A KIR x CD8 targeting bispecific modulator enhances regulatory CD8 T cell functions, and reduces inflammation in models of autoimmune disease

Orchestrating The Immune System

Gardell, JL; Boster, D; Bowser, J; Fasnacht, R; Fitzpatrick, K; Hermansky, G; Julien, S; Maurer, M; McMahan, C; Meengs, B; Patton, D; Pham, M; Therriault, J; Swiderek, KM; and Crane, CA.

Introduction

We have characterized a subset of CD8 T cells (CD8 Treg) with immunosuppressive characteristics in inflammatory disease settings. CD8 Treg activation results in their oligoclonal expansion and elimination of pathogenic CD4 T cells. Here we describe a bispecific CD8 Treg Modulator that binds KIR2DL1/2/3 and CD8 to activate autoimmune patient CD8 Tregs resulting in the cytolytic elimination of pathogenic CD4 T cell in vitro, ex vivo, and in vivo.

CD8 Treg Modulator is targeting a Novel Network to Restore Immune Balance in Autoimmune Disease

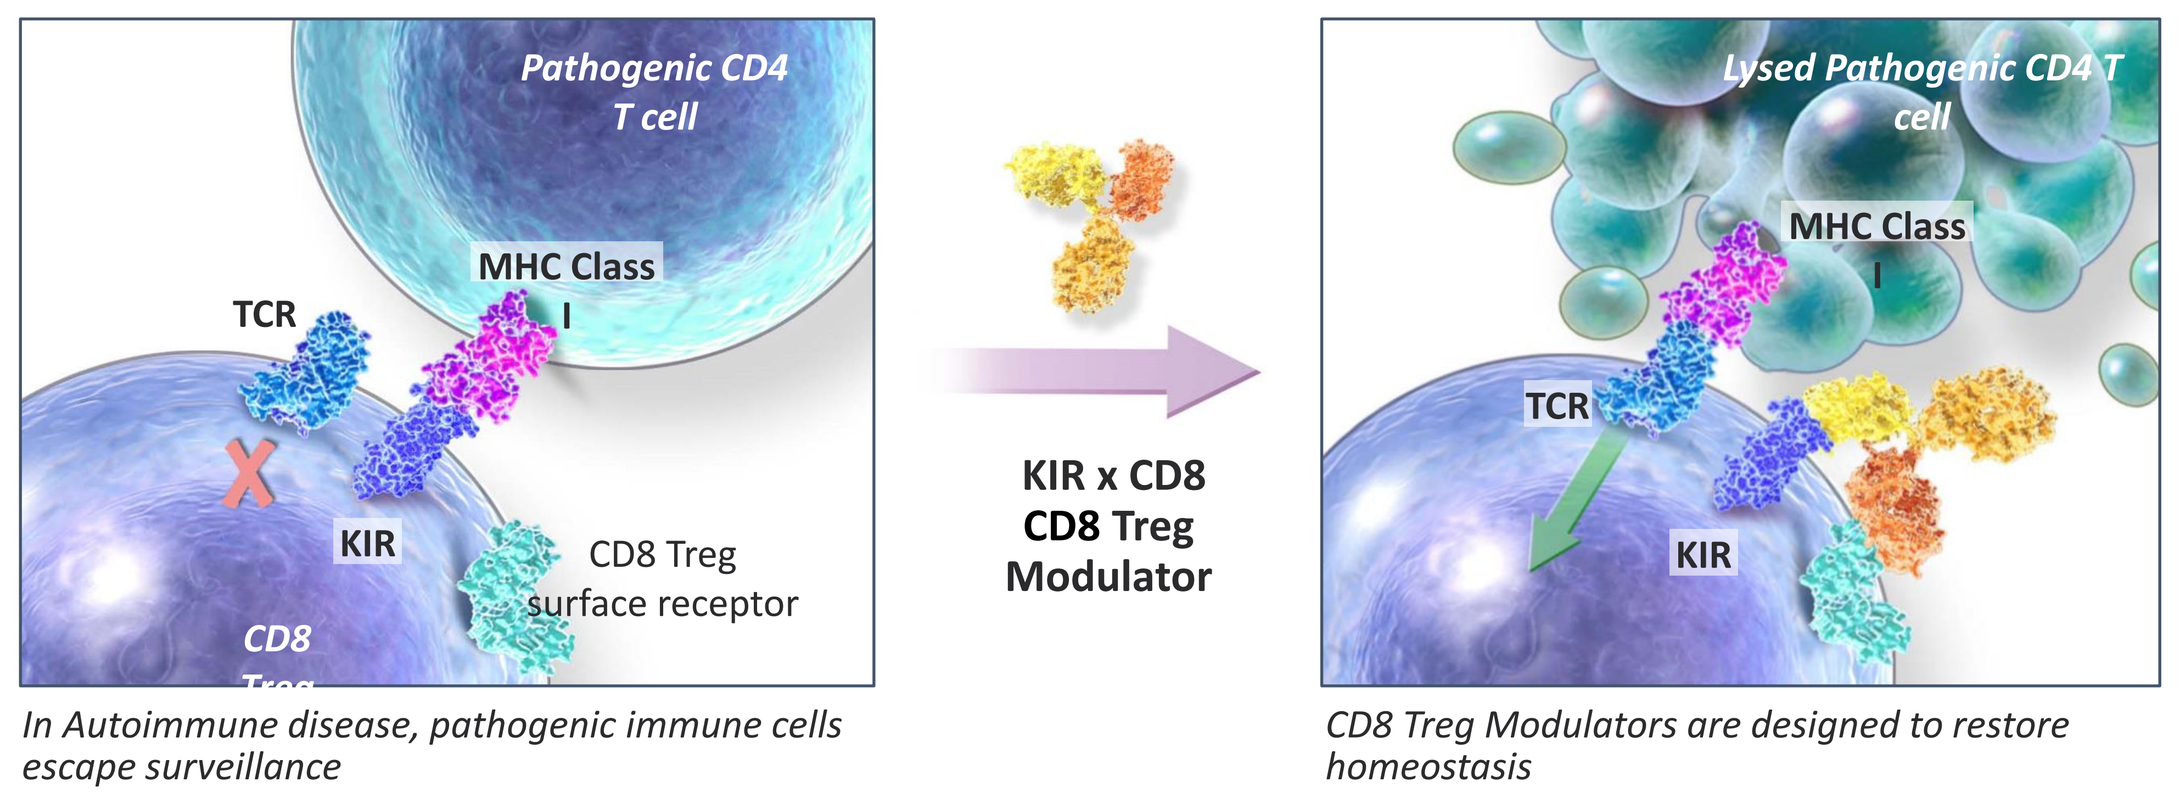
Pathogenic CD4 T cell
MHC Class I
TCR
KIR
CD8 Treg surface receptor
CD8 Treg
KIR x CD8 CD8 Treg Modulator
Lysed Pathogenic CD4 T cell
MHC Class I
TCR
KIR

In Autoimmune disease, pathogenic immune cells escape surveillance

CD8 Treg Modulators are designed to restore homeostasis

Approach seeks to increase cytolytic activity of dysfunctional CD8 Treg

Methods

We tested target binding of a novel bispecific CD8 Treg Modulator by Octet, and to immune cell subsets in healthy donor PBMC by flow cytometry.

The functional impact of the CD8 Treg Modulator was evaluated in healthy, Celiac, and Crohn's PBMC by CD8 Treg activation, cytotoxicity assays, and supernatant analysis in vitro, with Celiac patient derived biopsies ex vivo, and in acute GVHD studies using healthy donor human PBMC.

Simultaneous antigen binding and selective targeting of CD8 Treg Modulator to dual target expressing CD8 T cells

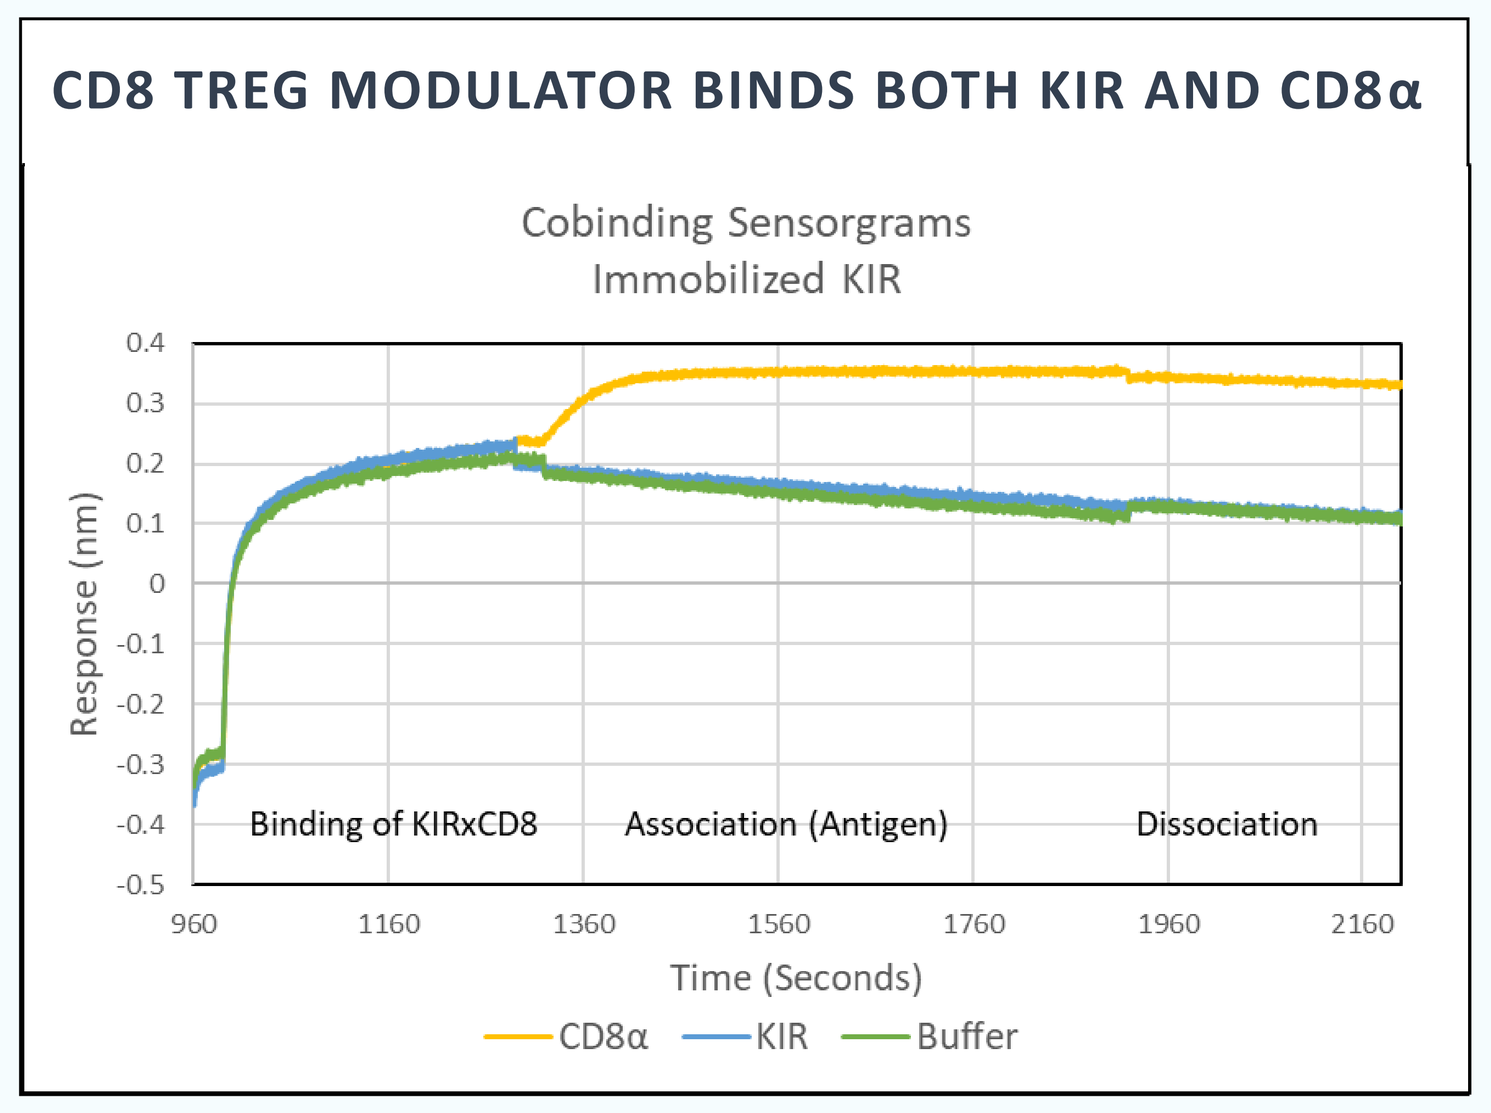
CD8 TREG MODULATOR BINDS BOTH KIR AND CD8α
Cobinding Sensorgrams
Immobilized KIR
Response (nm)
Binding of KIRxCD8
Association (Antigen)
Dissociation
Time (Seconds)
CD8α
KIR
Buffer

Figure 1. Binding of CD8 Treg Modulator to target recombinant proteins as detected by Octet. The CD8 Treg Modulator was captured onto immobilized KIR followed by CD8α binding.

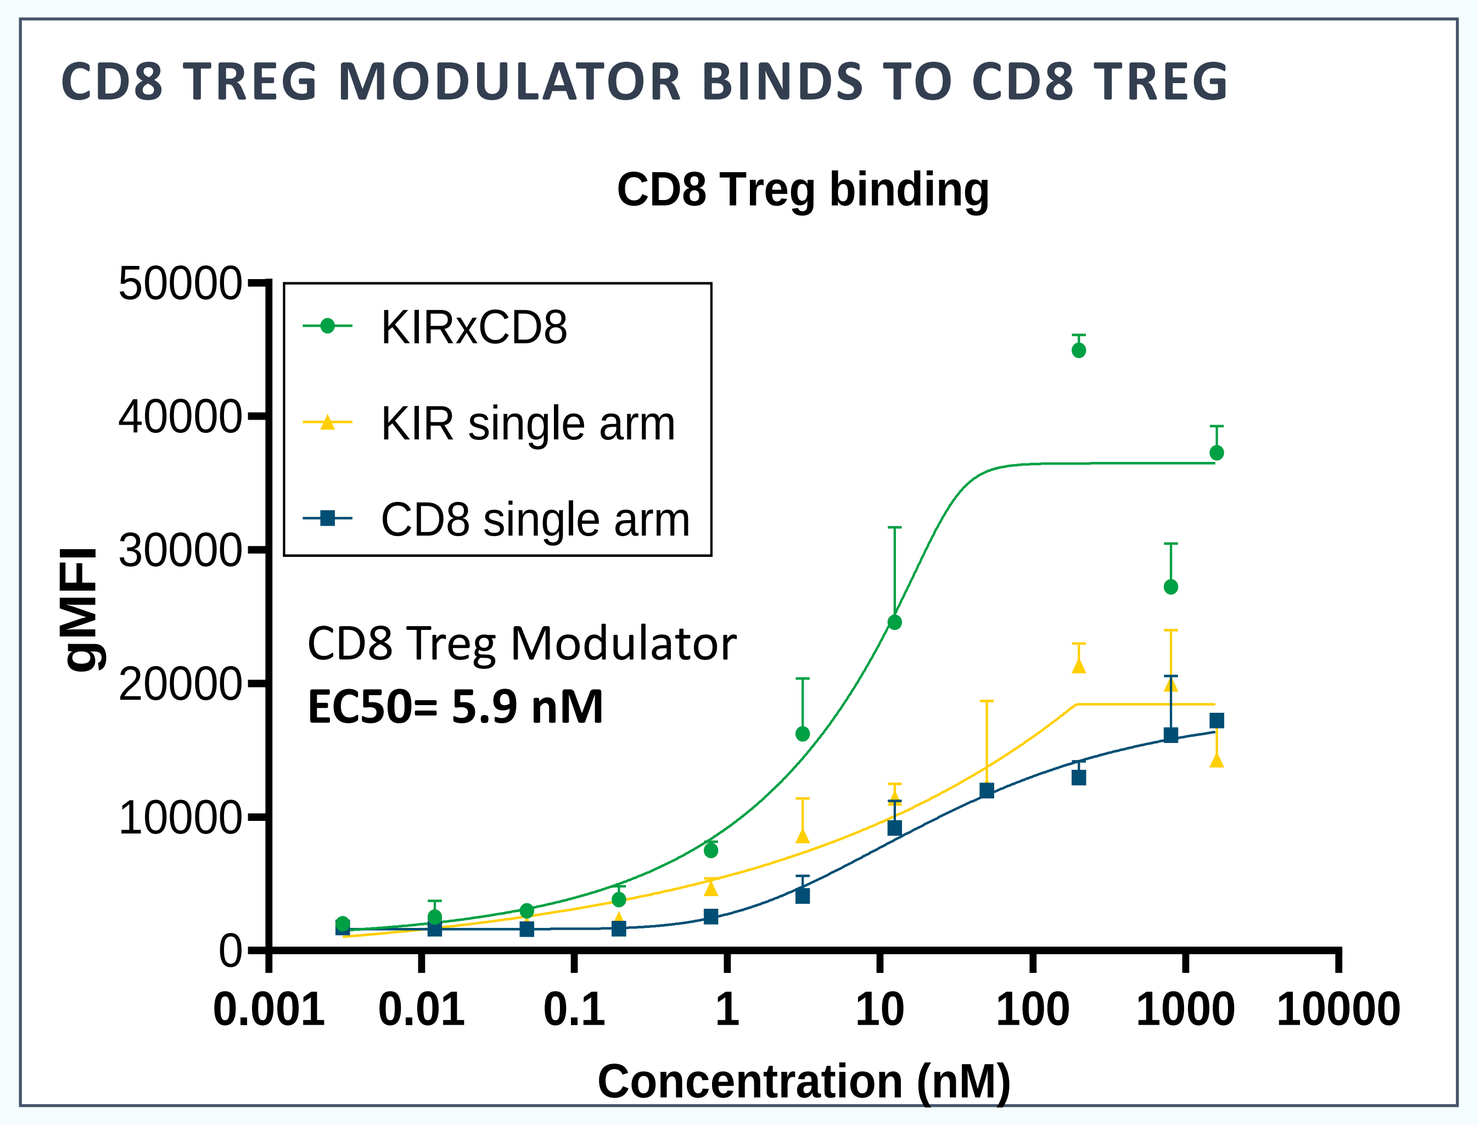
CD8 TREG MODULATOR BINDS TO CD8 TREG
CD8 Treg binding
KIRxCD8
KIR single arm
CD8 single arm
CD8 Treg Modulator
EC50= 5.9 nM
gMFI
Concentration (nM)

Figure 2. Mixed PBMC from a healthy donor were incubated with the bispecific CD8 Treg Modulator or each of the single arm control molecules. Binding of the antibodies to different cell populations was detected using an anti-human IgG1 Fc secondary antibody. A nonblocking KIR antibody was used to selectively gate on the KIR+ CD8 Tregs within the total PBMC.

CD8 Treg Modulator preferentially binds to CD8 Tregs inducing their activation with limited NK cell binding and no activation

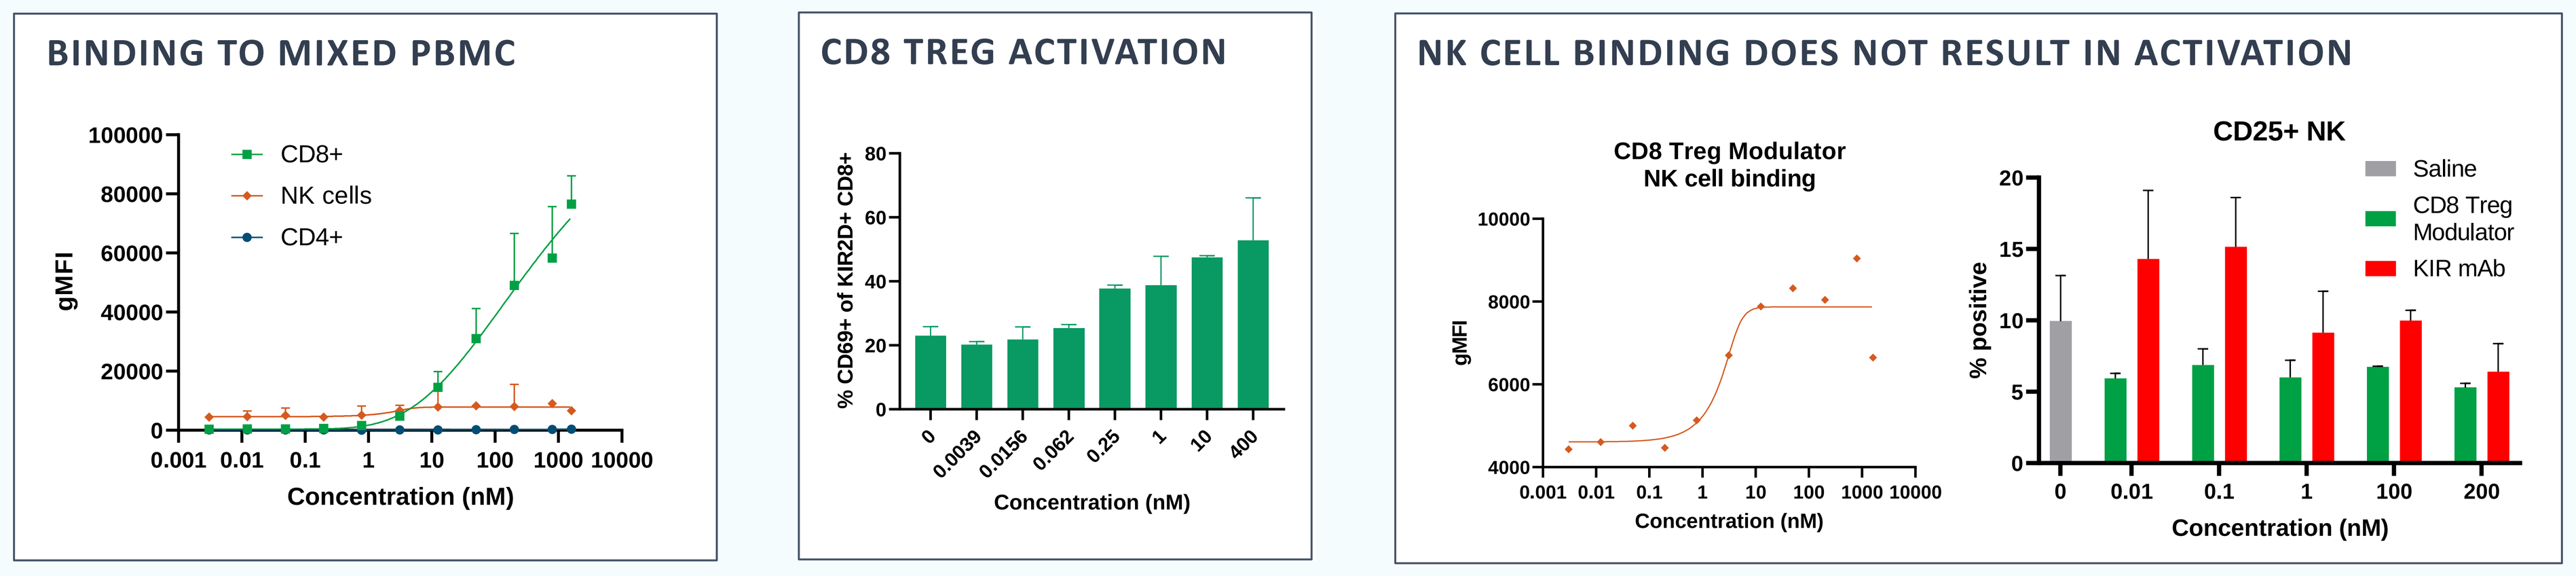
BINDING TO MIXED PBMC
CD8+
NK cells
CD4+
gMFI
Concentration (nM)
CD8 TREG ACTIVATION
% CD69+ of KIR2D+ CD8+
Concentration (nM)
NK CELL BINDING DOES NOT RESULT IN ACTIVATION
CD8 Treg Modulator
NK cell binding
CD25+ NK
Saline
CD8 Treg Modulator
KIR mAb
% positive
Concentration (nM)

Figure 3. PBMC from a healthy donor were incubated with KIRxCD8 bispecific CD8 Treg Modulator and binding of the antibodies to different cell populations was detected using an anti-human IgG1 Fc secondary antibody. Healthy donor CD8 Tregs were incubated with the KIRxCD8 bispecific CD8 Treg Modulator for 3 days and then CD8 Treg activation was determined by gating on CD8 Treg with non-blocking KIR2D and CD69 antibodies. KIRxCD8 bispecific CD8 Treg Modulator binding to NK cells was evaluated over a range of doses, and NK cell activation measured following incubation with KIRxCD8 bispecific CD8 Treg Modulator or KIR single arm control antibody.

CD8 Treg Modulator enhances the elimination of Ag-responsive CD4s in Celiac and Crohn's PBMC

Celiac PBMC

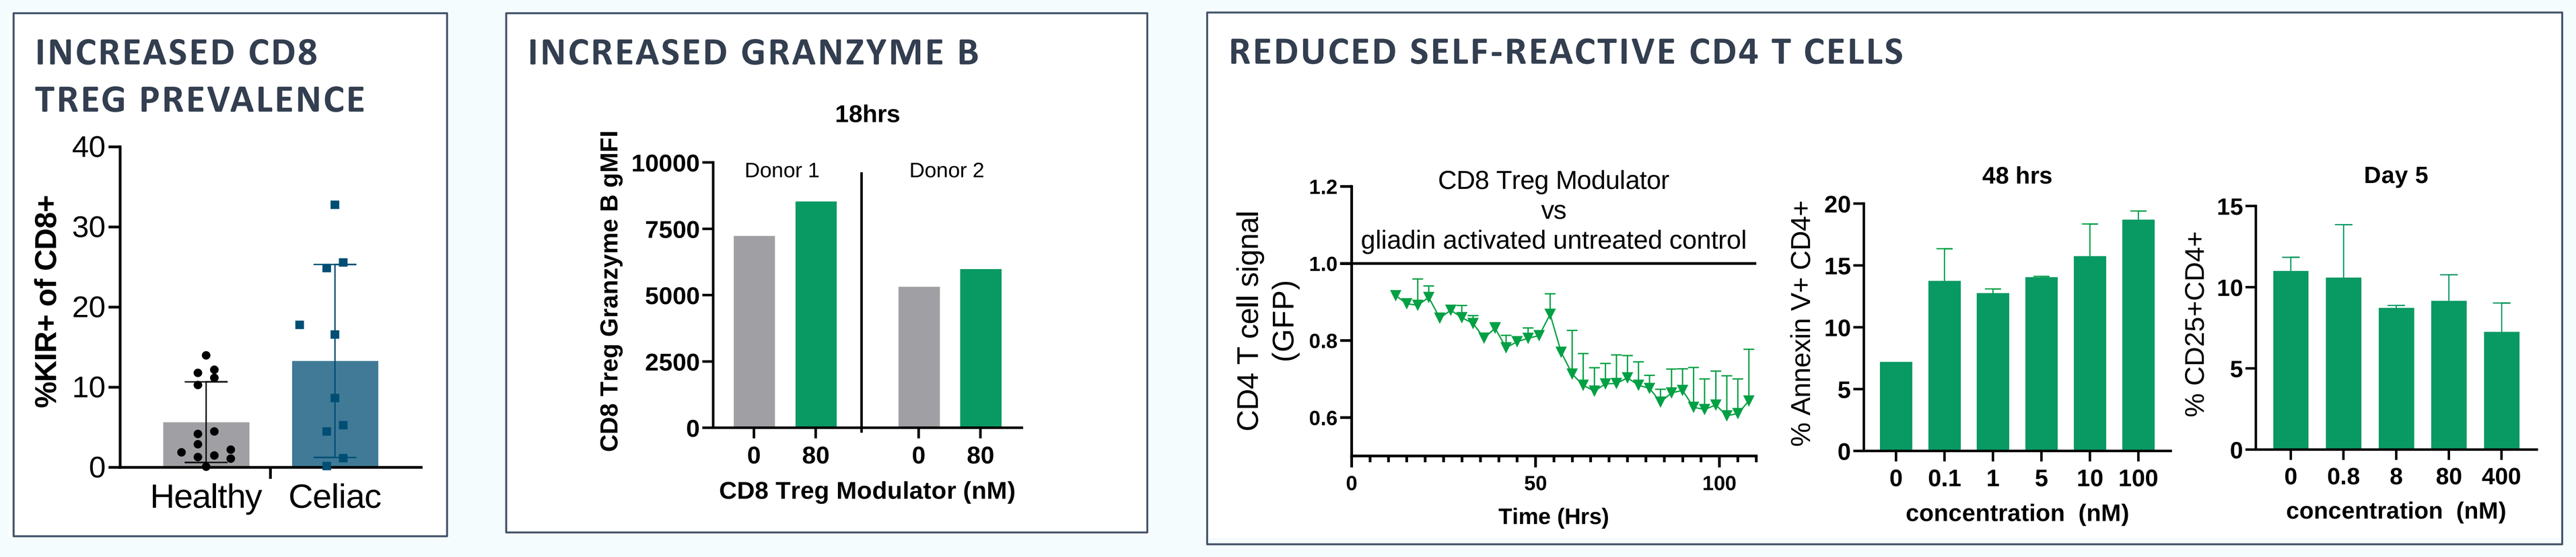
INCREASED CD8 TREG PREVALENCE
%KIR+ of CD8+
Healthy
Celiac
INCREASED GRANZYME B
18hrs
Donor 1
Donor 2
CD8 Treg Granzyme B gMFI
CD8 Treg Modulator (nM)
REDUCED SELF-REACTIVE CD4 T CELLS
CD8 Treg Modulator vs gliadin activated untreated control
CD4 T cell signal (GFP)
Time (Hrs)
48 hrs
% Annexin V+ CD4+
Day 5
% CD25+CD4+
concentration (nM)

Crohn's PBMC

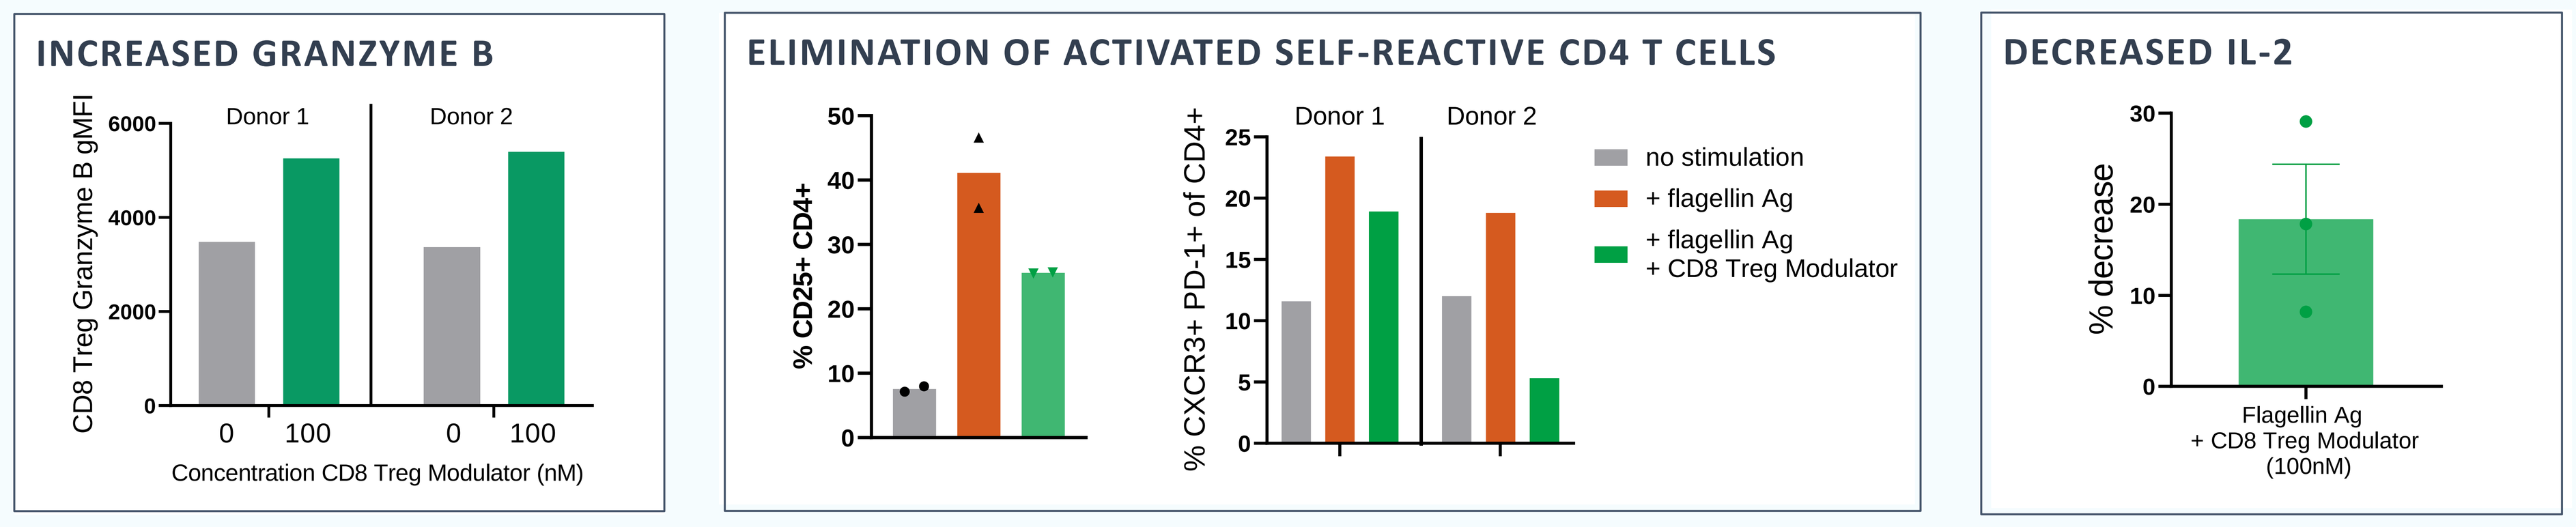
INCREASED GRANZYME B
Donor 1
Donor 2
CD8 Treg Granzyme B gMFI
Concentration CD8 Treg Modulator (nM)
ELIMINATION OF ACTIVATED SELF-REACTIVE CD4 T CELLS
% CD25+ CD4+
% CXCR3+ PD-1+ of CD4+
no stimulation
+ flagellin Ag
+ flagellin Ag + CD8 Treg Modulator
DECREASED IL-2
% decrease
Flagellin Ag + CD8 Treg Modulator (100nM)

Figure 4. CD8 Tregs detected in either healthy or Celiac PBMC. Increase in Granzyme B content in Celiac donor CD8 Tregs following–co-culture with gliadin-stimulated CD4 T cells and 80nM KIRxCD8 bispecific CD8 Treg Modulator for 18 hours. CD8 Treg elimination of celiac-responsive CD4 T cells as detected by a decrease in GFP signal over time by Incucyte. Elimination of gliadin-responsive Celiac donor CD4 T cells as detected by increase in Annexin V+ CD4 T cells at 48hrs following addition of KIRxCD8 bispecific CD8 Treg Modulator and decrease in CD25+ CD4s on Day 5. Increase in Granzyme B content in Crohn's donor CD8 Tregs following flagellin-specific stimulation and treatment with KIRxCD8 bispecific CD8 Treg Modulator. Flagellin-specific activation of Crohn's donor CD4 T cells was reduced by treatment with KIRxCD8 bispecific CD8 Treg Modulator as detected by reduction in CD25^{+} CD4 T cells, CXCR3^{+} PD-1^{+} CD4 T cells, and IL-2.

CD8 Treg Modulator treatment reduces epithelial cell death in Celiac duodenal biopsy organoid cultures following gliadin-specific stimulation

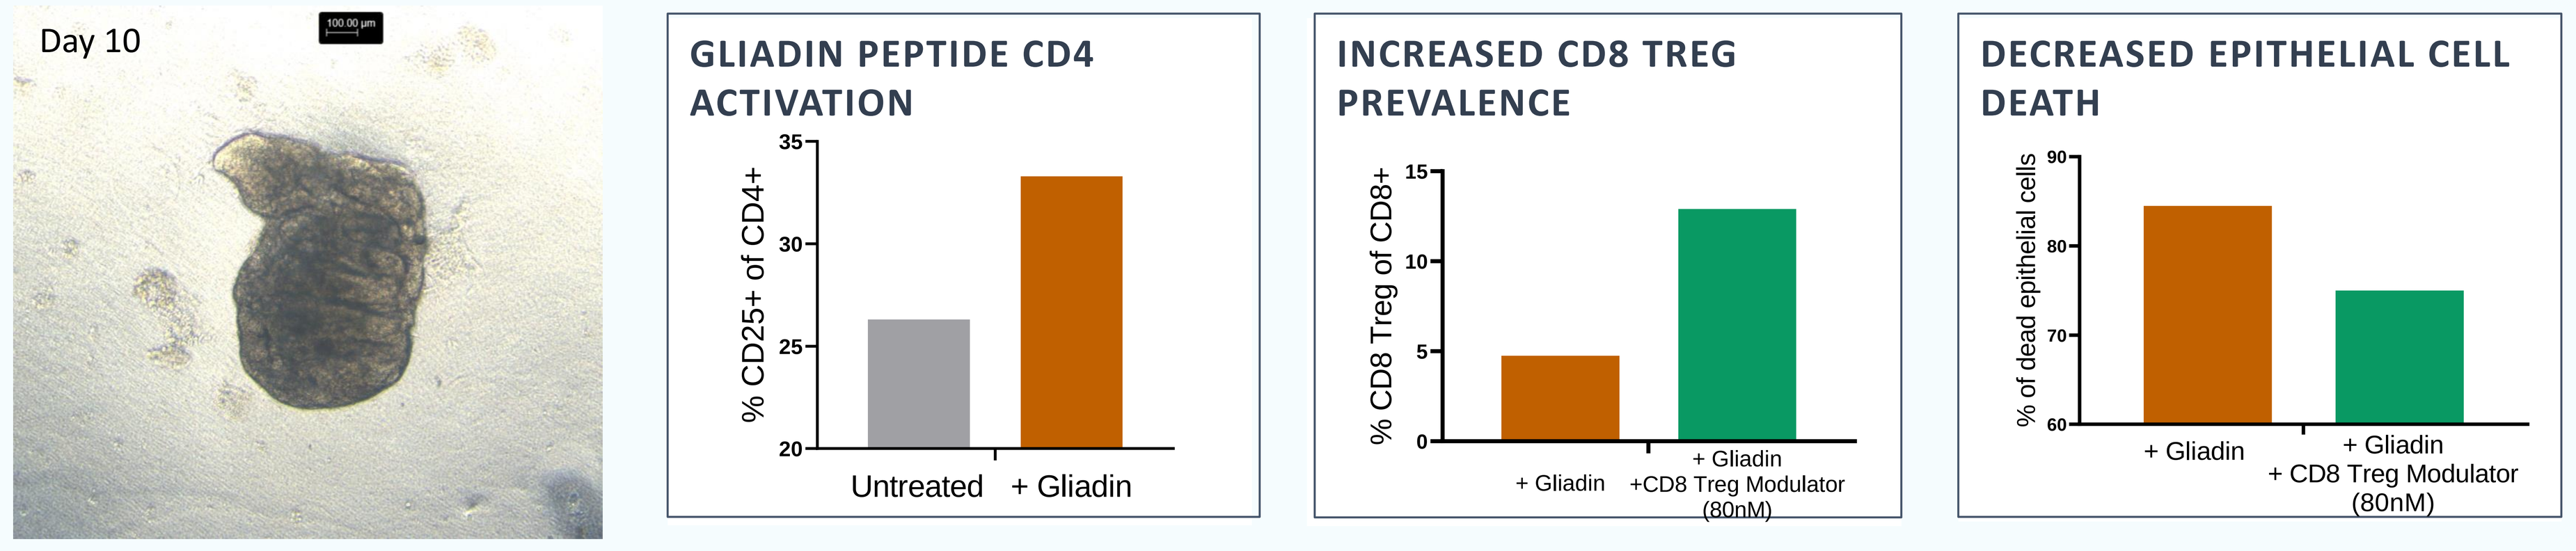
Day 10
GLIADIN PEPTIDE CD4 ACTIVATION
% CD25+ of CD4+
Untreated
+ Gliadin
INCREASED CD8 TREG PREVALENCE
% CD8 Treg of CD8+
+ Gliadin
+ Gliadin +CD8 Treg Modulator (80nM)
DECREASED EPITHELIAL CELL DEATH
% of dead epithelial cells
+ Gliadin
+ Gliadin + CD8 Treg Modulator (80nM)

Figure 5. Image of an organoid generated from a duodenal biopsy from a Celiac patient that was cultured for ten days in a collagen matrix in the presence of essential growth factors. Organoid were treated, harvested following culture, and processed for flow cytometry. Gliadin peptide stimulation activated CD4 T cells and increased epithelial cell death that was reduced with the addition of CD8 Treg Modulator. CD8 Treg Modulator treatment also expanded CD8 Tregs within the organoid culture.

CD8 Treg Modulator activates and expands CD8 Tregs in Human PBMC-NSG™ GvHD Model

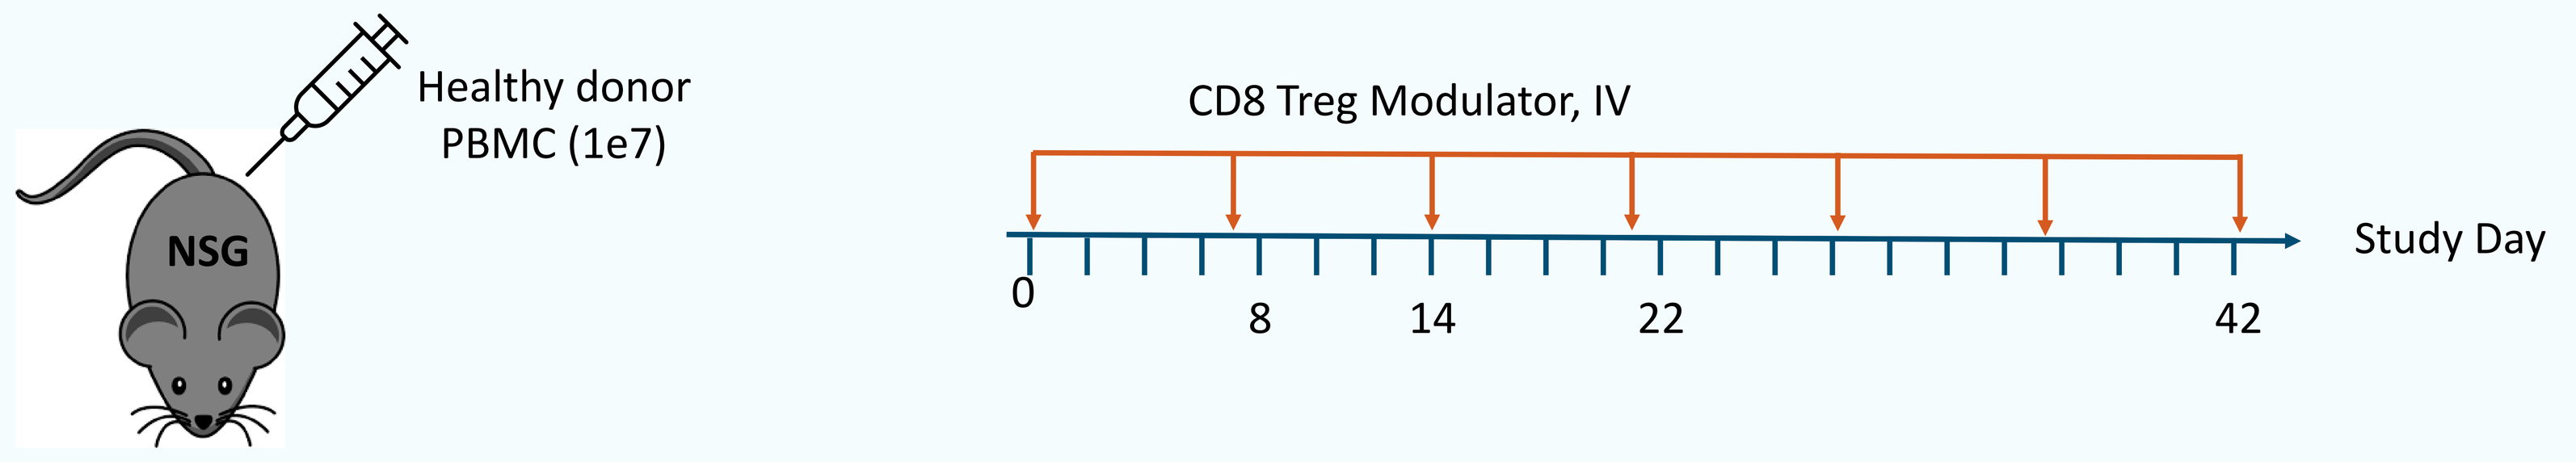
NSG
Healthy donor PBMC (1e7)
CD8 Treg Modulator, IV
Study Day
0
8
14
22
42

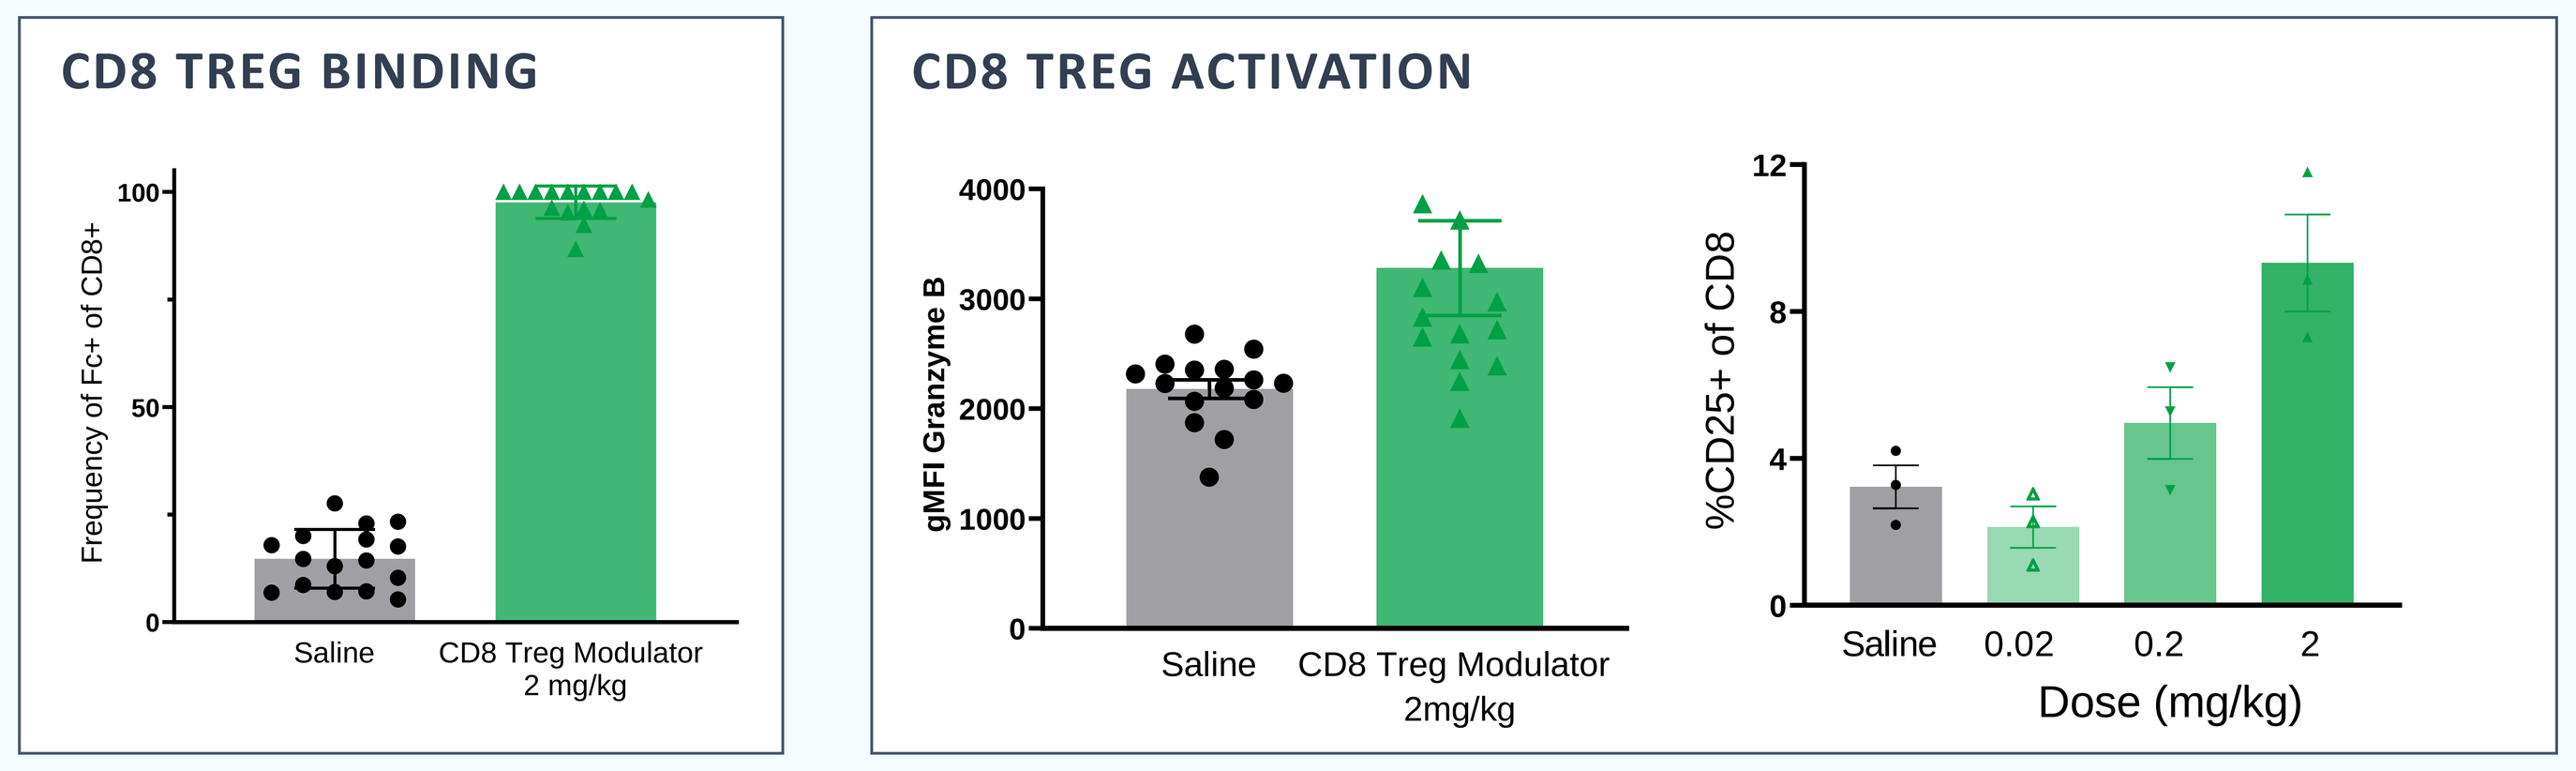
CD8 TREG BINDING
Frequency of Fc+ of CD8+
Saline
CD8 Treg Modulator 2 mg/kg
CD8 TREG ACTIVATION
gMFI Granzyme B
Saline
CD8 Treg Modulator 2mg/kg
%CD25+ of CD8
Dose (mg/kg)

Figure 6. NSG Mice were engrafted with healthy human PBMC followed by weekly doses of CD8 Treg Modulator or Saline control. Peripheral blood was collected at day 14 and CD8 Treg Modulator bound to CD8 Tregs was detected with an anti-human IgG1 Fc secondary antibody. CD8 Treg Modulator treated CD8 Tregs had increased Granzyme B content and a dose-dependent increase in the activation marker CD25.

CD8 Treg modulator ameliorates CD4 mediated pathology in Human PBMC-NSG™ GvHD Model

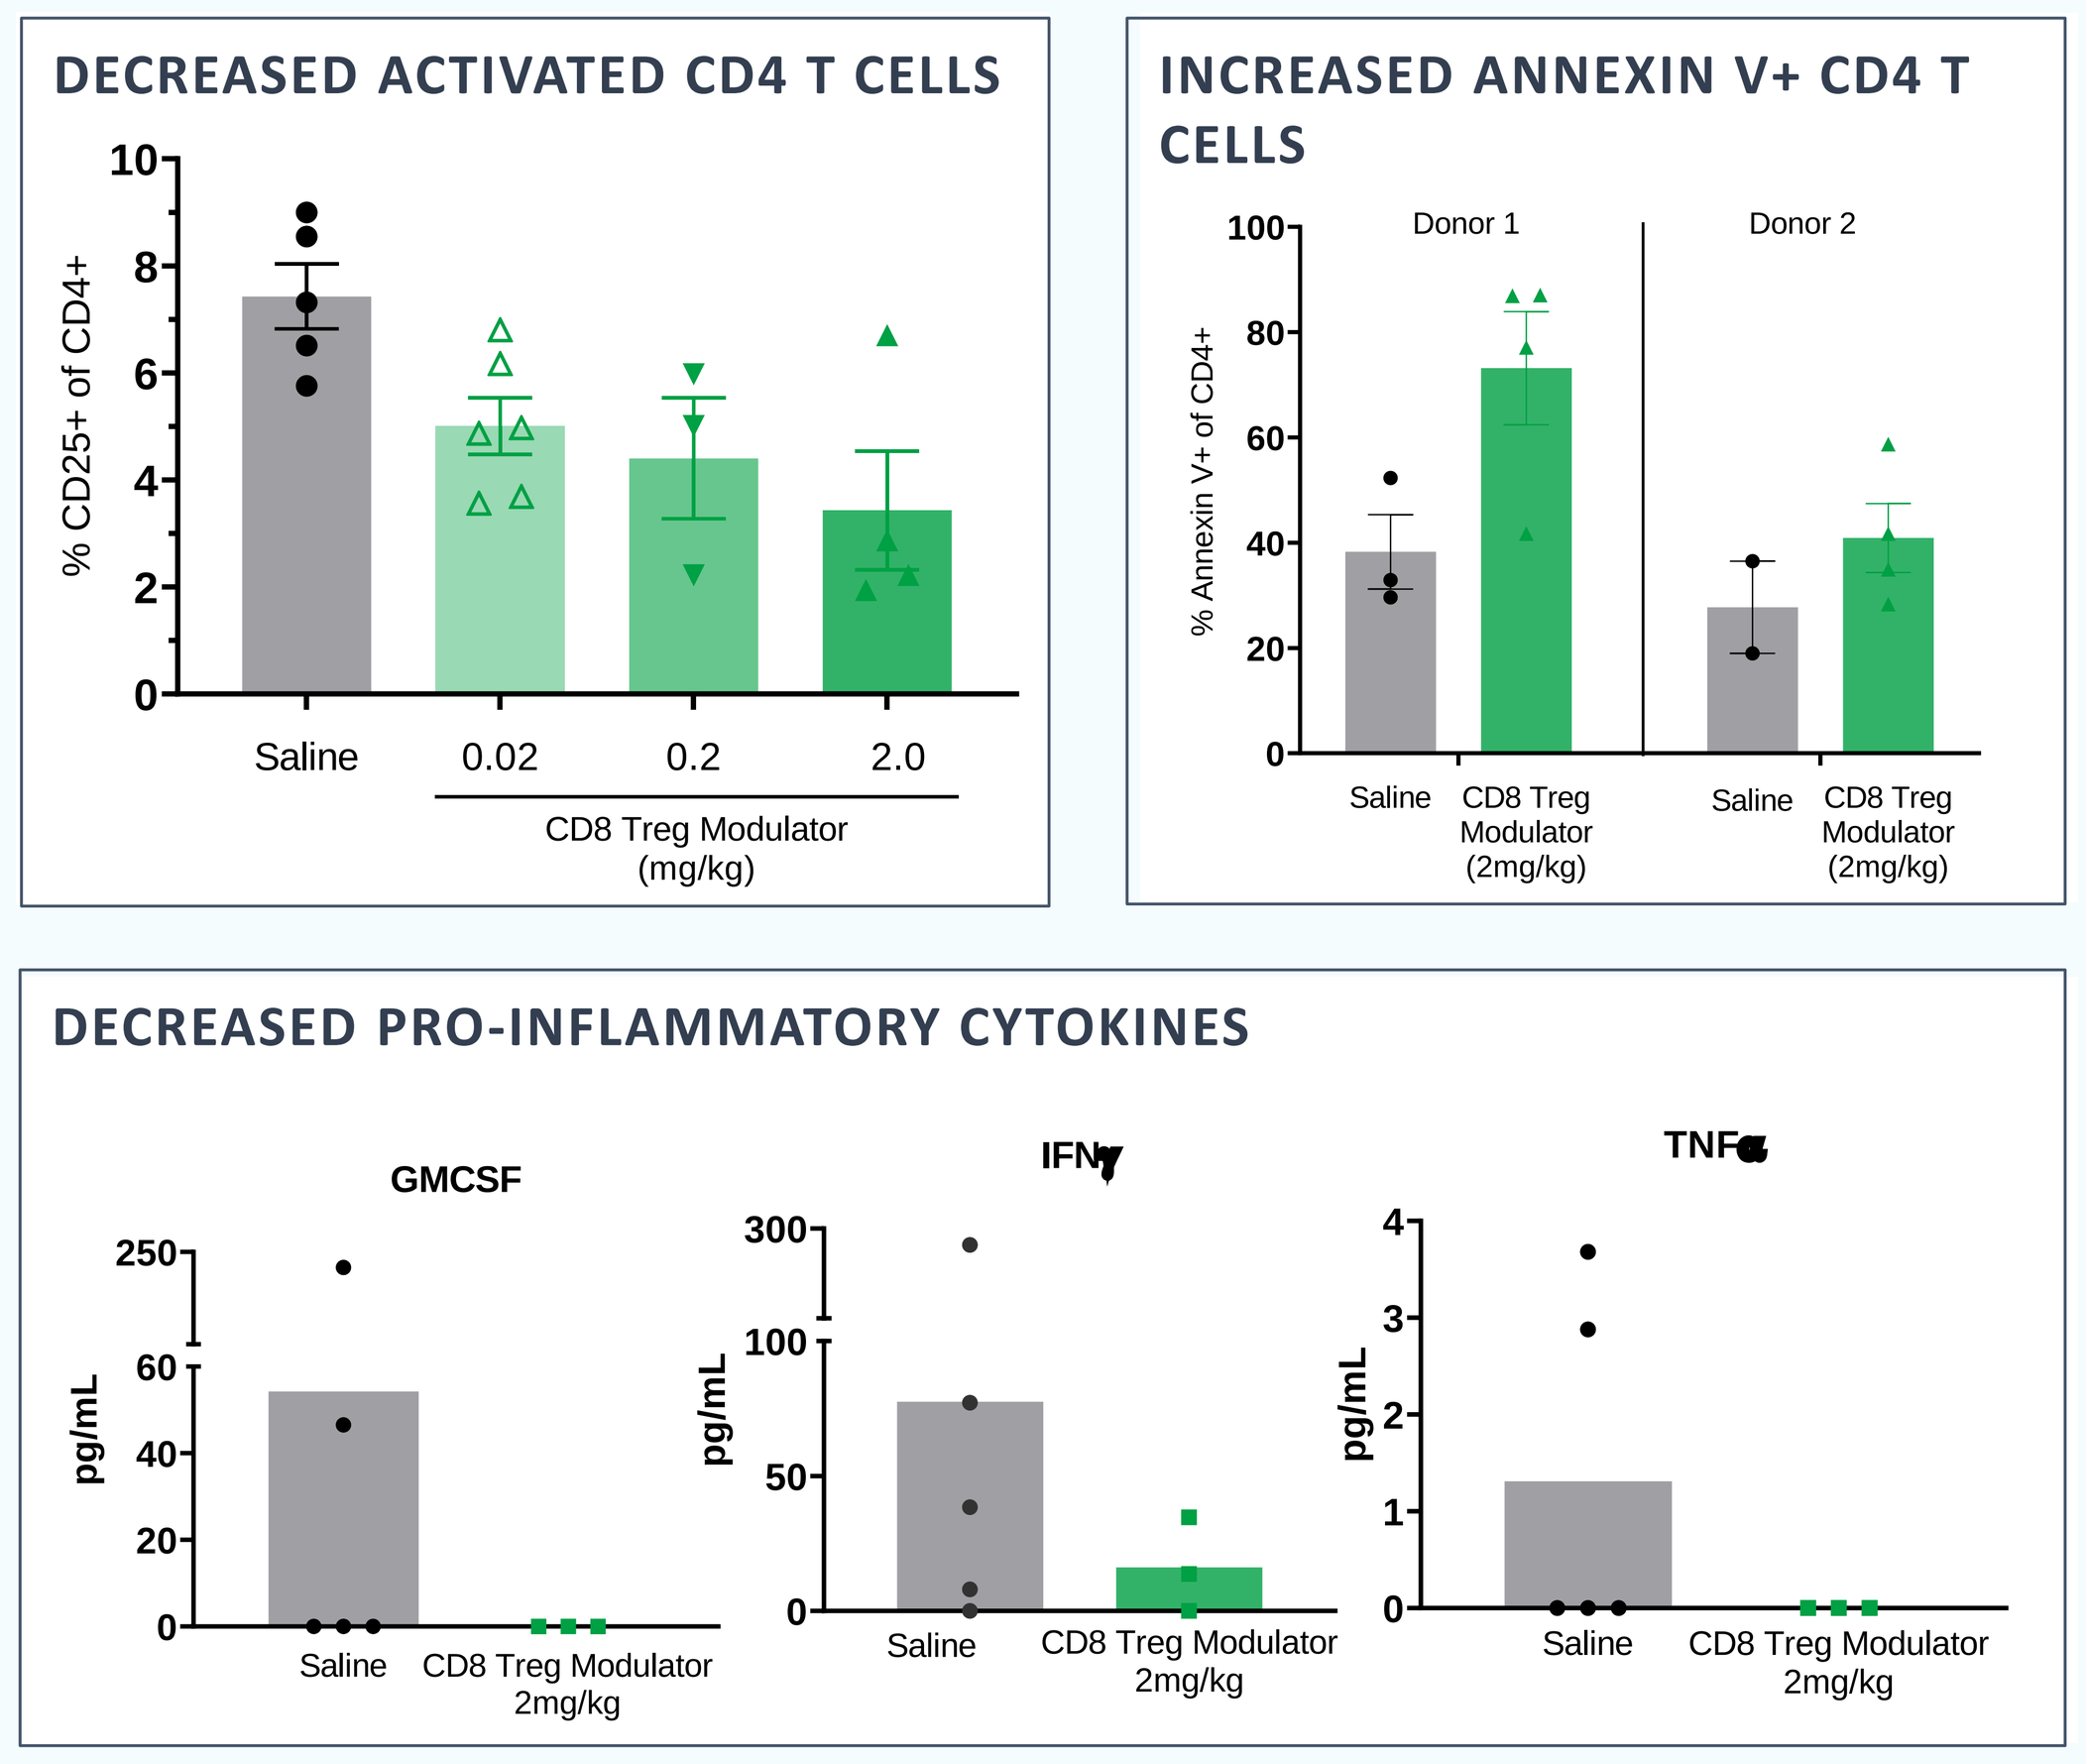
DECREASED ACTIVATED CD4 T CELLS
% CD25+ of CD4+
Saline
CD8 Treg Modulator (mg/kg)
INCREASED ANNEXIN V+ CD4 T CELLS
% Annexin V+ of CD4+
Donor 1
Donor 2
Saline
CD8 Treg Modulator (2mg/kg)
DECREASED PRO-INFLAMMATORY CYTOKINES
GMCSF
IFNγ
TNFα
pg/mL
Saline
CD8 Treg Modulator 2mg/kg

Figure 7. NSG Mice injected with human PBMC followed by weekly doses of CD8 Treg Modulator. Activated CD25+ CD4 T cells and the proinflammatory cytokines GMCSF, IFNγ, and TNFα were decreased with increasing dose of CD8 Treg Modulator with a corresponding increase in Annexin V+ CD4 T cells.

Dose-dependent concentration of CD8 Treg Modulator detected in humanized mice PBMC-NSG™ GvHD Model

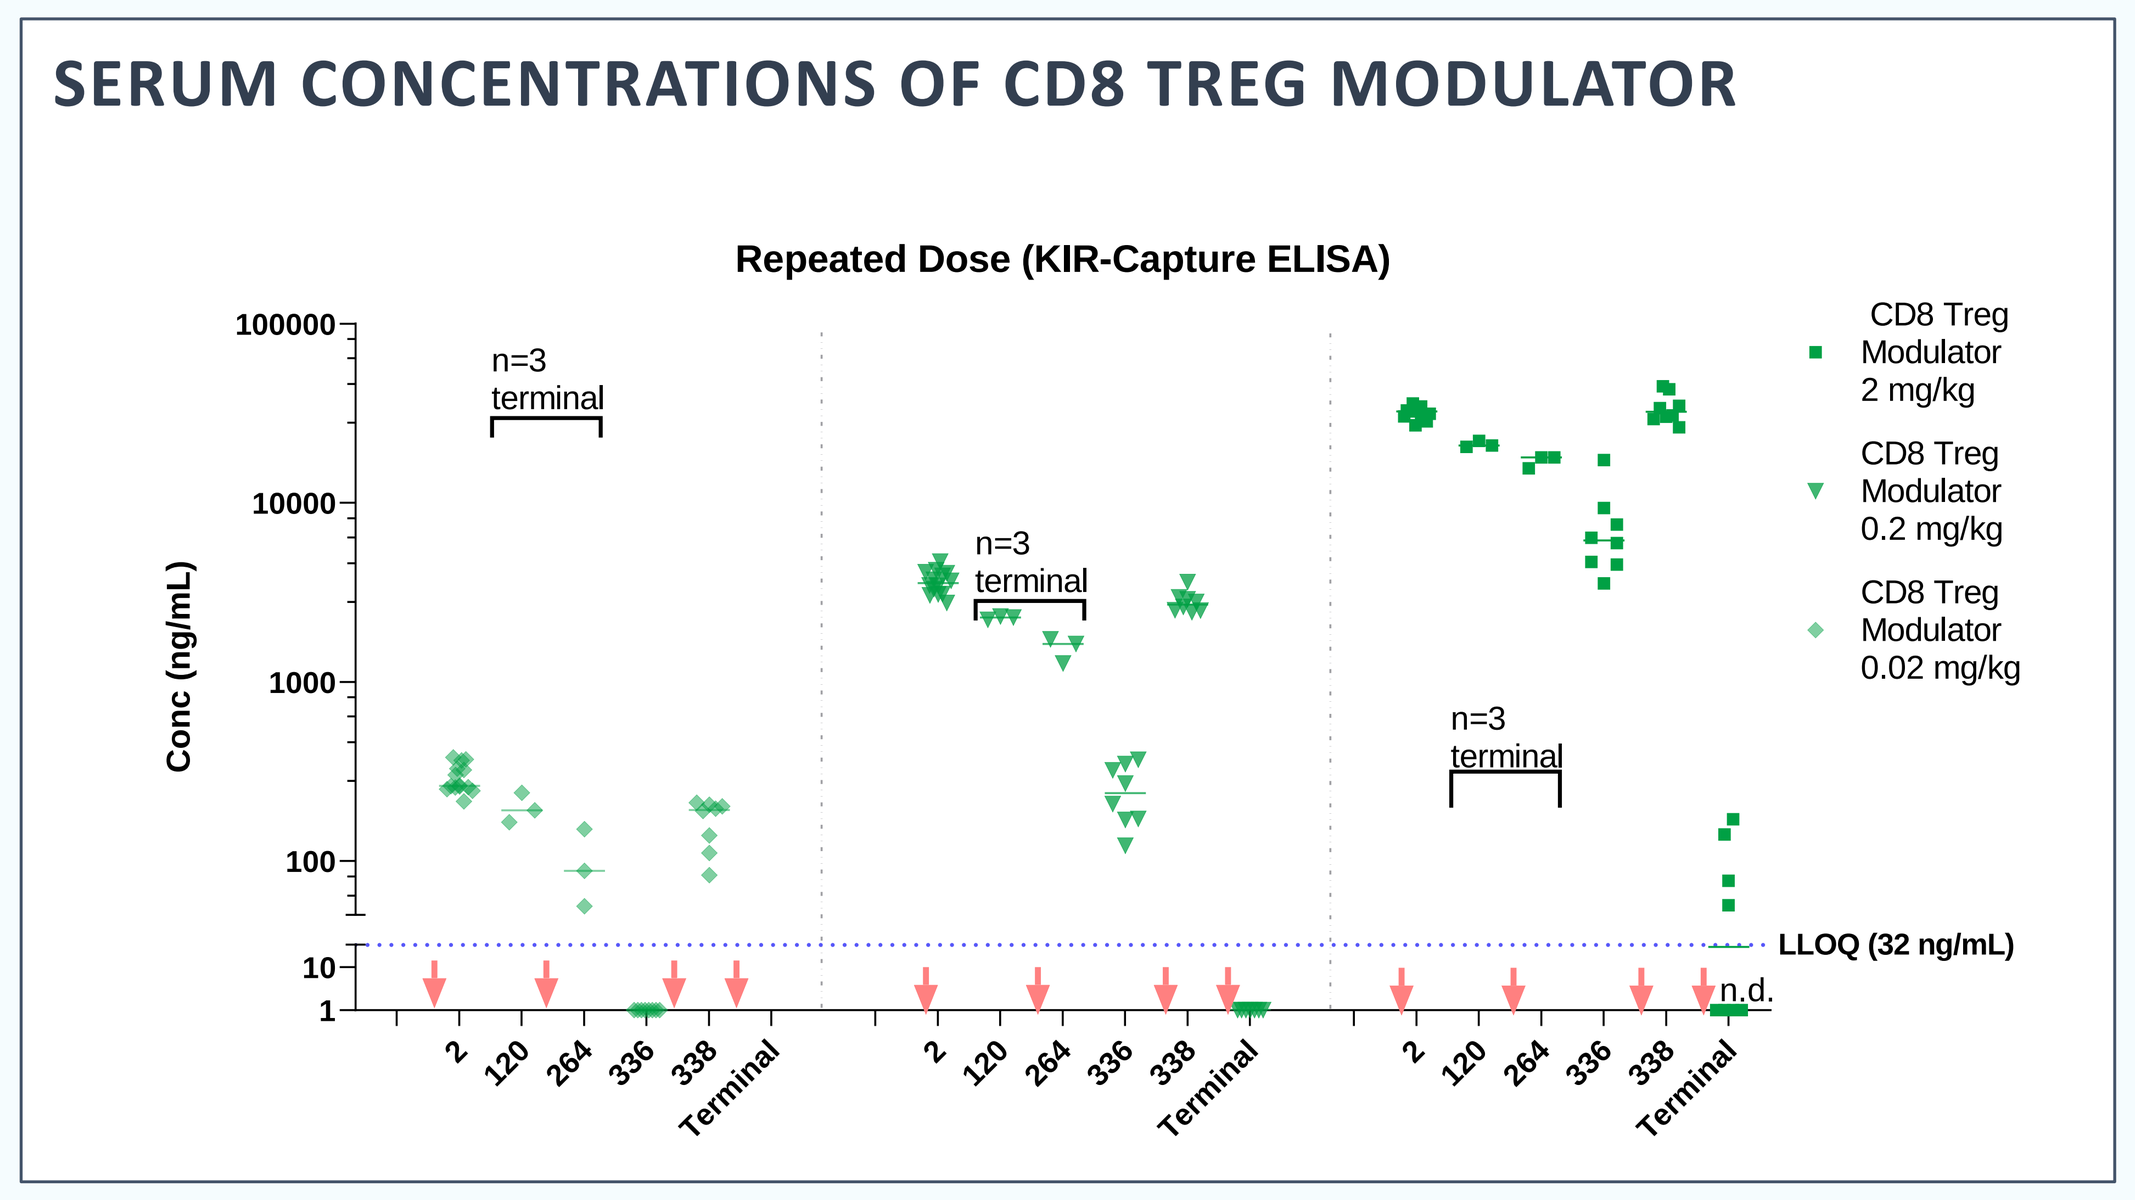
SERUM CONCENTRATIONS OF CD8 TREG MODULATOR
Repeated Dose (KIR-Capture ELISA)
Conc (ng/mL)
n=3 terminal
LLOQ (32 ng/mL)
n.d.
CD8 Treg Modulator 2 mg/kg
CD8 Treg Modulator 0.2 mg/kg
CD8 Treg Modulator 0.02 mg/kg
Terminal

Figure 8. Serum was collected at different time points following intravenous injection of different doses of KIRxCD8 bispecific CD8 Treg Modulator. Serum concentration was detected by KIR capture ELISA. n=14 mice at SD0, 2h; n=8 mice at 336h ; up to n=8 mice for terminal time points (ranging from 768 hr to 1008 hr), Interim terminal on 120 hr, 264 hr, n=3/time point Terminal analysis for 0.02 mg/kg group not done (n.d.)

Please see Mozart Poster #P970:** ***"Pre-clinical pharmacologic and tolerability characterization of a novel KIRxCD8 targeting bispecific CD8 Treg modulator"

Conclusions

- A novel KIR x CD8 targeting bispecific CD8 Treg Modulator selectively binds and activates CD8 Tregs to restore cytolytic function towards pathogenic CD4 T cells in Celiac and Crohn's donor derived PBMC.
- In ex vivo Celiac duodenal organoid cultures, treatment with the CD8 Treg Modulator increases CD8 Treg prevalence and reduces epithelial cell death induced by gluten-reactive CD4s.
- CD8 Treg Modulator increases CD8 Treg prevalence and activation while reducing CD4 pathology in a human PBMC NSG mouse model of acute GVHD.
- We postulate that the CD8 Treg Modulator can reduce autoreactive CD4 T cell prevalence to reestablish immune balance and may represent a selective, broadly applicable and novel therapeutic approach for the treatment of autoimmune diseases.

Acknowledgements:
The authors would like to thank Dr. Lord and patients for duodenal biopsies, members of the Kuo laboratory for assistance with the organoid culturing methods, and Jackson laboratories for performing GVHD studies.

Contact:
Follow up questions can be directed to Kristine Swiderek, Chief Scientific Officer, Mozart Therapeutics kswiderek@mozart-tx.com or by visiting the website at https://www.mozart-tx.com/

References:
Li et al. KIR $^{+}$ CD8 $^{+}$ T cells suppress pathogenic T cells and are active in autoimmune diseases and COVID-19. *Science.* 2022
DOI: 10.1126/science.abi9591