study.

Contact:
Follow up questions can be directed to Kristine Swiderek, Chief Scientific Officer, Mozart Therapeutics
kswiderek@mozart-tx.com or by visiting the website at https://www.mozart-tx.com/

References:

Li, et al. KIR + CD8 + T cells suppress pathogenic T cells and are active in autoimmune diseases and COVID-19. *Science.* 2022, 376, eabi9591.
Abeynaike, et al. Human Hematopoietic Stem Cell Engrafted IL-15 Transgenic NSG Mice Support Robust NK Cell Responses and Sustained HIV-1 Infection. Viruses 2023, 15, 365.